circVec: A novel circular RNA-based expression system to increase AAV potency and reduce therapeutic dose

PosterID: P0089
Presented at ESGCT 2026

O'Leary ET[1], Zhang J[1], Warsame M[1], Stefanelli S[1], Akter F[1], An Y[1], Liu J[1], Subramanian N[1], Melin O[1], Wiklund ED[1], Levitsky V[1], Hansen TB[1]
1. Circio AB, Huddinge, Sweden

Graphical Abstract

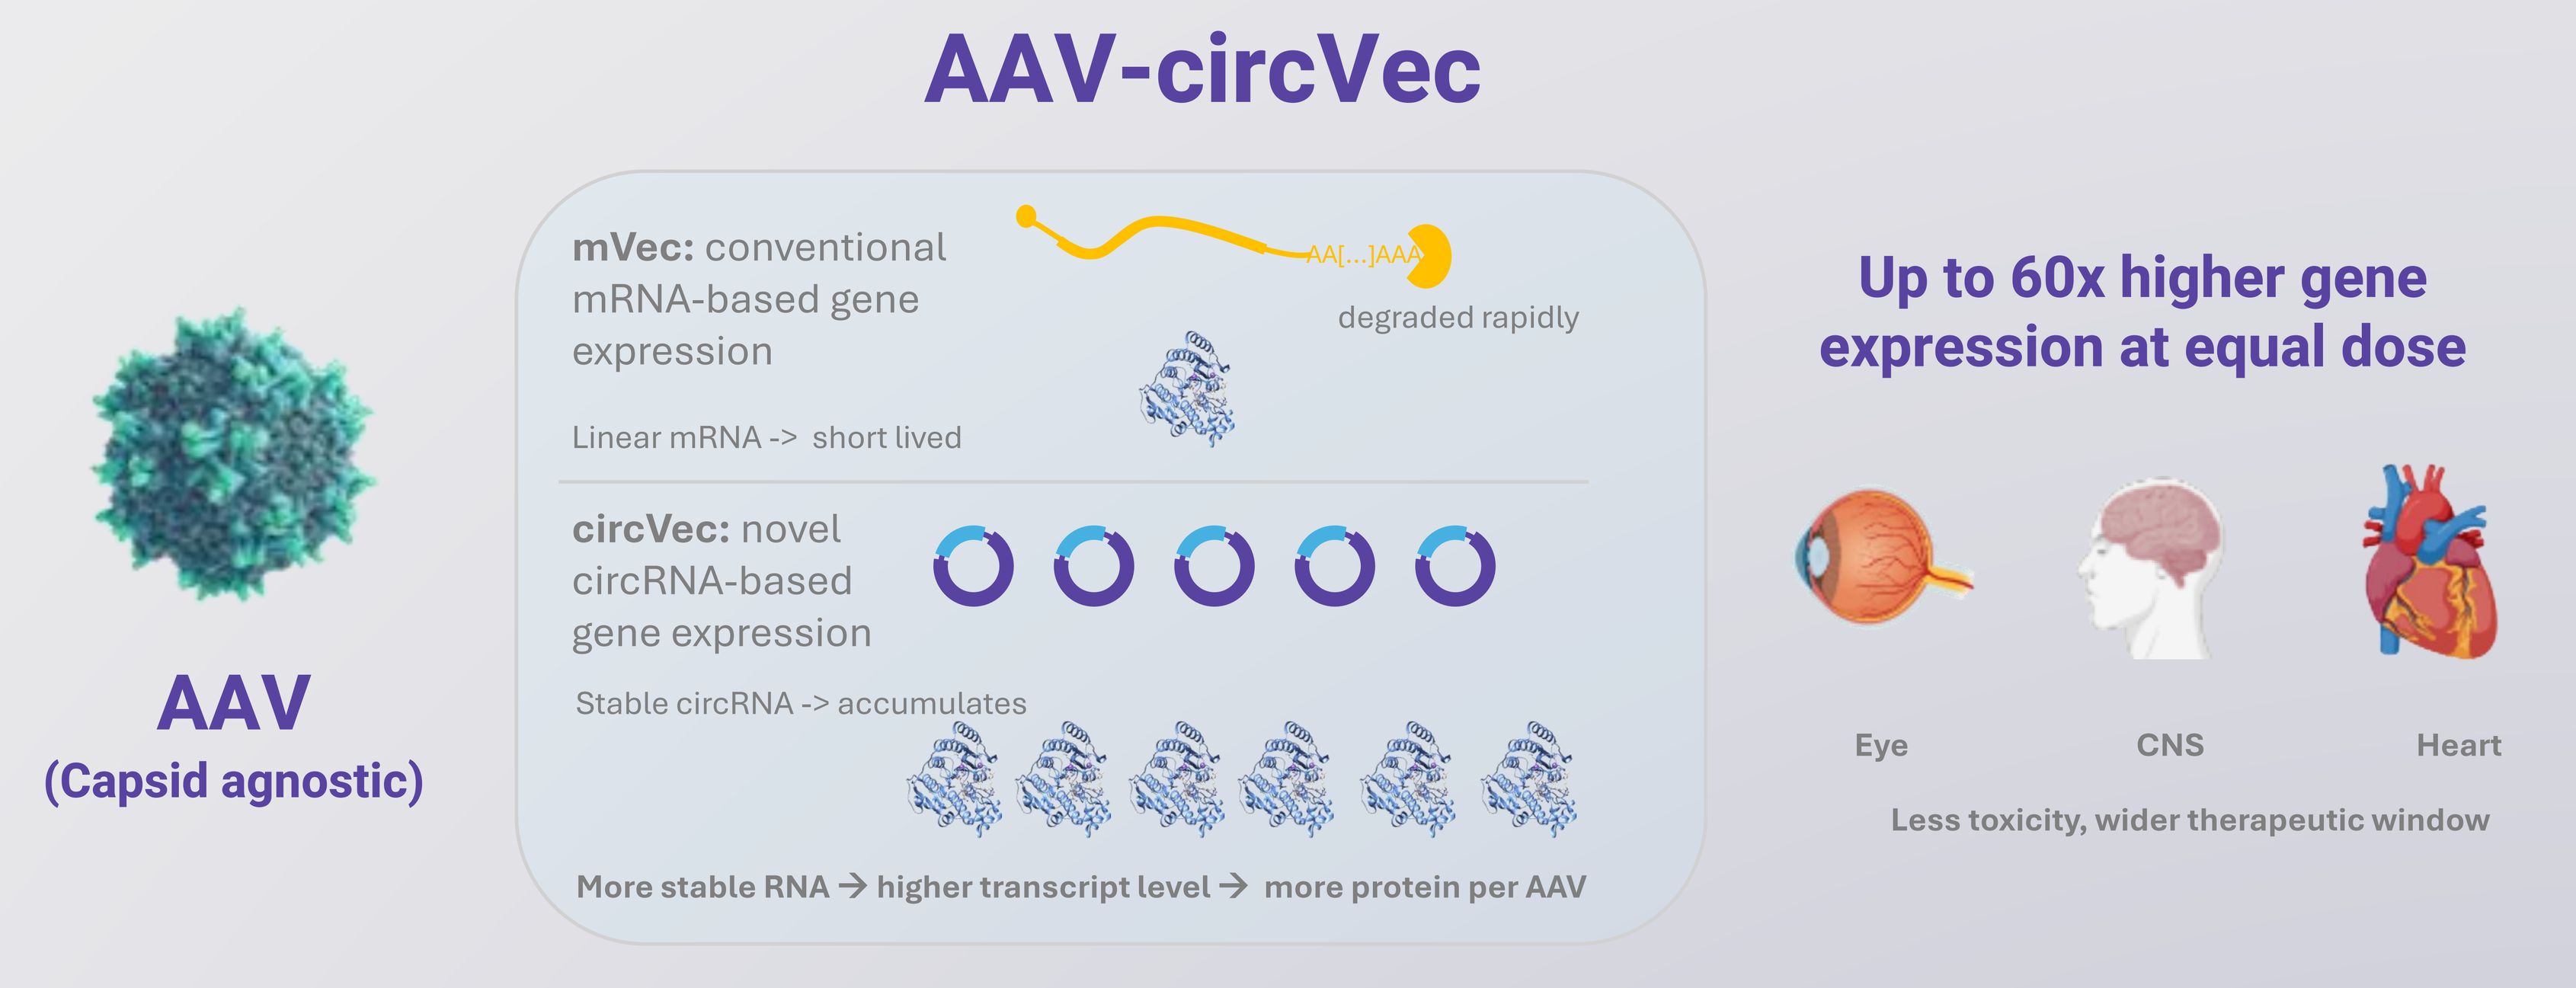
AAV-circVec
AAV
(Capsid agnostic)
mVec: conventional mRNA-based gene expression
degraded rapidly
Linear mRNA -> short lived
circVec: novel circRNA-based gene expression
Stable circRNA -> accumulates
More stable RNA → higher transcript level → more protein per AAV
Up to 60x higher gene expression at equal dose
Eye
CNS
Heart
Less toxicity, wider therapeutic window

