Circular RNA-based expression enhances AAV potency in heart, whilst minimizing dose and cellular stress

PosterID: P0604
Presented at ESGCT 2026

O'Leary ET[1], Zhang J[1], Warsame M[1], Stefanelli S[1], Akter F[1], An Y[1], Liu J[1], Subramanian N[1], Melin O[1], Wiklund ED[1], Levitsky V[1], Hansen TB[1]

1. Circio AB, Huddinge, Sweden

circVec technology introduction

Circular RNA (circRNA) is a novel class of endogenously expressed RNA. CircRNAs are generated by a non-linear splicing event, known as **backsplicing**, where an upstream splice acceptor attacks a downstream splice donor. circRNAs **are resistant to exonucleolytic decay**, which results in **increased intra-cellular stability and persistence compared to mRNA**. Circio has developed a circRNA-based gene expression platform, **circVec**, which utilizes this natural stability advantage to **increase and prolong protein expression from viral and non-viral vectors**

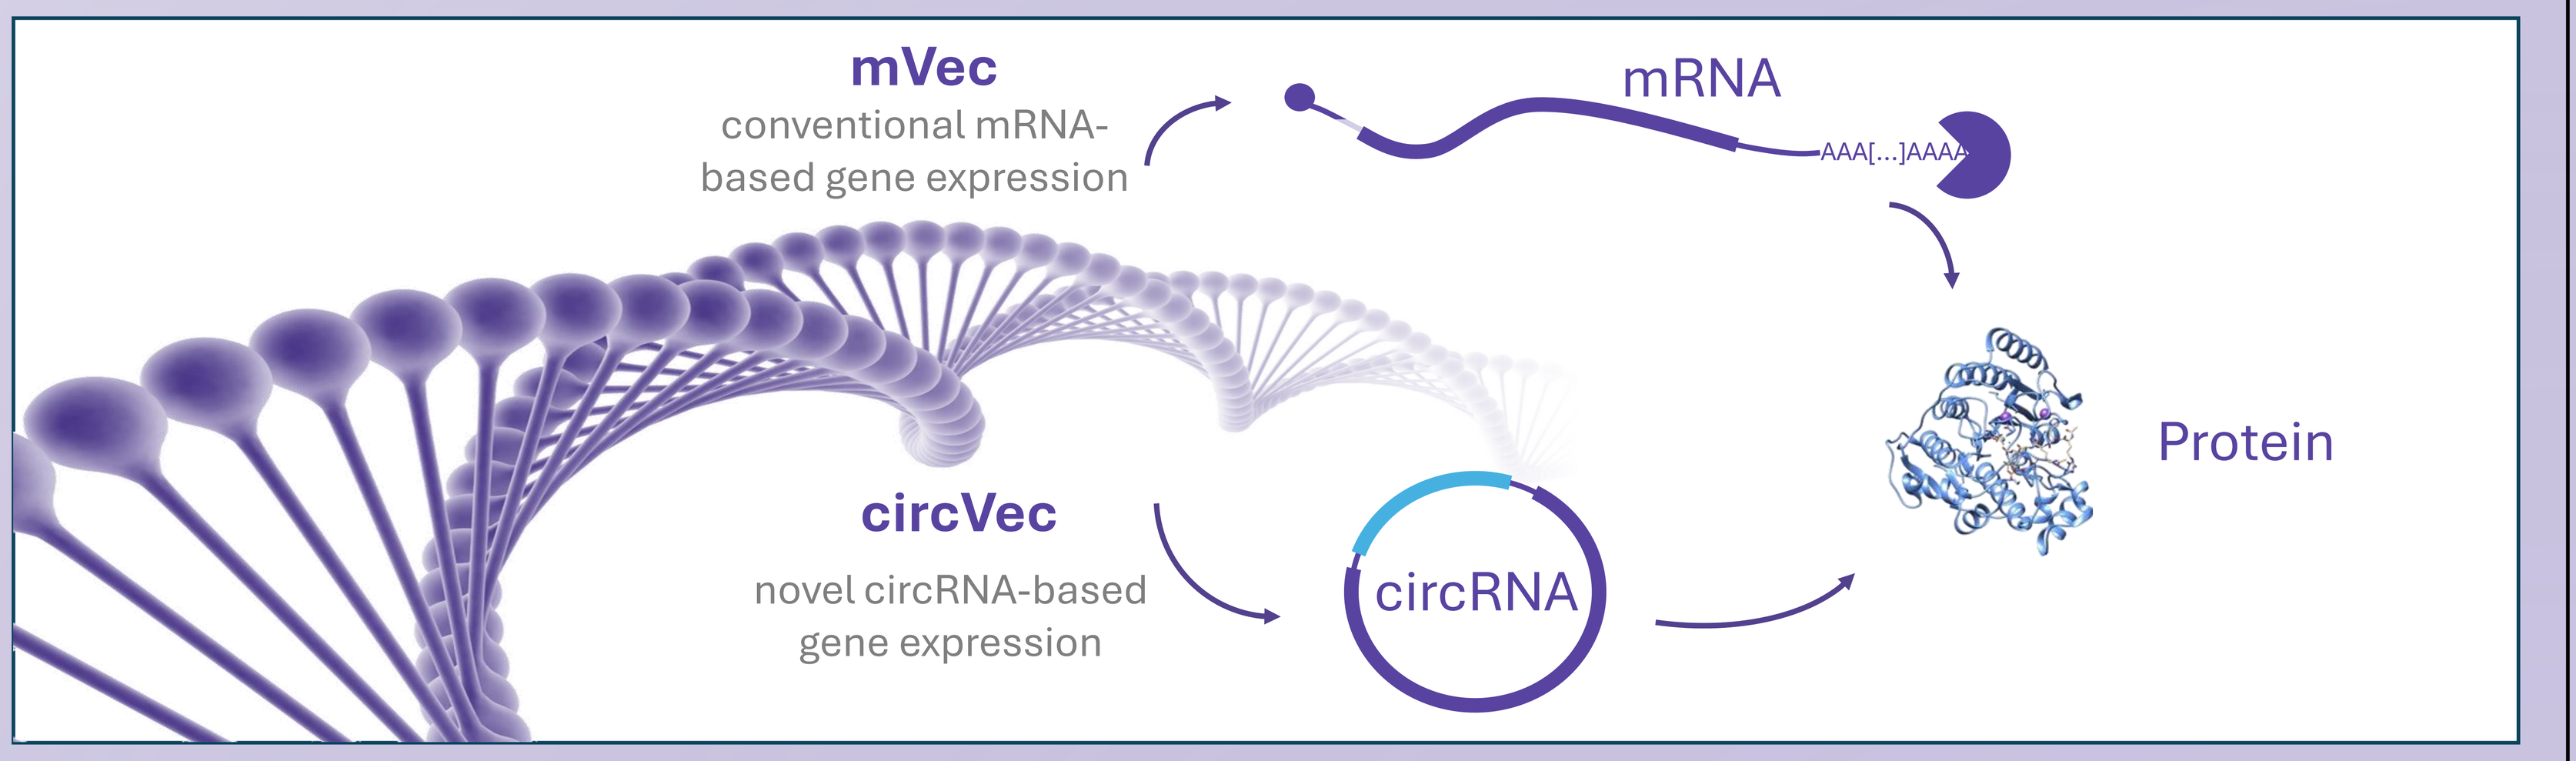
mVec
conventional mRNA-based gene expression
mRNA
AAA[...]AAAA
Protein
circVec
novel circRNA-based gene expression
circRNA