1. Optimized circVec architecture outperforms conventional circRNA expression constructs by ~36×

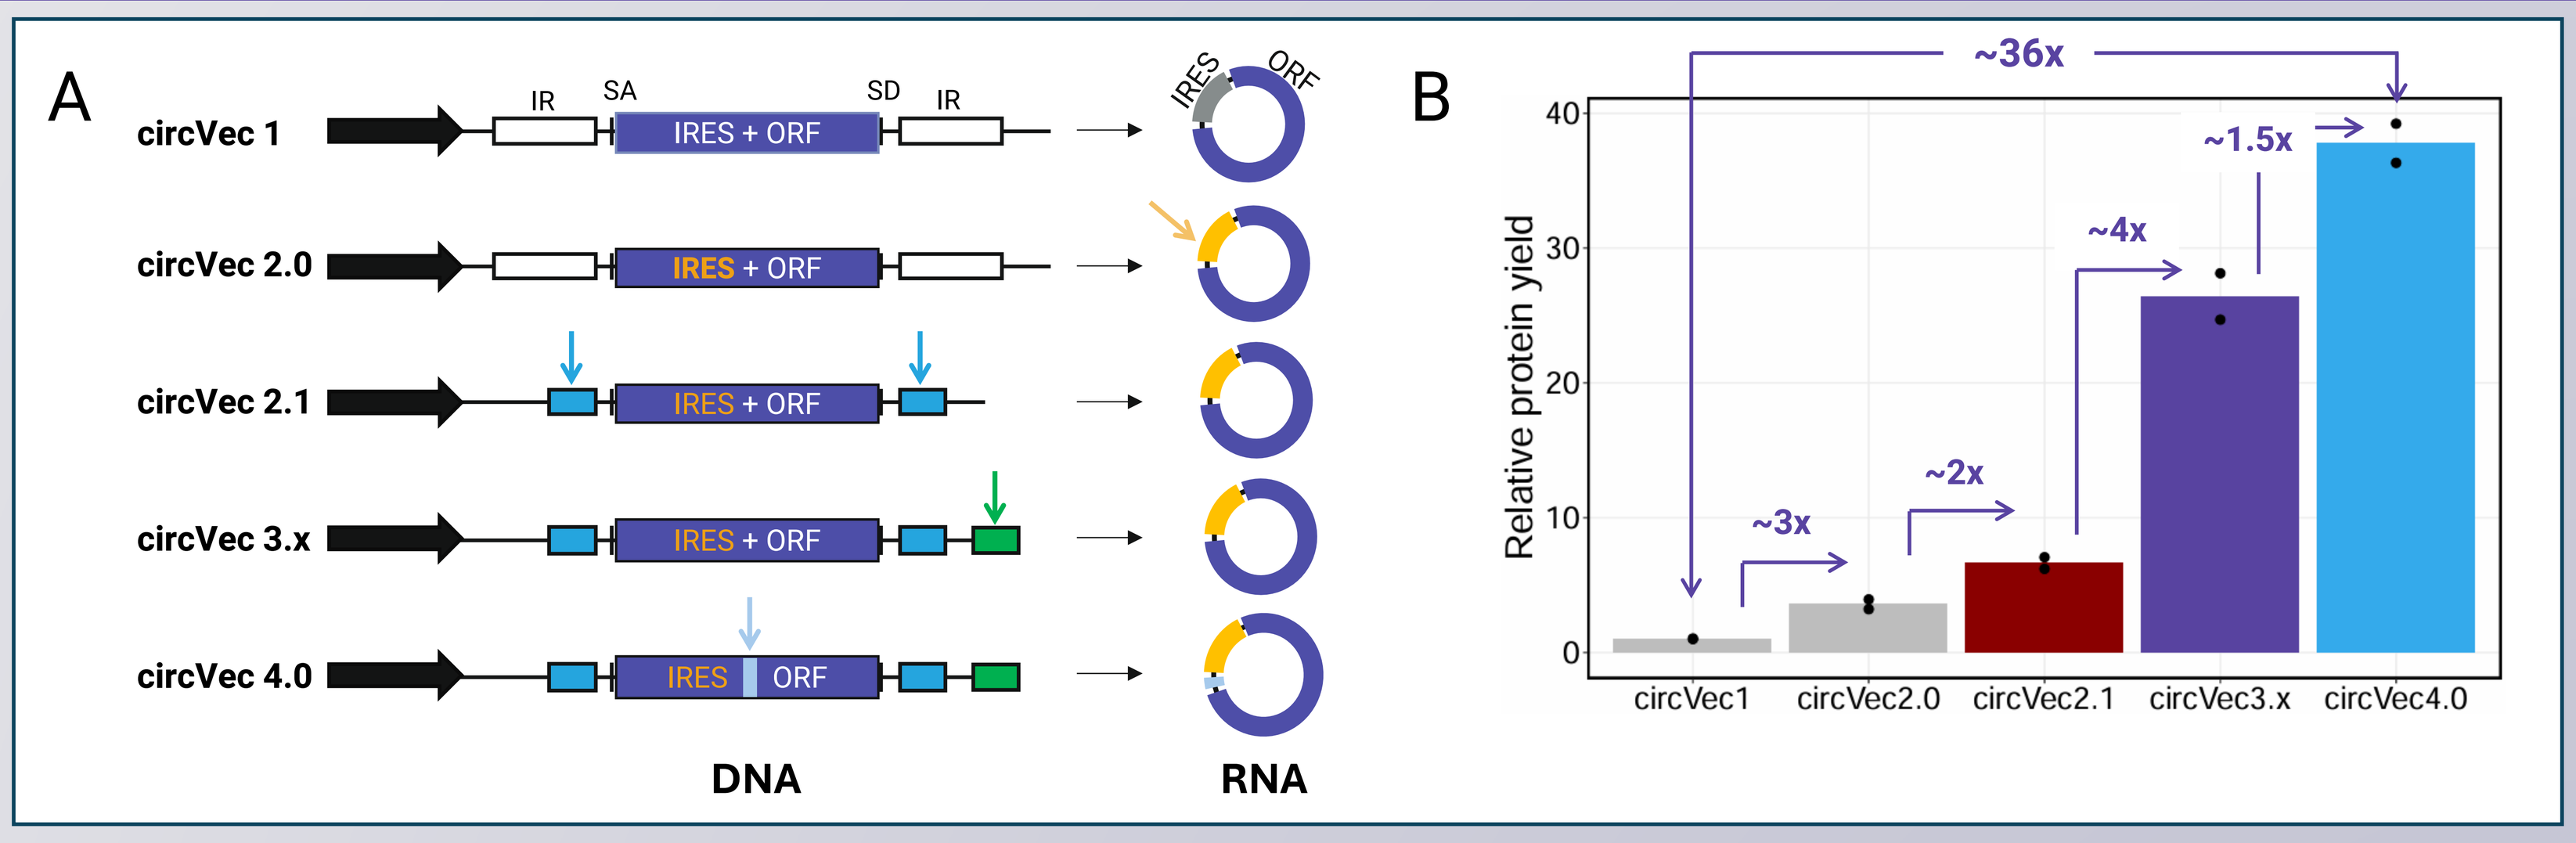
A
circVec 1
circVec 2.0
circVec 2.1
circVec 3.x
circVec 4.0
IR
SA
SD
IRES + ORF
DNA
RNA
B
Relative protein yield
~36x
~1.5x
~4x
~2x
~3x
circVec1
circVec2.0
circVec2.1
circVec3.x
circVec4.0

Figure 1: circVec platform evolution. A) Schematic representation of circVec designs from generation 1 → 4. **B)** Relative protein yield from different generations of circVec in HepG2 cells (n=2).

2. circRNA stability confers enhanced protein expression from equal vector dose *in vivo*

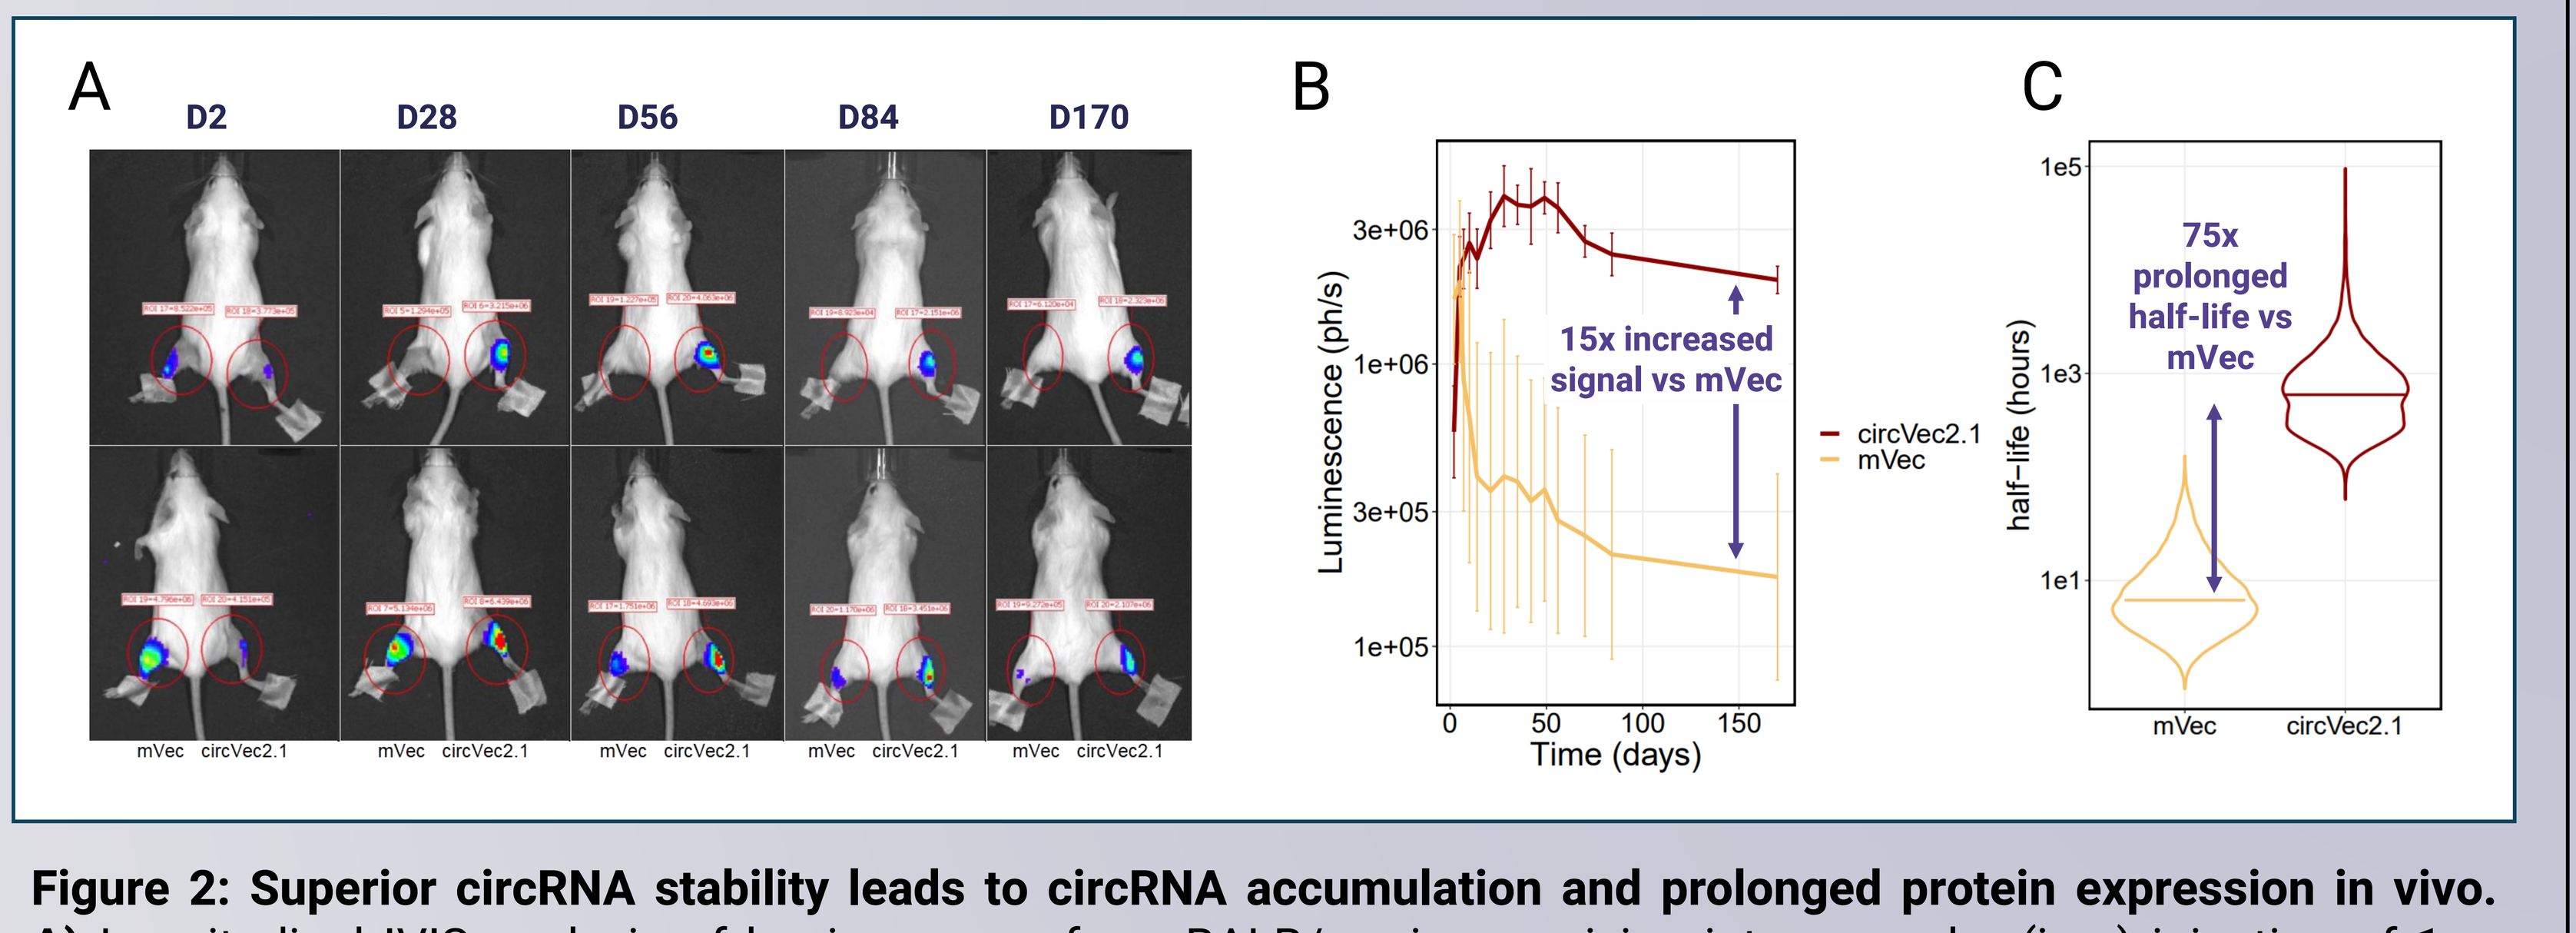
A
D2
D28
D56
D84
D170
B
Luminescence (ph/s)
15x increased signal vs mVec
circVec2.1
mVec
Time (days)
C
half-life (hours)
75x prolonged half-life vs mVec
mVec
circVec2.1

Figure 2: Superior circRNA stability leads to circRNA accumulation and prolonged protein expression in vivo. A) Longitudinal IVIS analysis of luminescence from BALB/c mice receiving intramuscular (i.m.) injection of 1ug pcDNA3 vectors expressing a Firefly luciferase-encoding mRNA (mVec, left hindleg) or circular RNA (circVec, right hindleg). **B)** Quantification of signal from hindleg muscles presented as geometric mean ± s.e.m. (n=4). **C)** Inference of RNA half-life based on MCMC modelling (10000 iterations) of expression profile over time presented as violin plot with median.