1 Optimized circVec architecture improves circRNA gene expression by ~36-fold

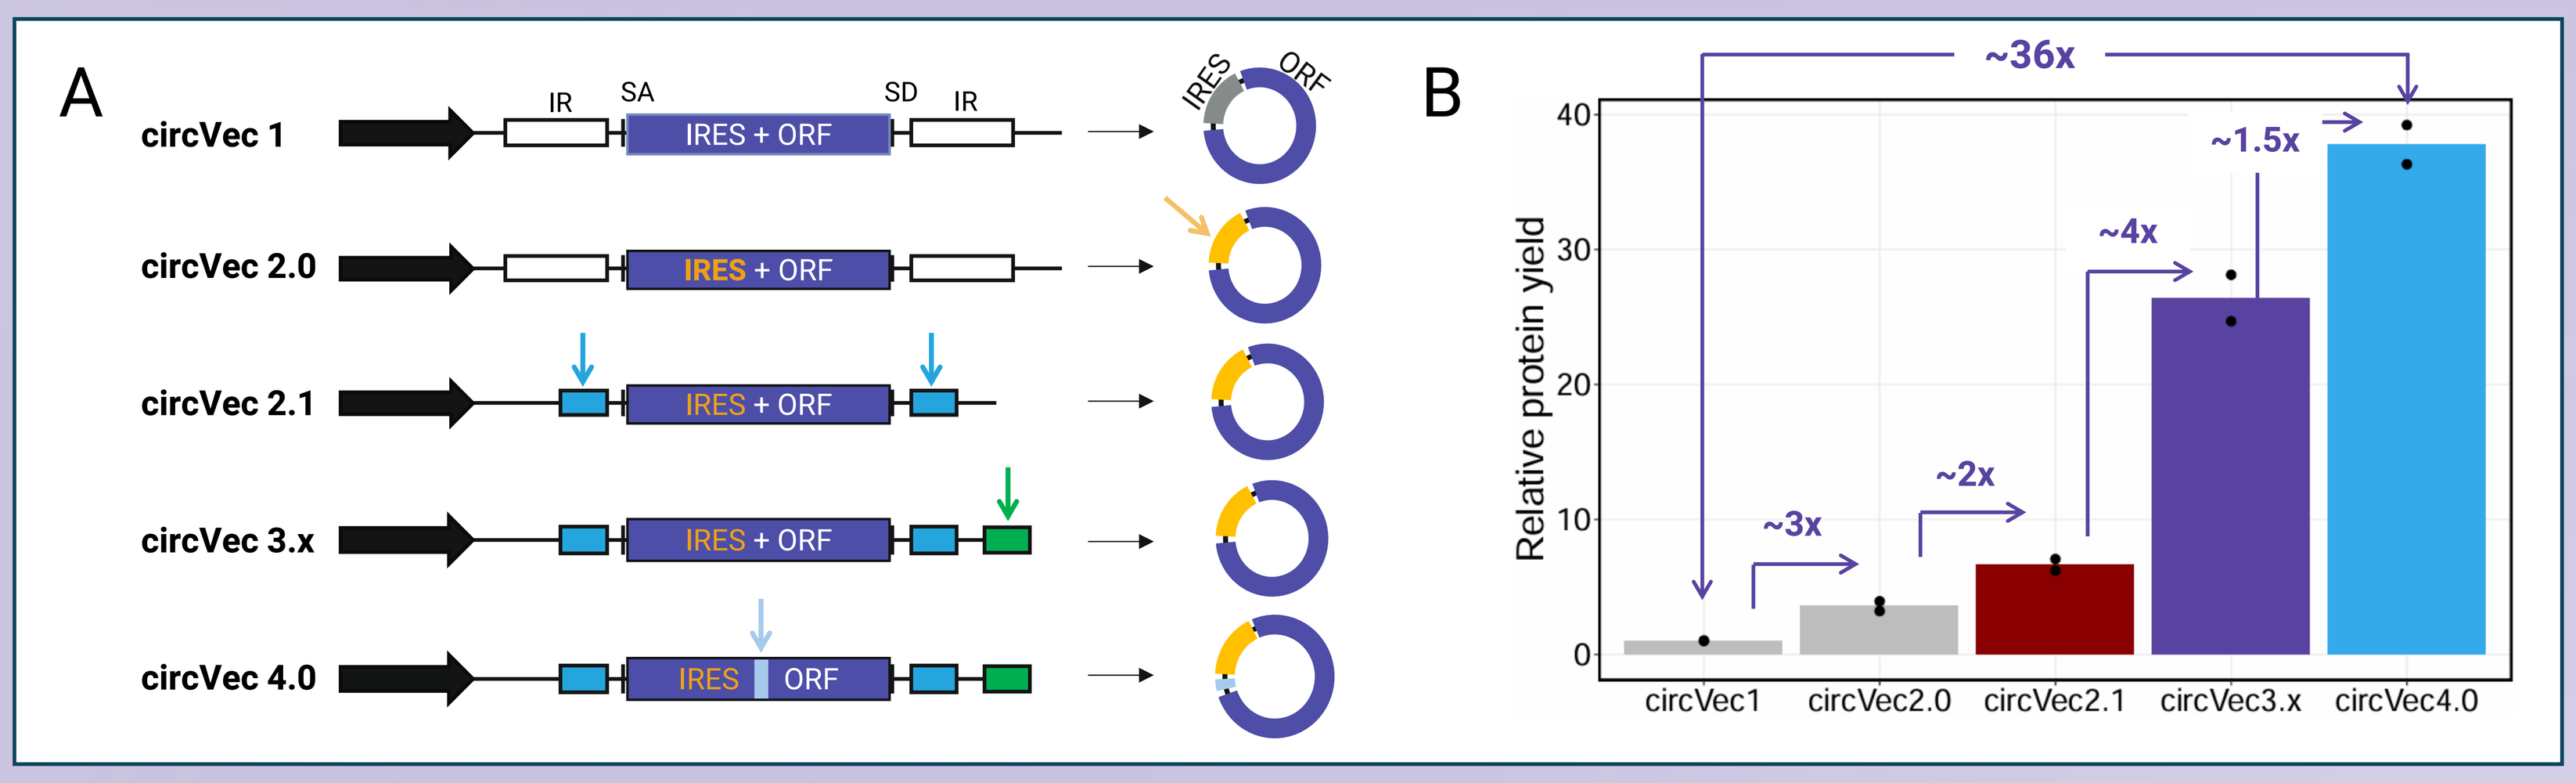
A
circVec 1
circVec 2.0
circVec 2.1
circVec 3.x
circVec 4.0
IR
SA
SD
IRES + ORF
IRES
ORF
B
Relative protein yield
~36x
~1.5x
~4x
~2x
~3x
circVec1
circVec2.0
circVec2.1
circVec3.x
circVec4.0