3. Intra-vitreal delivery of AAV2-circVec increases protein expression in eye by up to ~50x

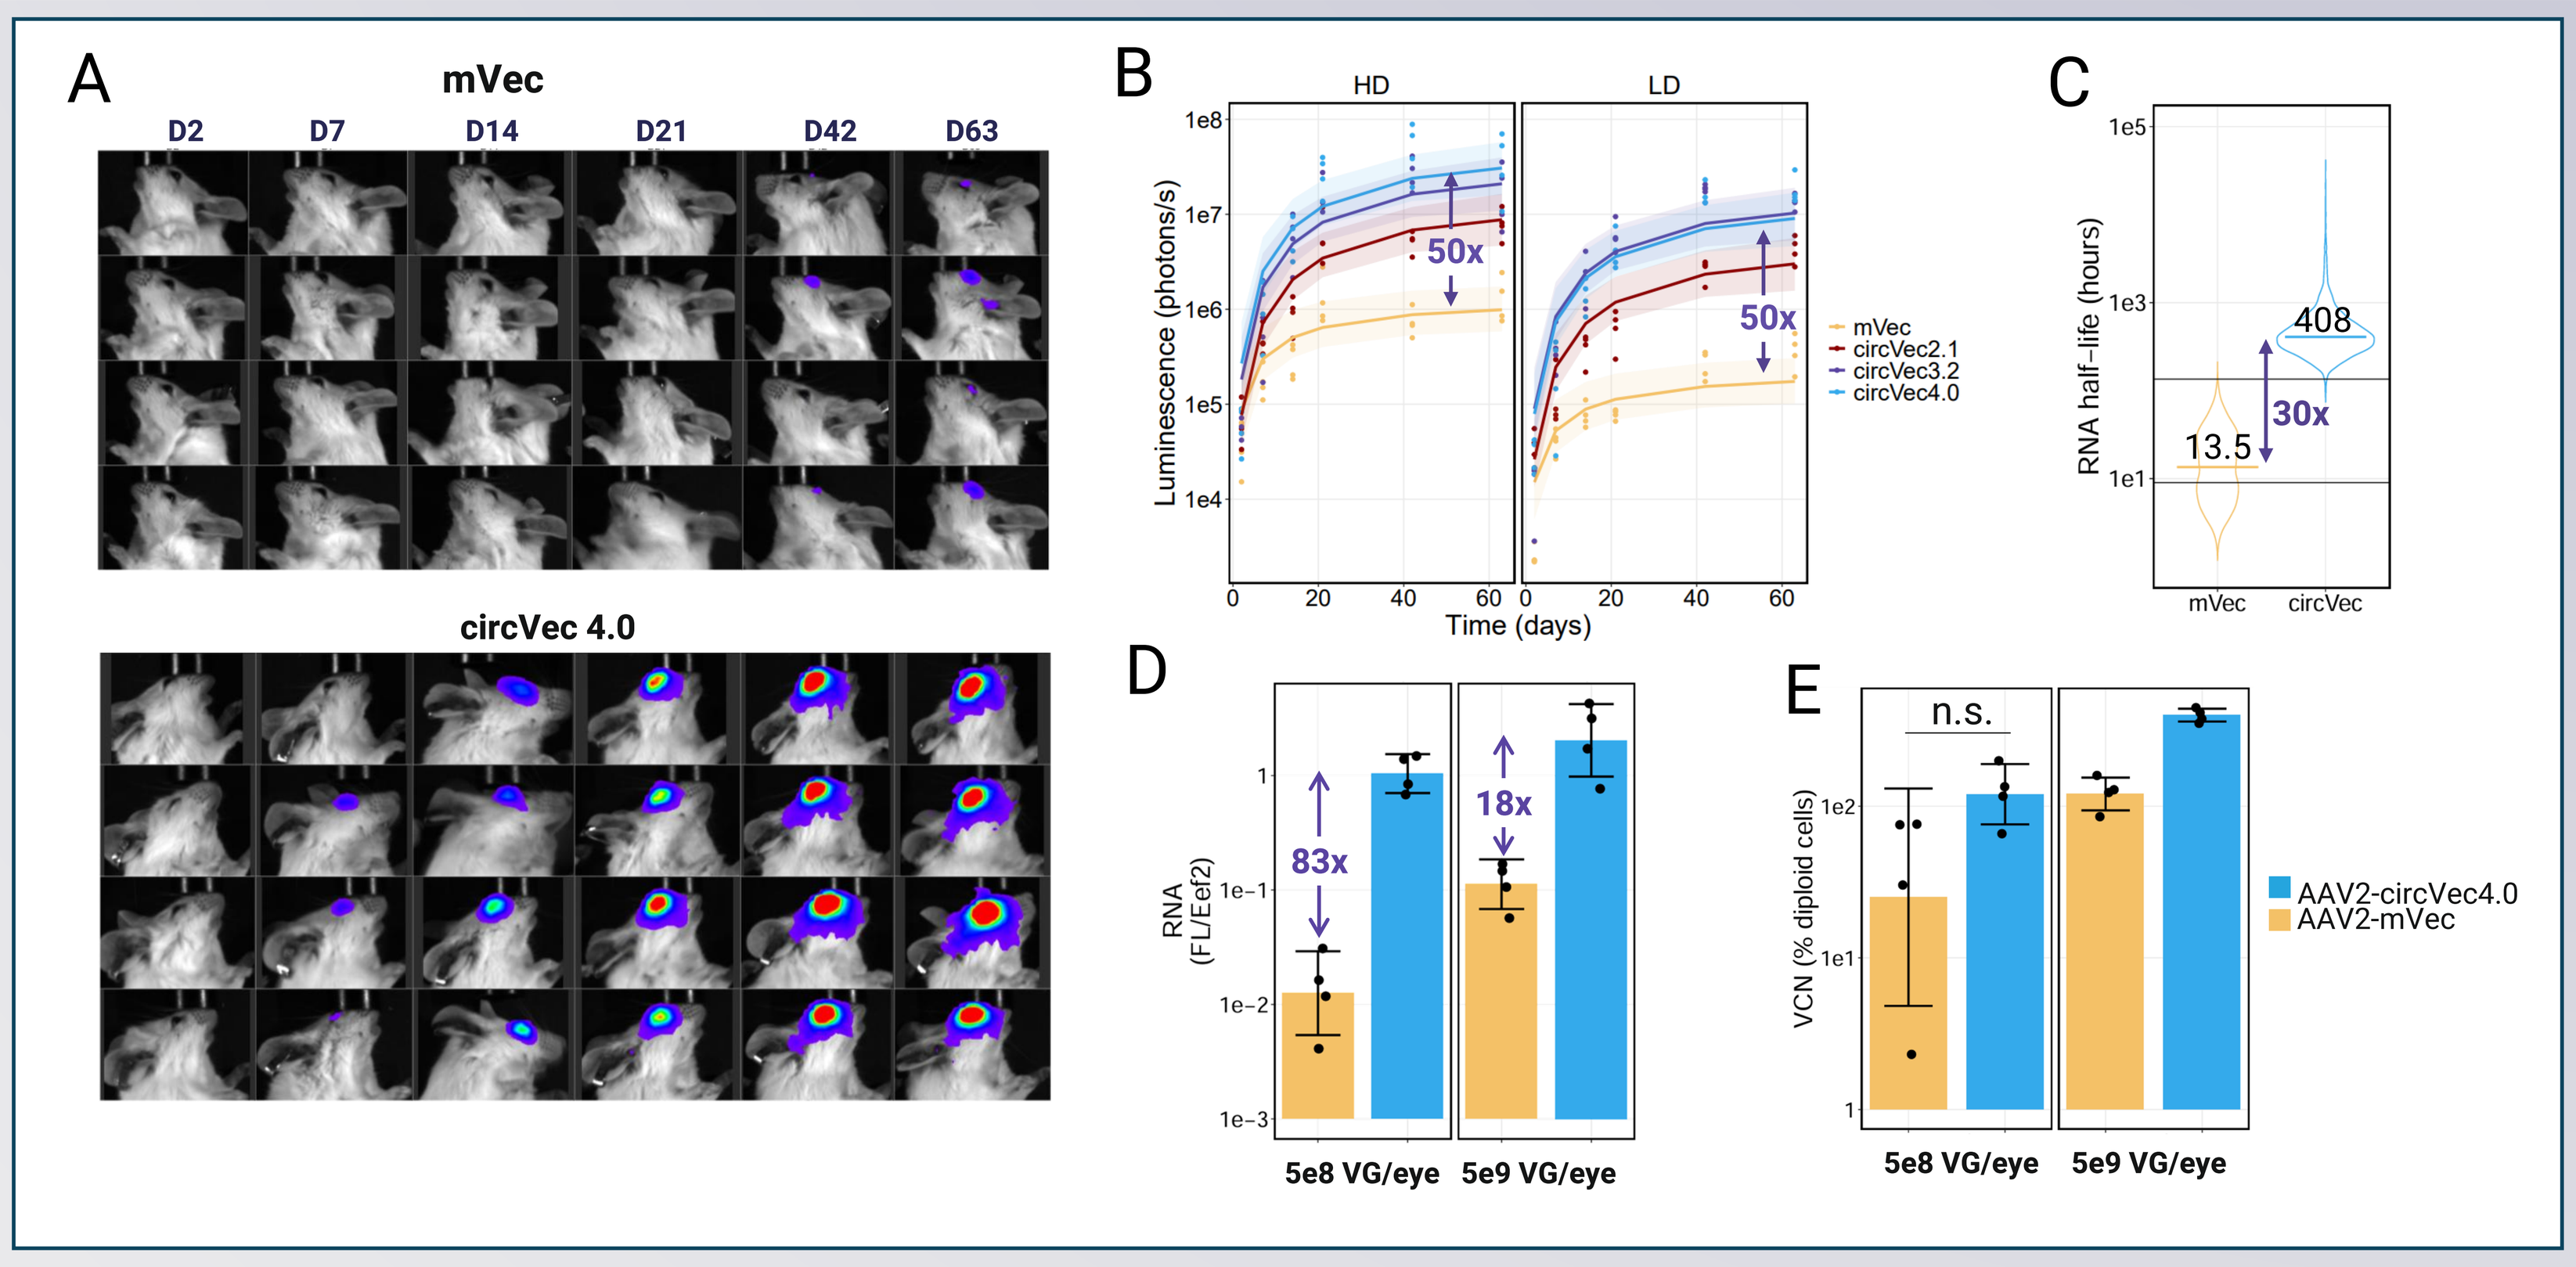
A
mVec
D2
D7
D14
D21
D42
D63
circVec 4.0
B
HD
LD
Luminescence (photons/s)
50x
50x
mVec
circVec2.1
circVec3.2
circVec4.0
Time (days)
C
RNA half-life (hours)
408
30x
13.5
mVec
circVec
D
RNA (FL/Eef2)
83x
18x
5e8 VG/eye
5e9 VG/eye
E
VCN (% diploid cells)
n.s.
AAV2-circVec4.0
AAV2-mVec
5e8 VG/eye
5e9 VG/eye