Figure 1: circVec genetic construct design and platform evolution. A) Schematic representation of circVec conctruct design at RNA and DNA level. **B)** Relative protein yield from circVec generation 1-4 in HepG2 cells (n=2).

2 circVec increases AAV9 gene expression by over 30-fold compared to benchmark AAV

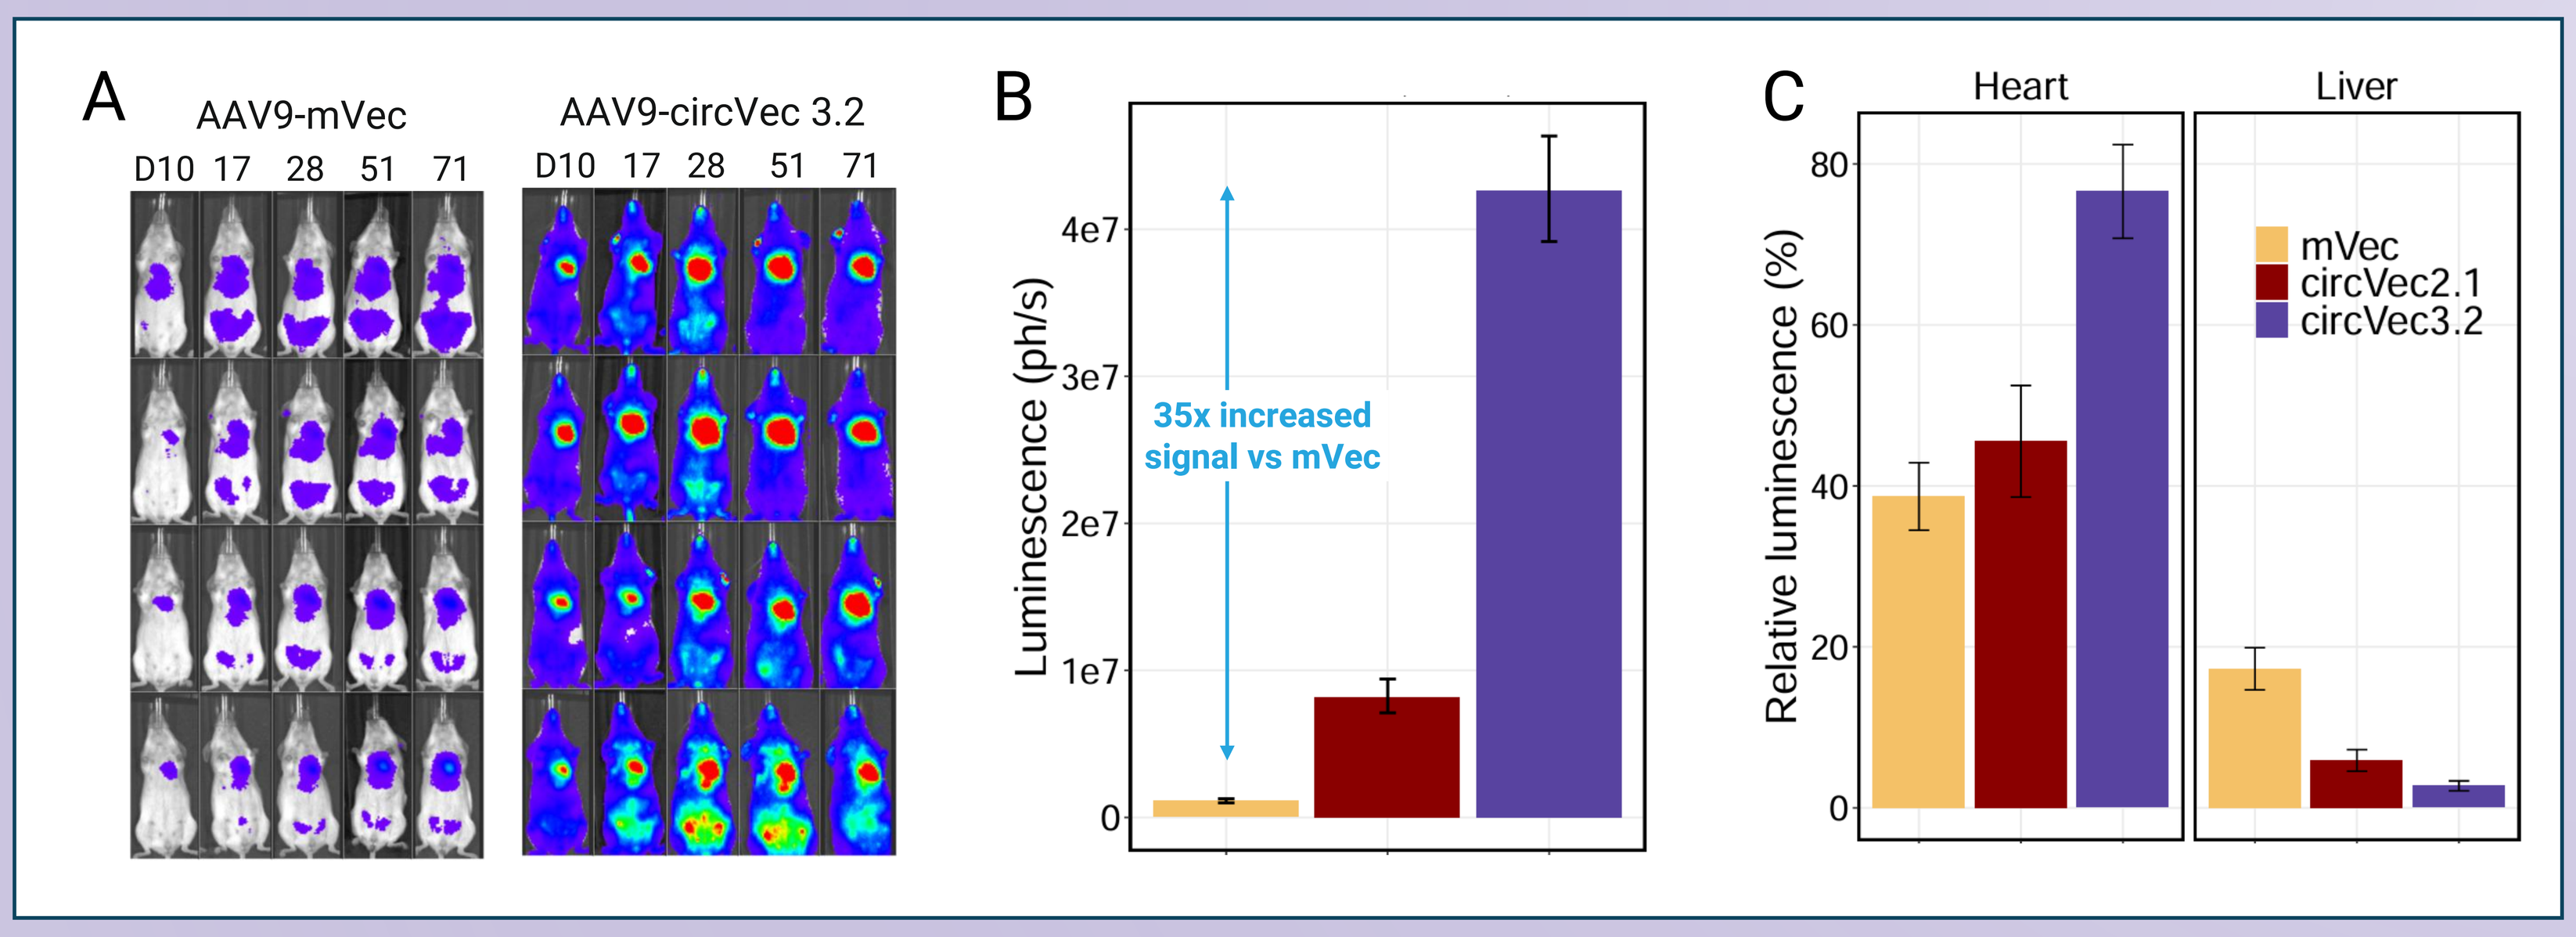
A
AAV9-mVec
AAV9-circVec 3.2
D10 17 28 51 71
B
Luminescence (ph/s)
35x increased signal vs mVec
C
Heart
Liver
Relative luminescence (%)
mVec
circVec2.1
circVec3.2