Figure 3: In vivo expression of CMV-driven AAV2-circVec 4.0 in the eye by intravitreal delivery. A) Longitudinal IVIS imaging of ocular luminescence at the indicated timepoints after intravitreal injection of 5e9 or 5e8 vg of AAV2-mVec (right eye) and AAV2-circVec4.0 (left eye) in BALB/c mice. **B)** Ocular luminescence over time for AAV2-mVec and the AAV2-circVec generations, shown as geometric mean ± s.e.m. **C)** RNA half-life inferred by MCMC modelling (10,000 iterations) of the expression profile over time, shown as a violin plot with median. **D, E)** Firefly RNA (D) and AAV genome (E) quantified in eye tissue 63 days after intravitreal injection of 5e8 or 5e9 vg/eye of AAV2-circVec4.0 or AAV2-mVec, as indicated. Significance was determined by t-test on log-transformed values; n.s., not significant.

4. Local delivery of AAV9-circVec increases protein expression in CNS by up to 60x

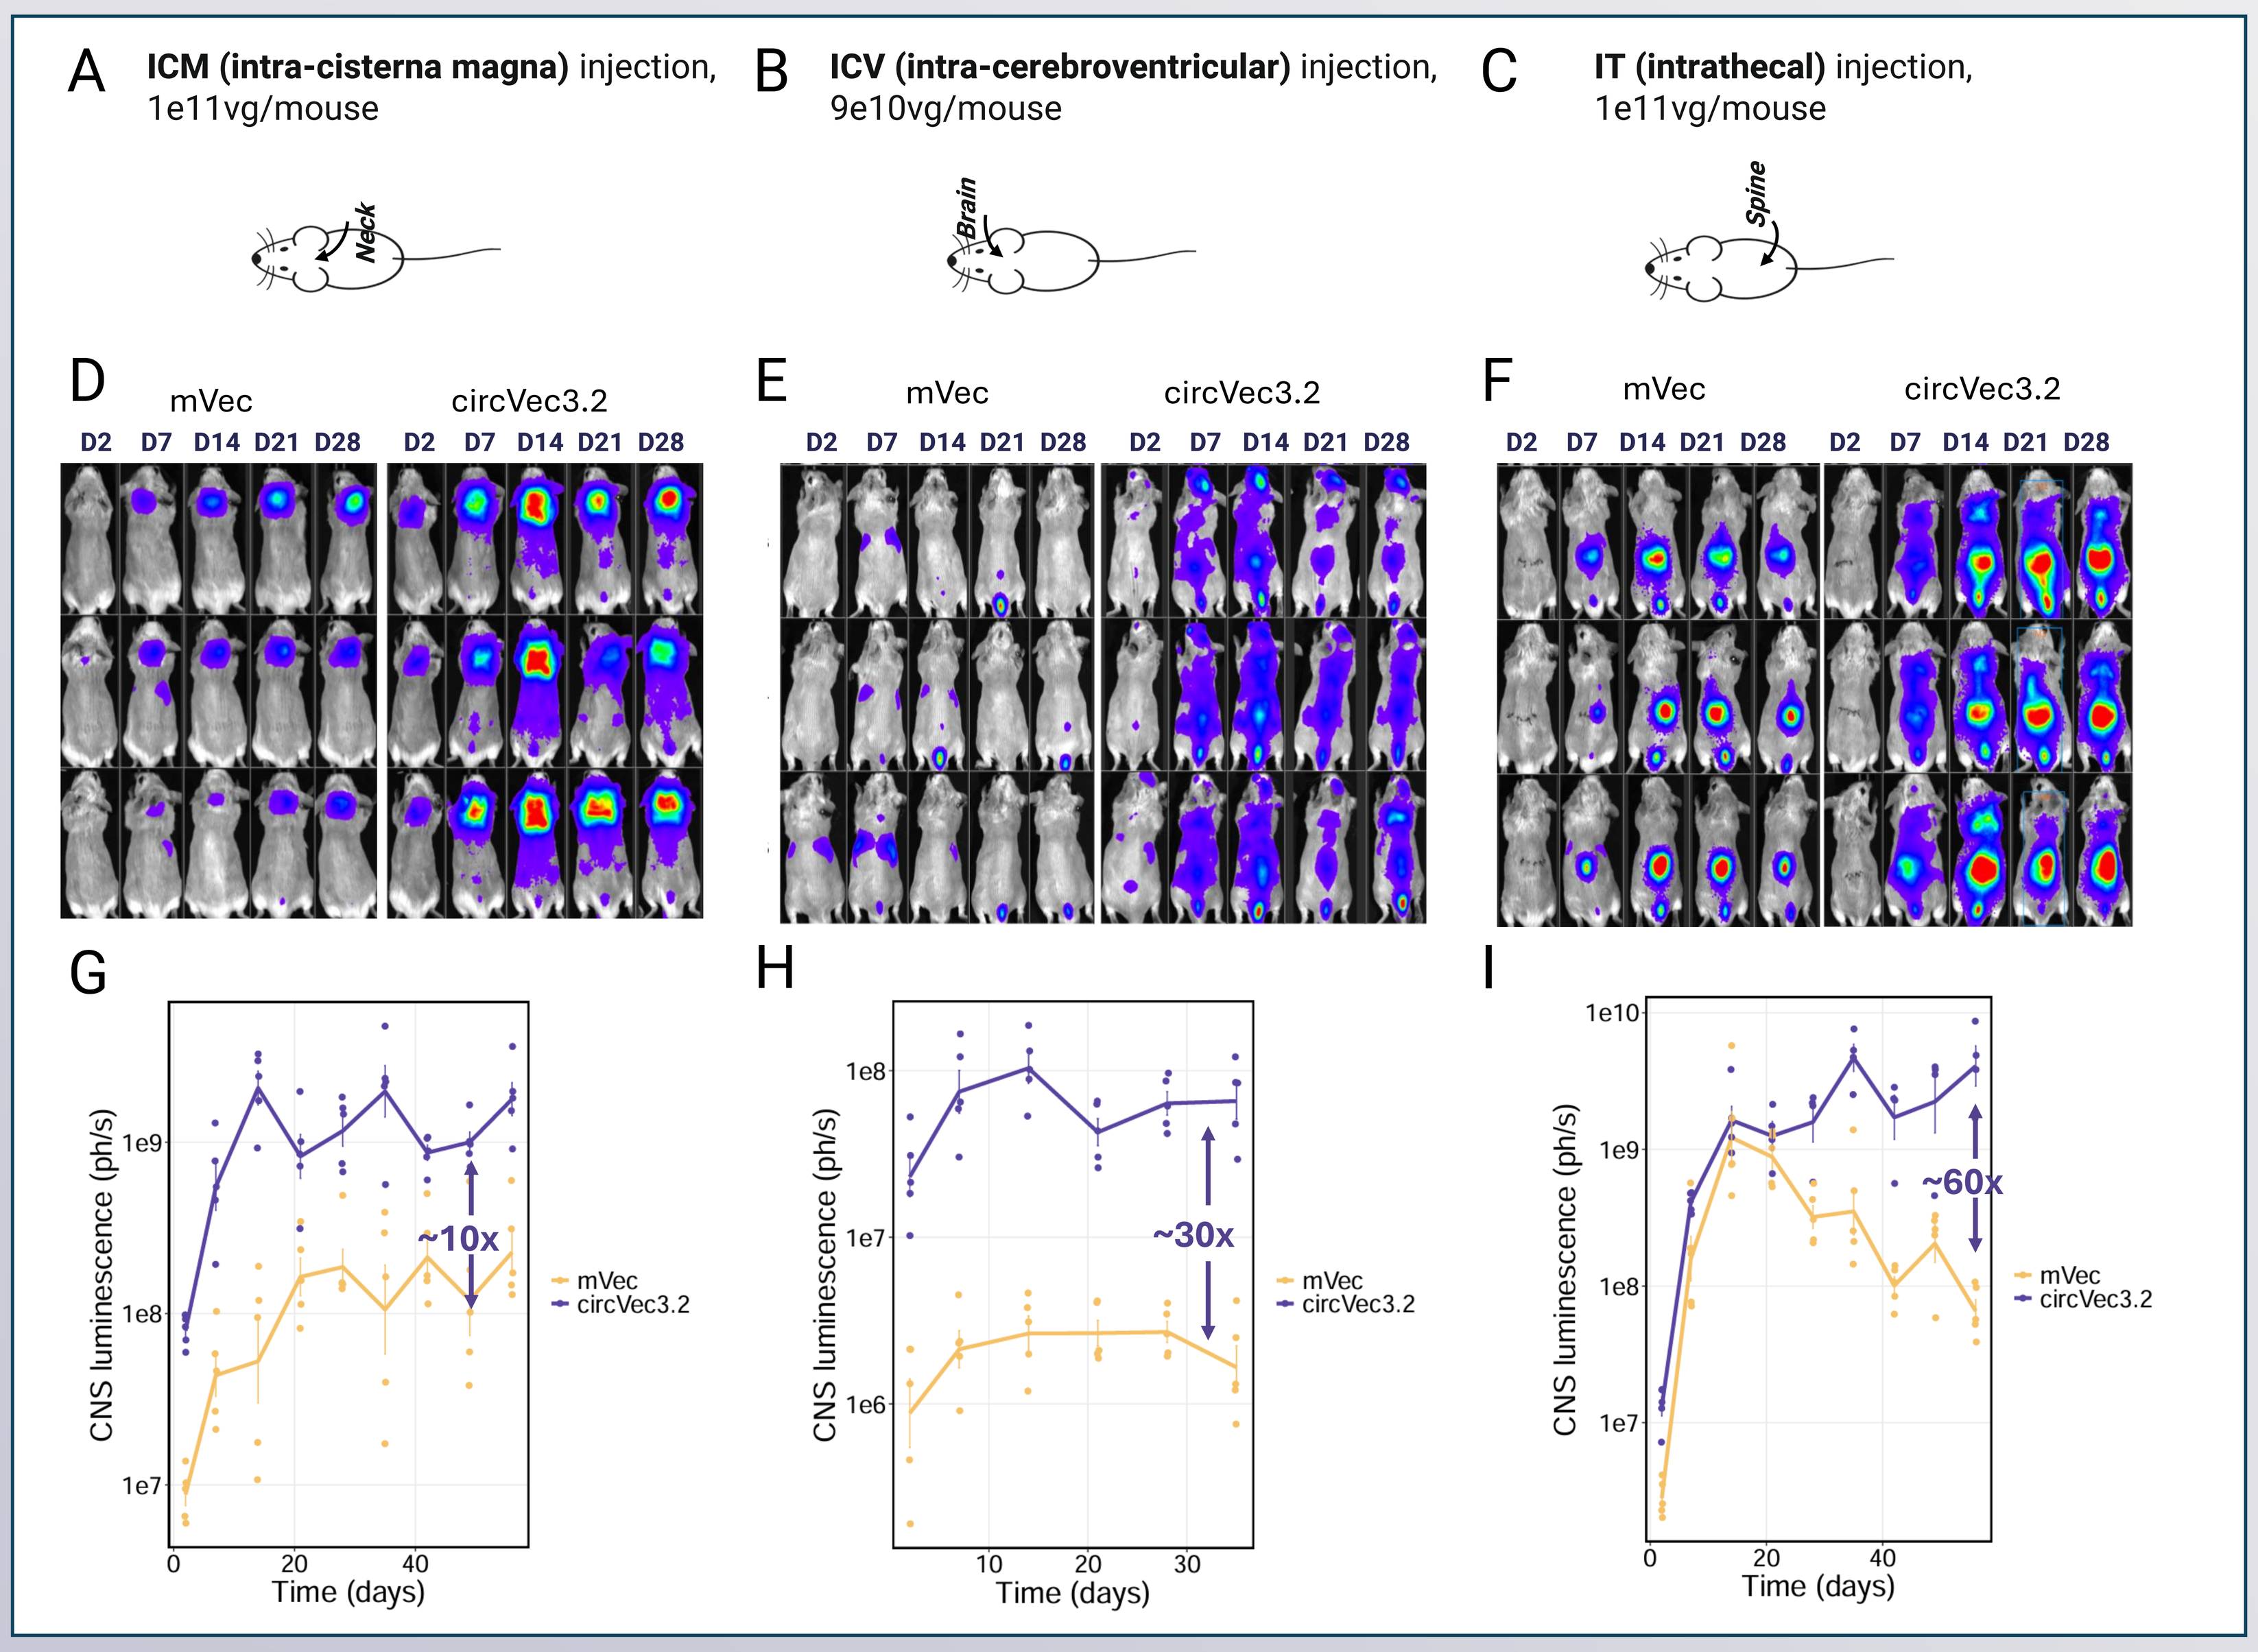
A ICM (intra-cisterna magna) injection, 1e11vg/mouse
B ICV (intra-cerebroventricular) injection, 9e10vg/mouse
C IT (intrathecal) injection, 1e11vg/mouse
Neck
Brain
Spine
D
E
F
mVec
circVec3.2
D2 D7 D14 D21 D28
G
H
I
CNS luminescence (ph/s)
~10x
~30x
~60x
mVec
circVec3.2
Time (days)