Figure 2: In vivo performance of AAV9-circVec3.2 with heart-specific promoter (cTnT). A) Longitudinal IVIS analysis at different timepoints after i.v. injection of different AAV9 vectors (5e12 vg/kg) in female BALB/c mice (n=4). **B)** Absolute ex vivo luminescence from heart 72-days post injection shown as geometric mean + s.e.m. **C)** Relative luminescence from heart and liver as determined from ex vivo luminescence at day 72.

3 Circular RNA accumulation drives enhanced gene expression and reduces cellular stress response

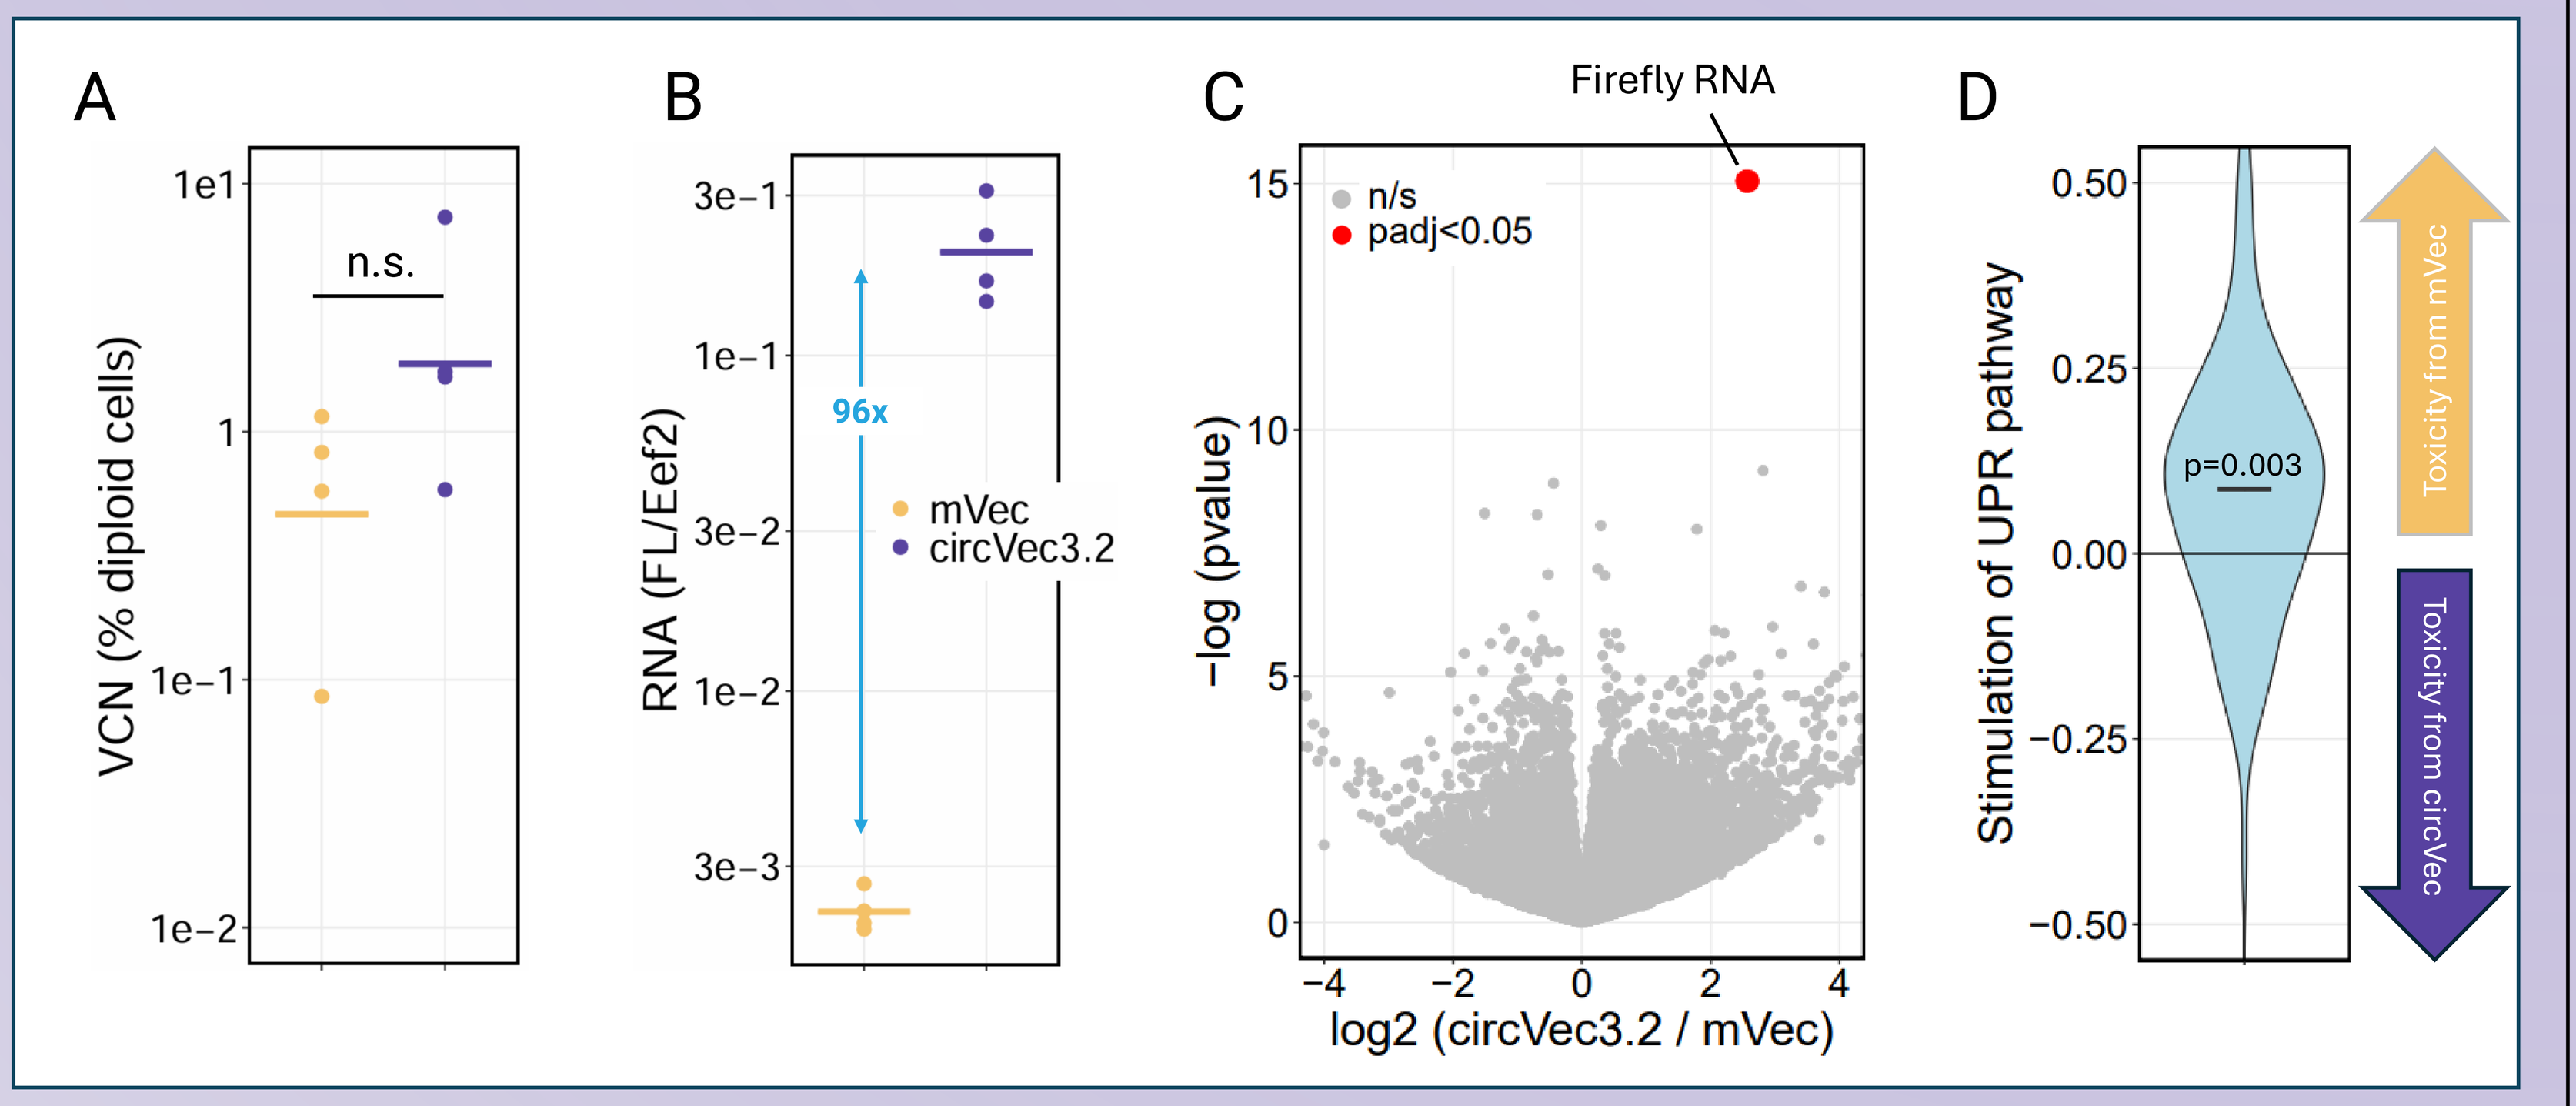
A
VCN (% diploid cells)
n.s.
B
RNA (FL/Eef2)
96x
mVec
circVec3.2
C
Firefly RNA
n/s
padj<0.05
−log (pvalue)
log2 (circVec3.2 / mVec)
D
Stimulation of UPR pathway
p=0.003
Toxicity from mVec
Toxicity from circVec

Figure 3: Viral genome and transcriptomic analysis on heart samples. A-B) AAV genome (A) and Firefly RNA (B) quantification on heart tissue 92 days after i.v. injection of 5e12vg/kg AAV9-circVec3.2 or AAV9-mVec, as denoted. T-test on log-transformed values is used to determine significance, n.s.: not significant. **C)** Volcano plot based on whole transcriptomic analysis of heart tissue (n=4/group). Only Firefly RNA shows a significant change in expression (red dot, BH-adjusted p-value (padj) = 0.01). **D)** Unfolded protein response (UPR) genes as a class are significantly upregulated in mVec heart tissue compared to circVec (p=0.003, Wilcoxon rank sum test).

4 RNAscope analysis shows 80% circRNA positive cardiomyocytes at 5e12 vg/kg dose

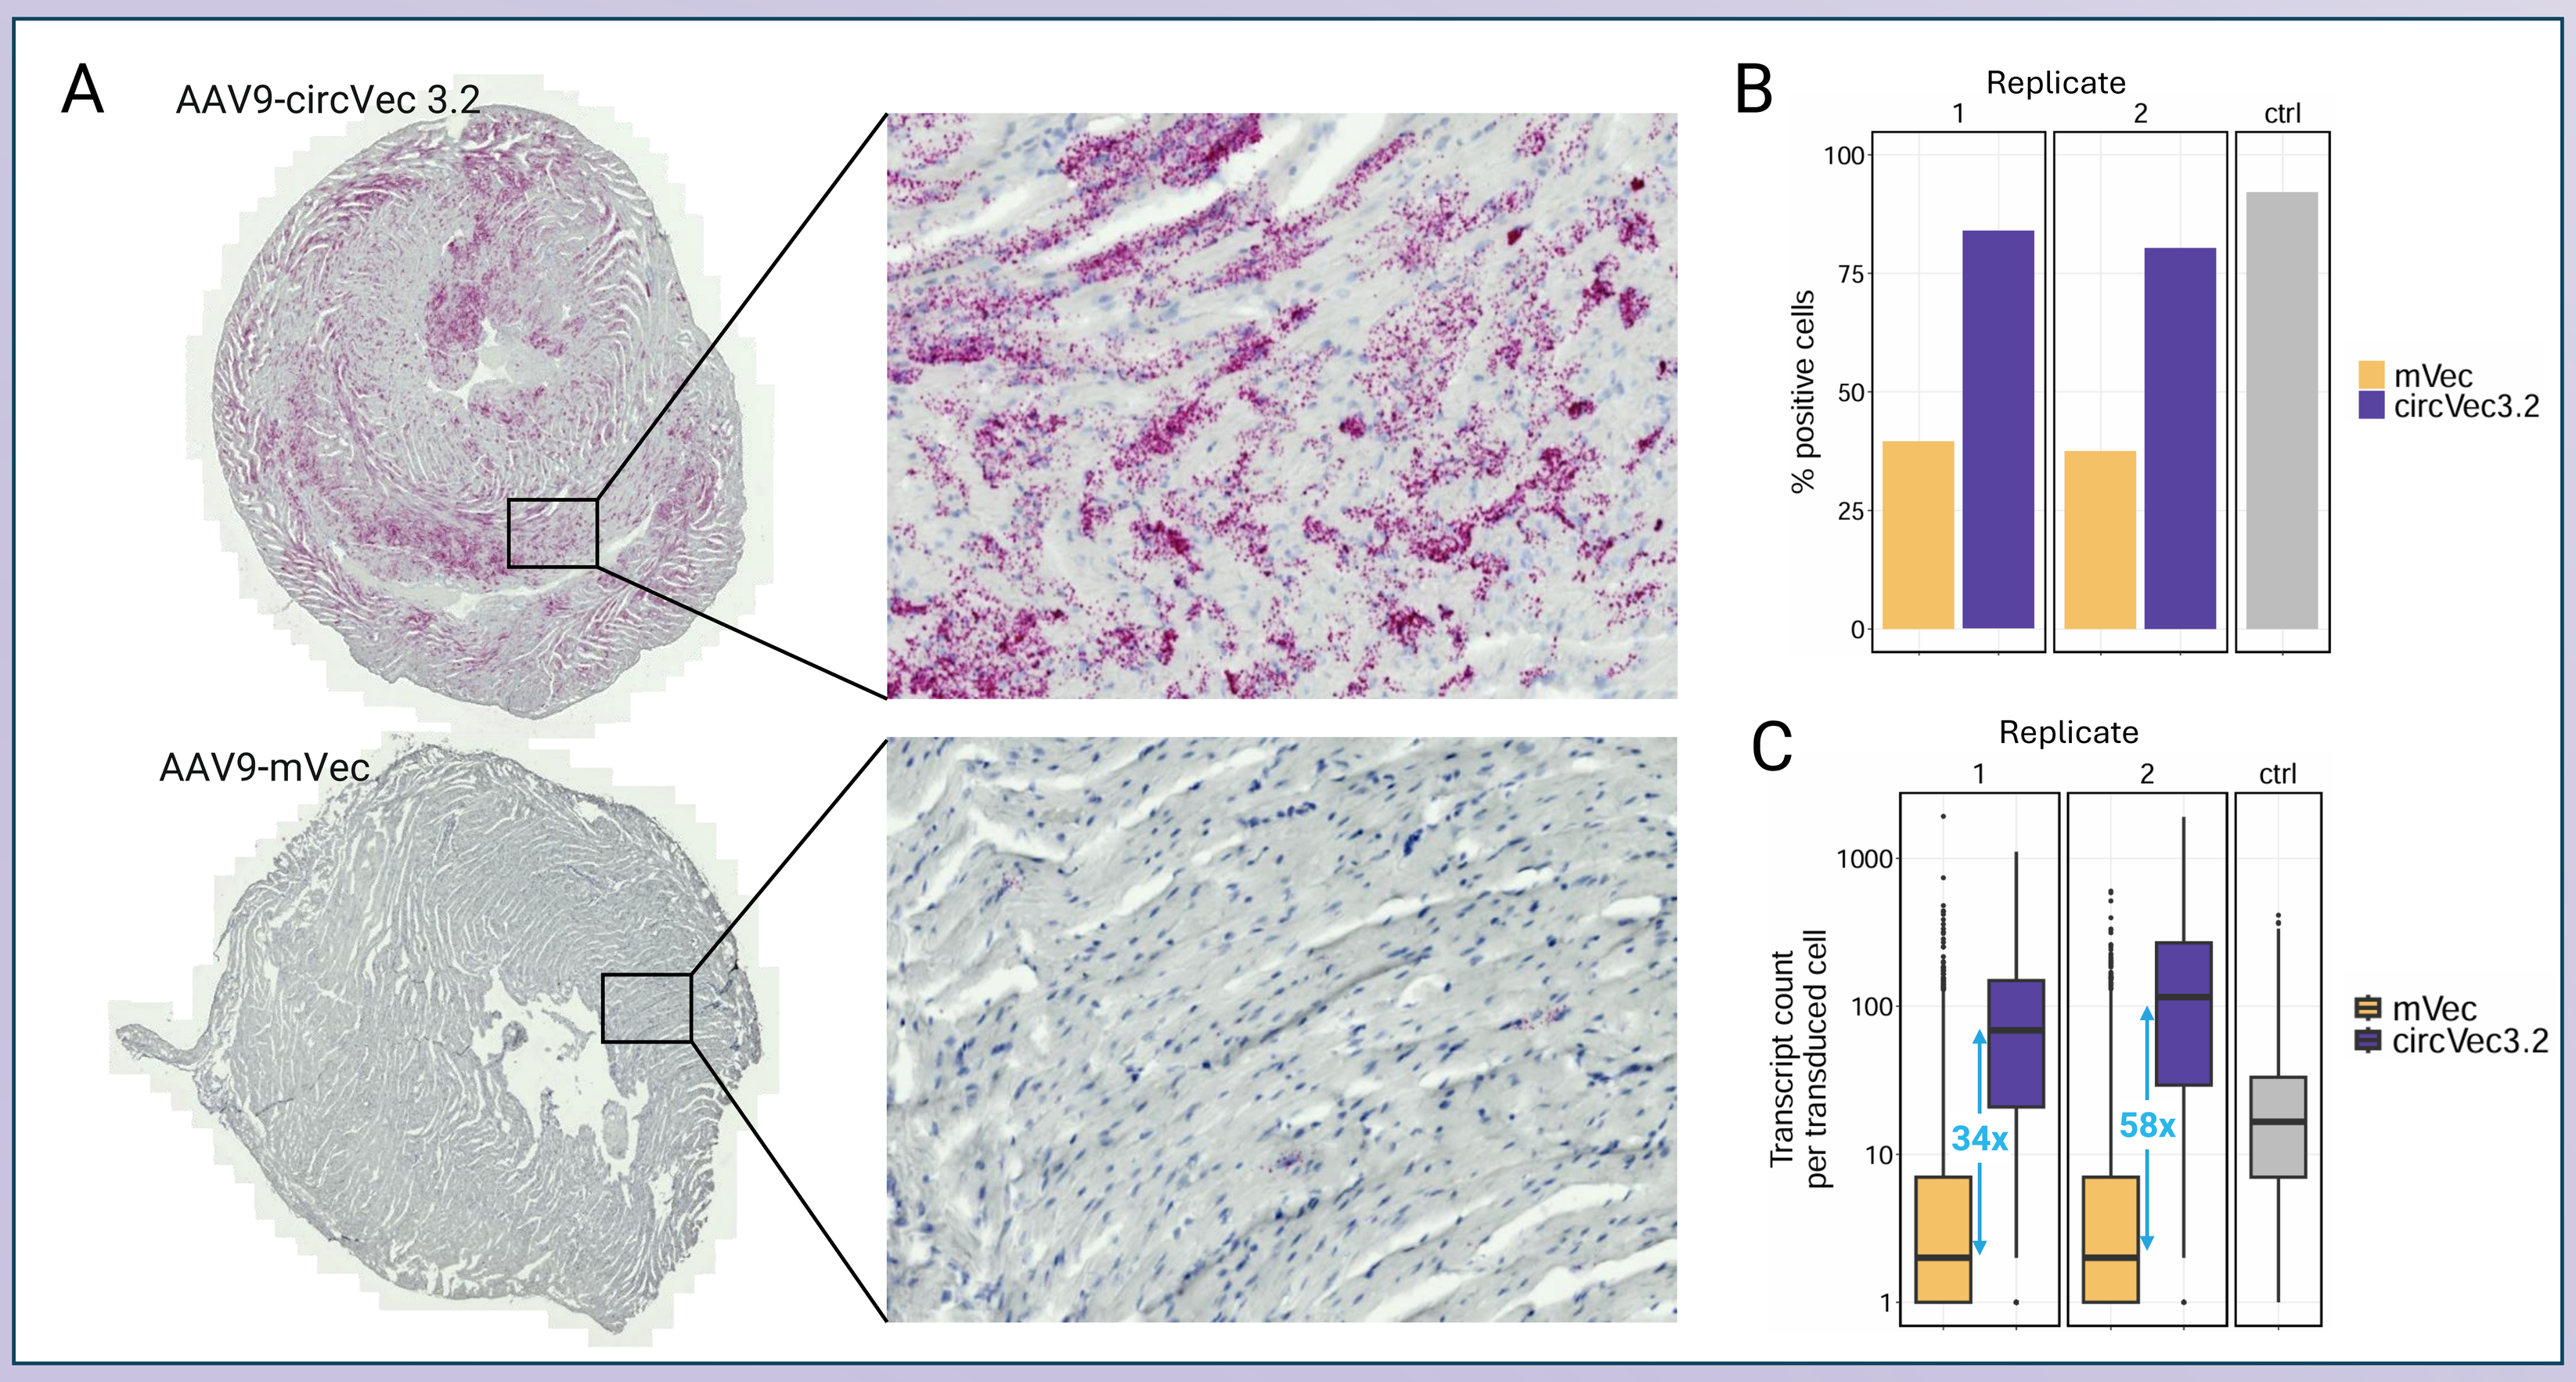
A
AAV9-circVec 3.2
AAV9-mVec
B
Replicate
1
2
ctrl
% positive cells
mVec
circVec3.2
C
Transcript count per transduced cell
34x
58x