Figure 4: In vivo expression of CMV-driven AAV9-circVec 3.2 in CNS by three delivery routes. A-C) Schematics of the routes of administration and doses of AAV9-circVec3.2 or AAV9-mVec expressing Firefly luciferase from a ubiquitous promoter in BALB/c mice: (A) intra-cisterna magna (ICM), 1e11 vg/mouse; (B) intra-cerebroventricular (ICV), 9e10 vg/mouse; (C) intrathecal (IT), 1e11 vg/mouse. **D–F)** Longitudinal IVIS imaging of luminescence for AAV9-circVec3.2 vs AAV9-mVec after ICM (D), ICV (E) or IT (F) injection. **G–I)** Luminescence quantified from the site of injection after ICM (G), ICV (H) or IT (I) injection, shown as geometric mean ± s.e.m.

5. Novel circVec5 features unlock up to 5-fold higher cardiac expression

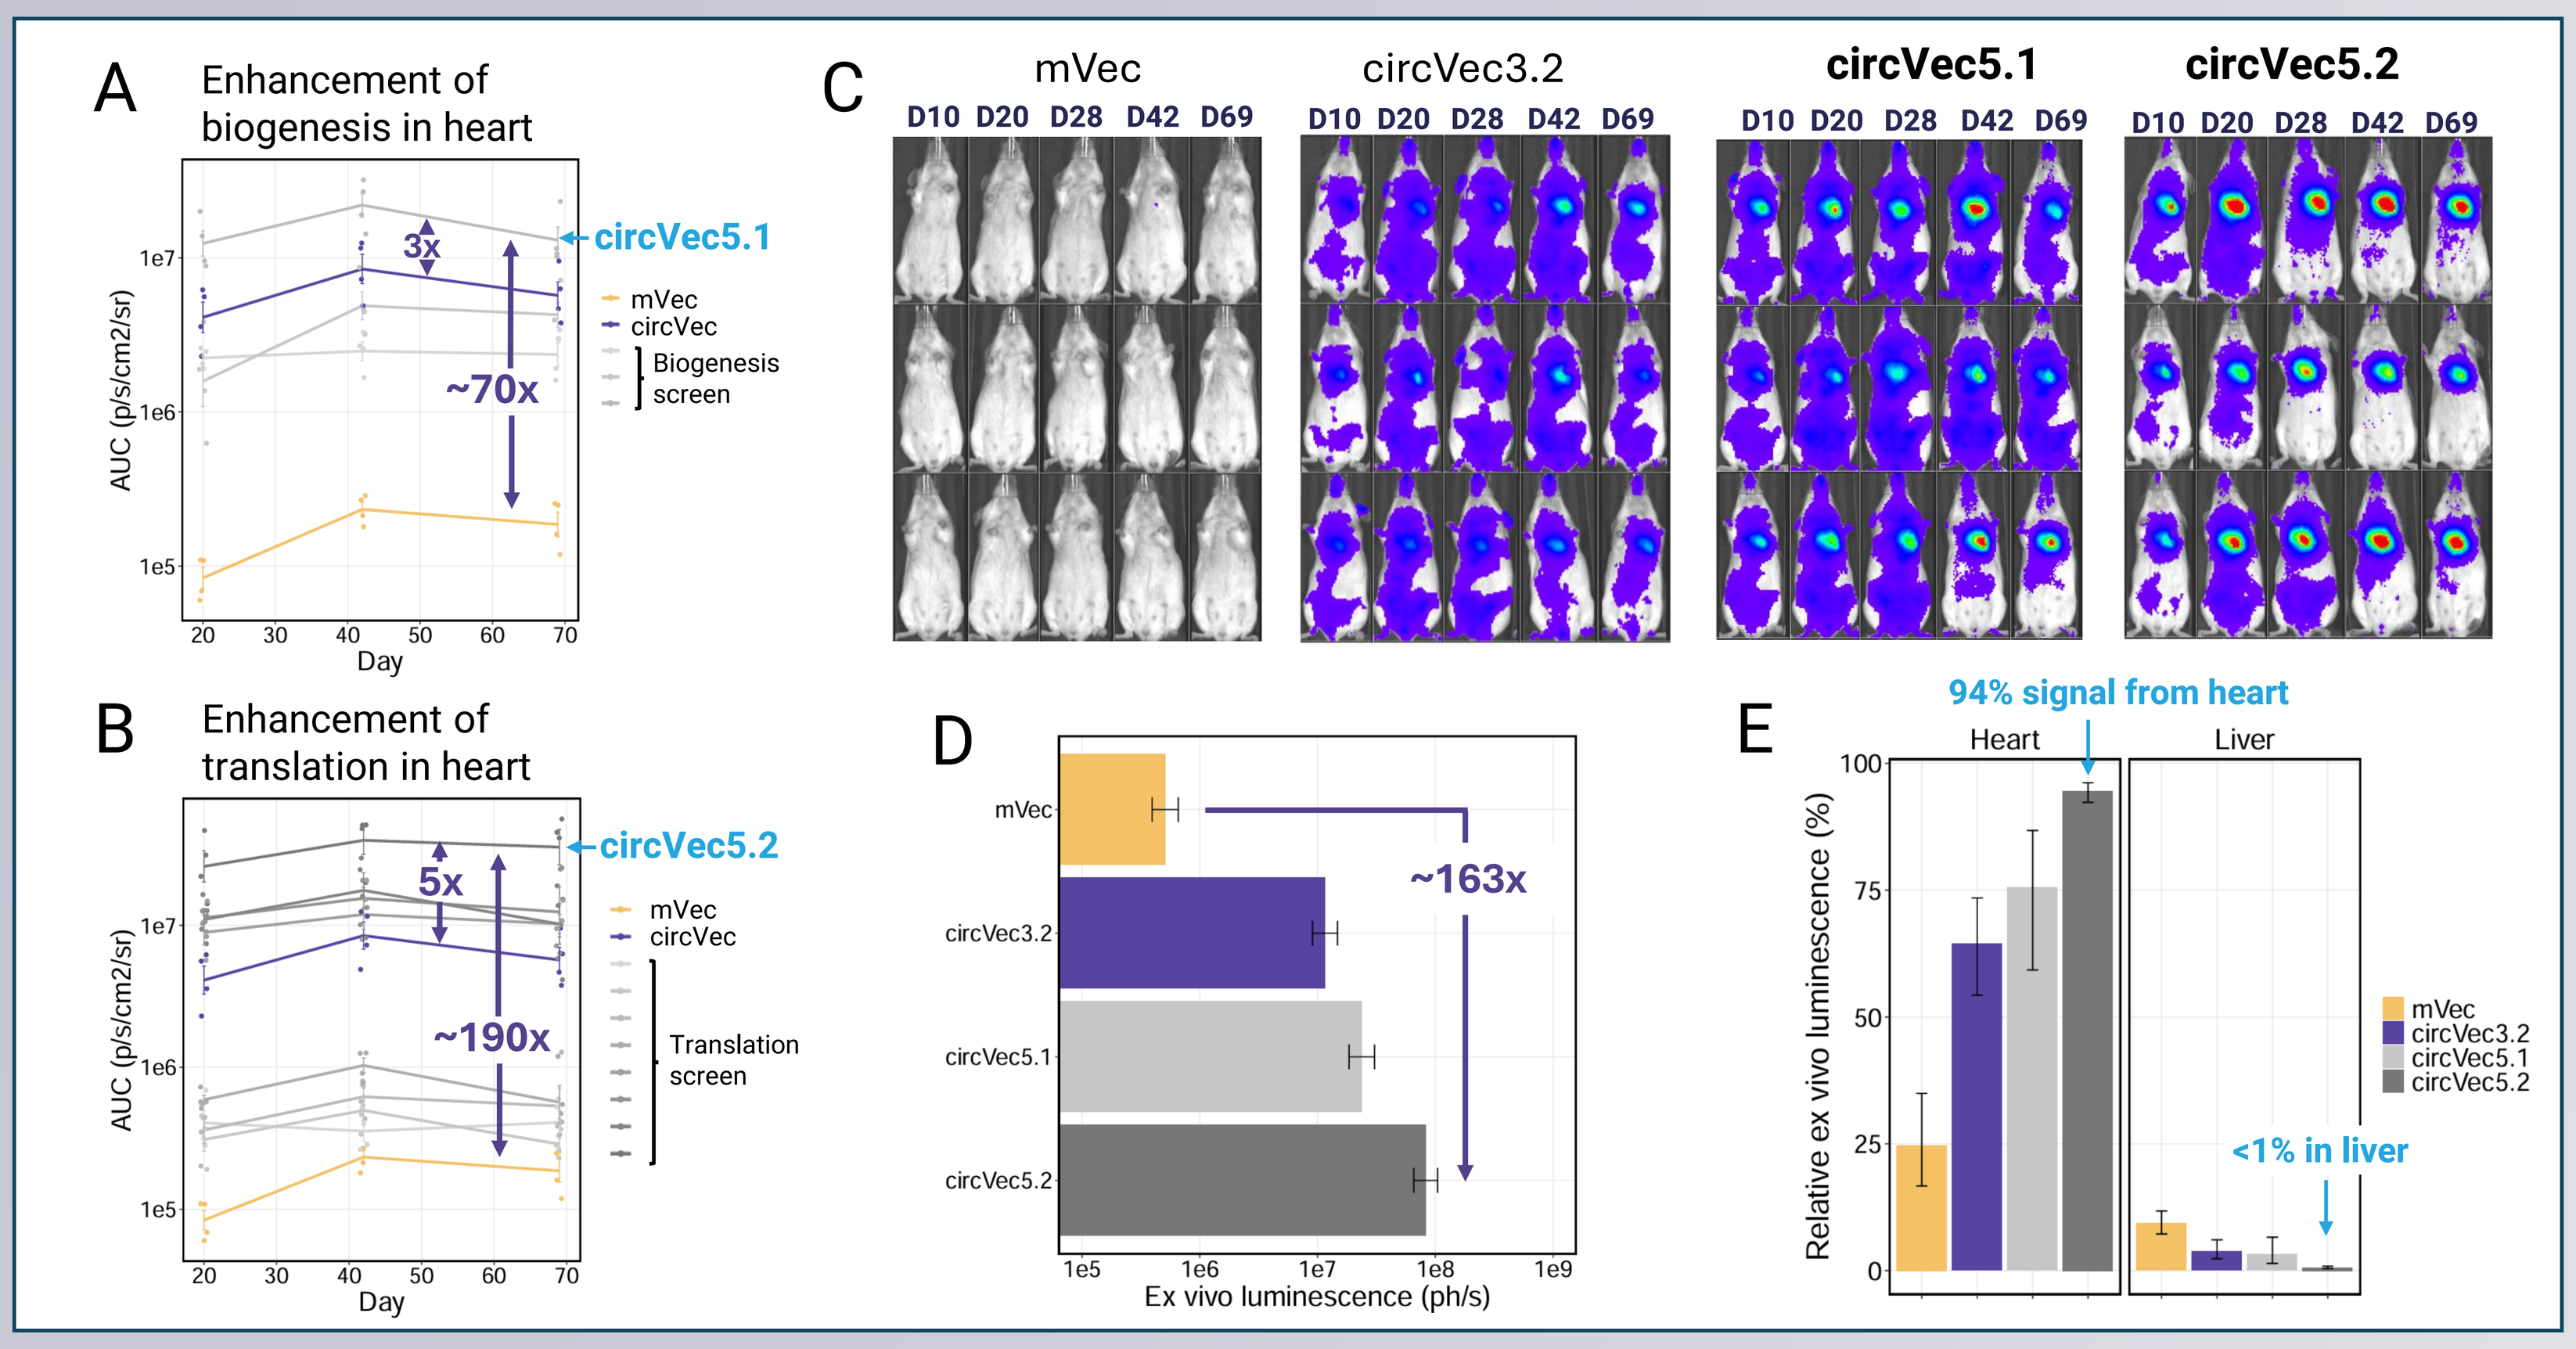
A Enhancement of biogenesis in heart
AUC (p/s/cm2/sr)
circVec5.1
3x
~70x
mVec
circVec
Biogenesis screen
Day
B Enhancement of translation in heart
circVec5.2
5x
~190x
Translation screen
C
mVec
circVec3.2
circVec5.1
circVec5.2
D10 D20 D28 D42 D69
D
~163x
Ex vivo luminescence (ph/s)
E
94% signal from heart
Heart
Liver
Relative ex vivo luminescence (%)
<1% in liver

Figure 5: Screening circVec design for enhanced biogenesis and translation in heart. A-B) Quantification of *in vivo* heart luminescence from an array of AAV9-circVec cassettes expressing firefly luciferase upon i.v. injection of AAV9 at dose 5e12vg/kg. The circVec cassettes aim to independently enhance biogenesis (A) or translation (B). The best performing circVecs are coined circVec5.1 and circVec5.2, respectively. **C)** Longitudinal IVIS imaging of luminescence for AAV9-mVec, AAV9-circVec3.2, AAV9-circVec5.1, and AAV9-circVec5.2. **D)** Ex vivo luminescence from heart at day 70 post transduction. **E)** Relative luminescence from heart and liver across all organs (heart, liver, diaphragm, ovaries, hindlegs, thyroid gland, spleen, intestine, and lung) as determined from ex vivo luminescence at day 70.

Circio pipeline

	Program (Technical concept → In vitro PoC → In vivo technical PoC → In vivo disease model → IND enabling)	Expression	Cross-correction	Stress	Off-target expression
Gene therapy circVec-AAV	**Genetic cardiomyopathy –** systemic delivery	↑	↑	↓	↓
	Ophthalmology - local delivery	↑	tbd	tbd	tbd
	CNS - local & systemic	↑	↑	tbd	tbd
In vivo CAR-T circVec DNA-LNP	**B-cell malignancies & autoimmunity** — Covered in Circio oral presentation SESSION 3d: Non-viral vectors/nanotechnology and RNA therapeutics I	↑	tbd	tbd	↓

Conclusions

Eoghan.oleary@circio.com

- **circVec is a platform technology to generate stable circRNA from viral and non-viral vectors:** circRNA accumulates intra-cellularly, leading to higher and more durable protein expression vs. standard linear-mRNA-based vectors
- **AAV-circVec outperforms conventional mRNA-based AAV by up to 60× across eye, heart and CNS in vivo**: the advantage is driven by increased RNA stability and transcript accumulation, with no change in AAV transduction
- **New heart-optimized circVec 5.2 design achieves significantly higher on target gene expression** than conventional mRNA-based AAV9, with 94% heart-specific expression and less than 1% off-target liver expression.
- **Next steps**: testing circVec in genetic heart disease models, single-cell analysis for eye & CNS, validation in NHPs

Get PDF using the QR code (do not distribute without permission)