Figure 4: RNAscope analysis on mouse heart subjected to intravenous injection of AAV9-circVec and AAV9-mVec (5e12vg/kg). A) Representative heart sections from AAV9-circVec3.2 or AAV9-mVec treated mice (as denoted) probed for Firefly RNA (violet) using RNAscope technology and strained with Dapi (blue). **B)** Using the QuPath software, number of positive cells were extracted. *Ppib* (peptidyl prolyl isomerase B) is used as the positive control marker (ctrl). **C)** Distribution of Firefly transcript copy-numbers in positive cells showing 34x-58x more RNA from circVec3.2 compared to mVec.

5 Systemic delivery of AAV9-circVec shows ~10–20x dose-sparing potential in heart

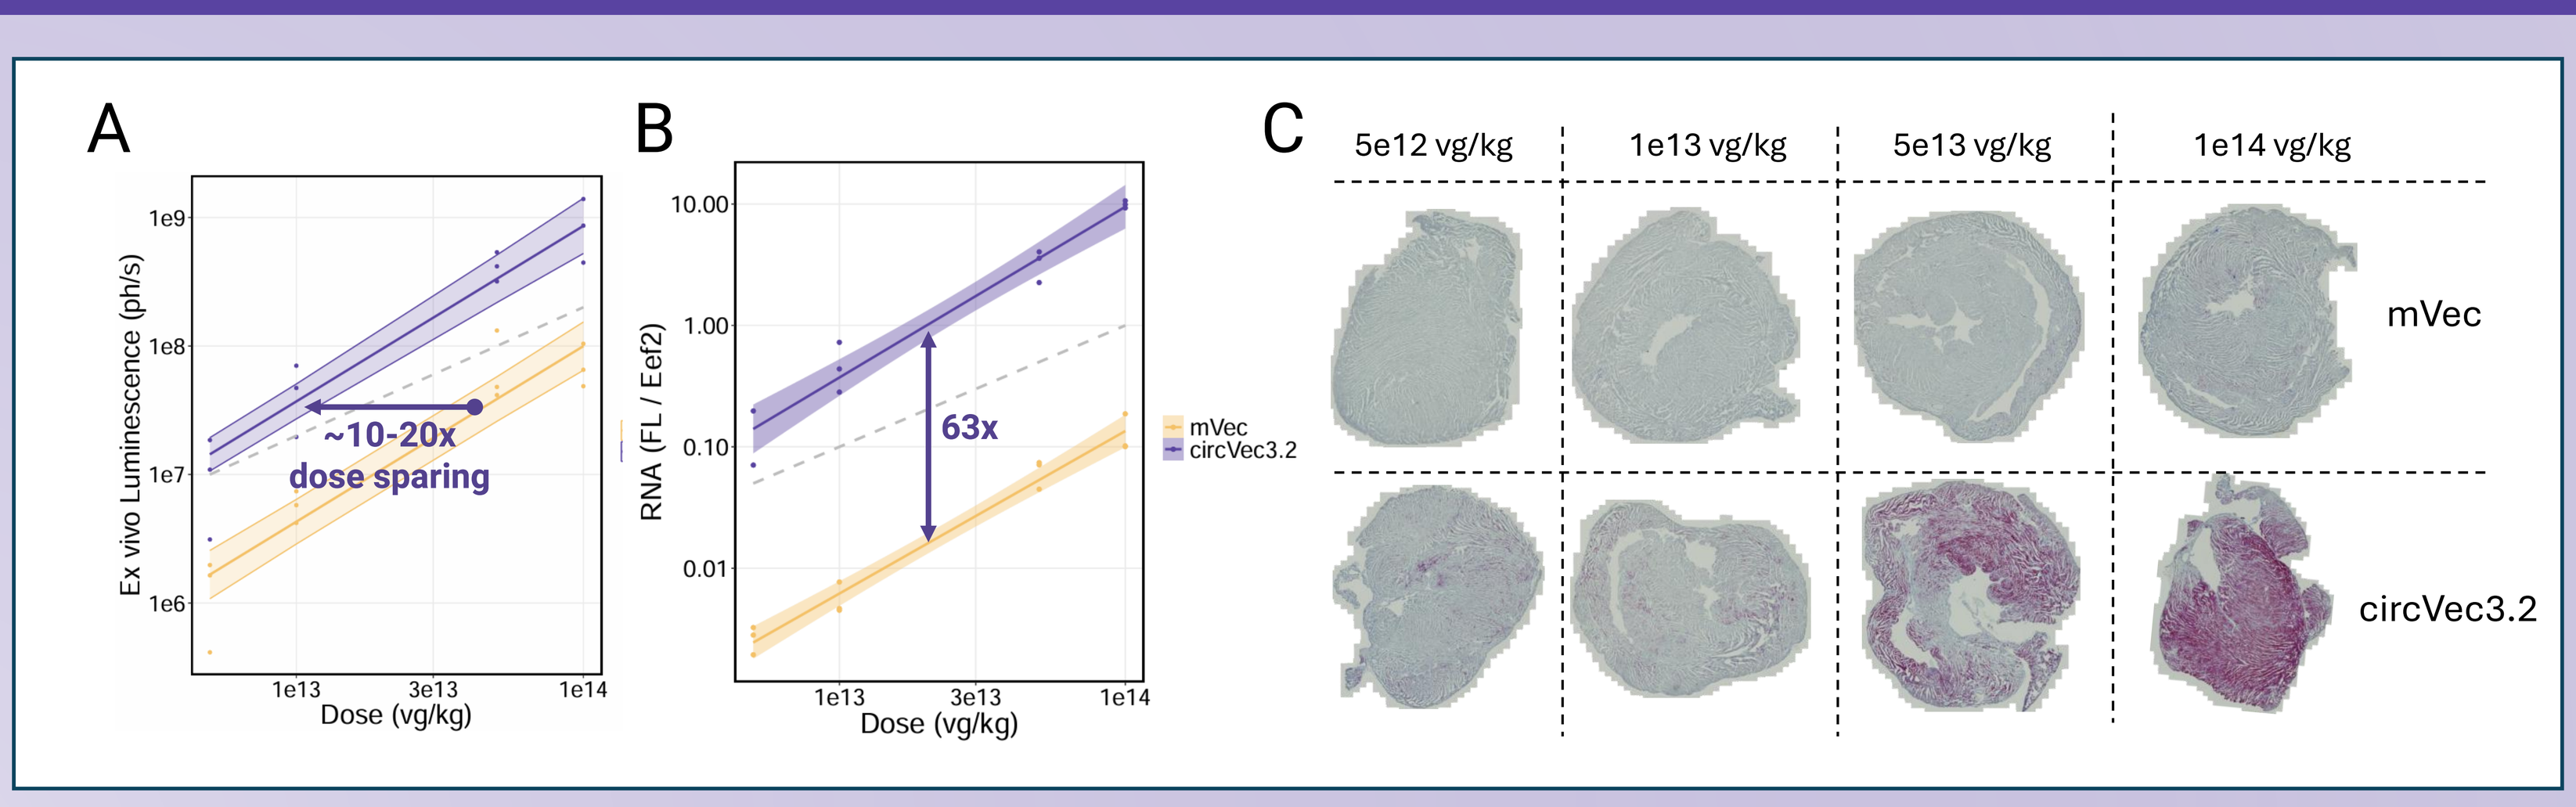
A
Ex vivo Luminescence (ph/s)
~10-20x
dose sparing
Dose (vg/kg)
B
RNA (FL / Eef2)
63x
mVec
circVec3.2
C
5e12 vg/kg
1e13 vg/kg
5e13 vg/kg
1e14 vg/kg
mVec
circVec3.2

Figure 5: Dose-response relationship for AAV9-circVec and AAV9-mVec in heart. A-B) Super-linear dose response curve on ex vivo luminescence (A) and Firefly RNA quantities (B) from heart 54 days after injection of AAV9-mVec or AAV9-circVec3.2 using 4 different dose levels: 5e12, 1e13, 5e13, and 1e14 vg/kg. Dashed line represents linear dose-response with a slope of 1. **C)** RNAscope images on representative hearts form mice subjected to the different doses of AAV9-mVec or AAV-circVec as denoted.

6 AAV-circVec advantage also applies to a therapeutically relevant gene (LAMP2) *in vivo*

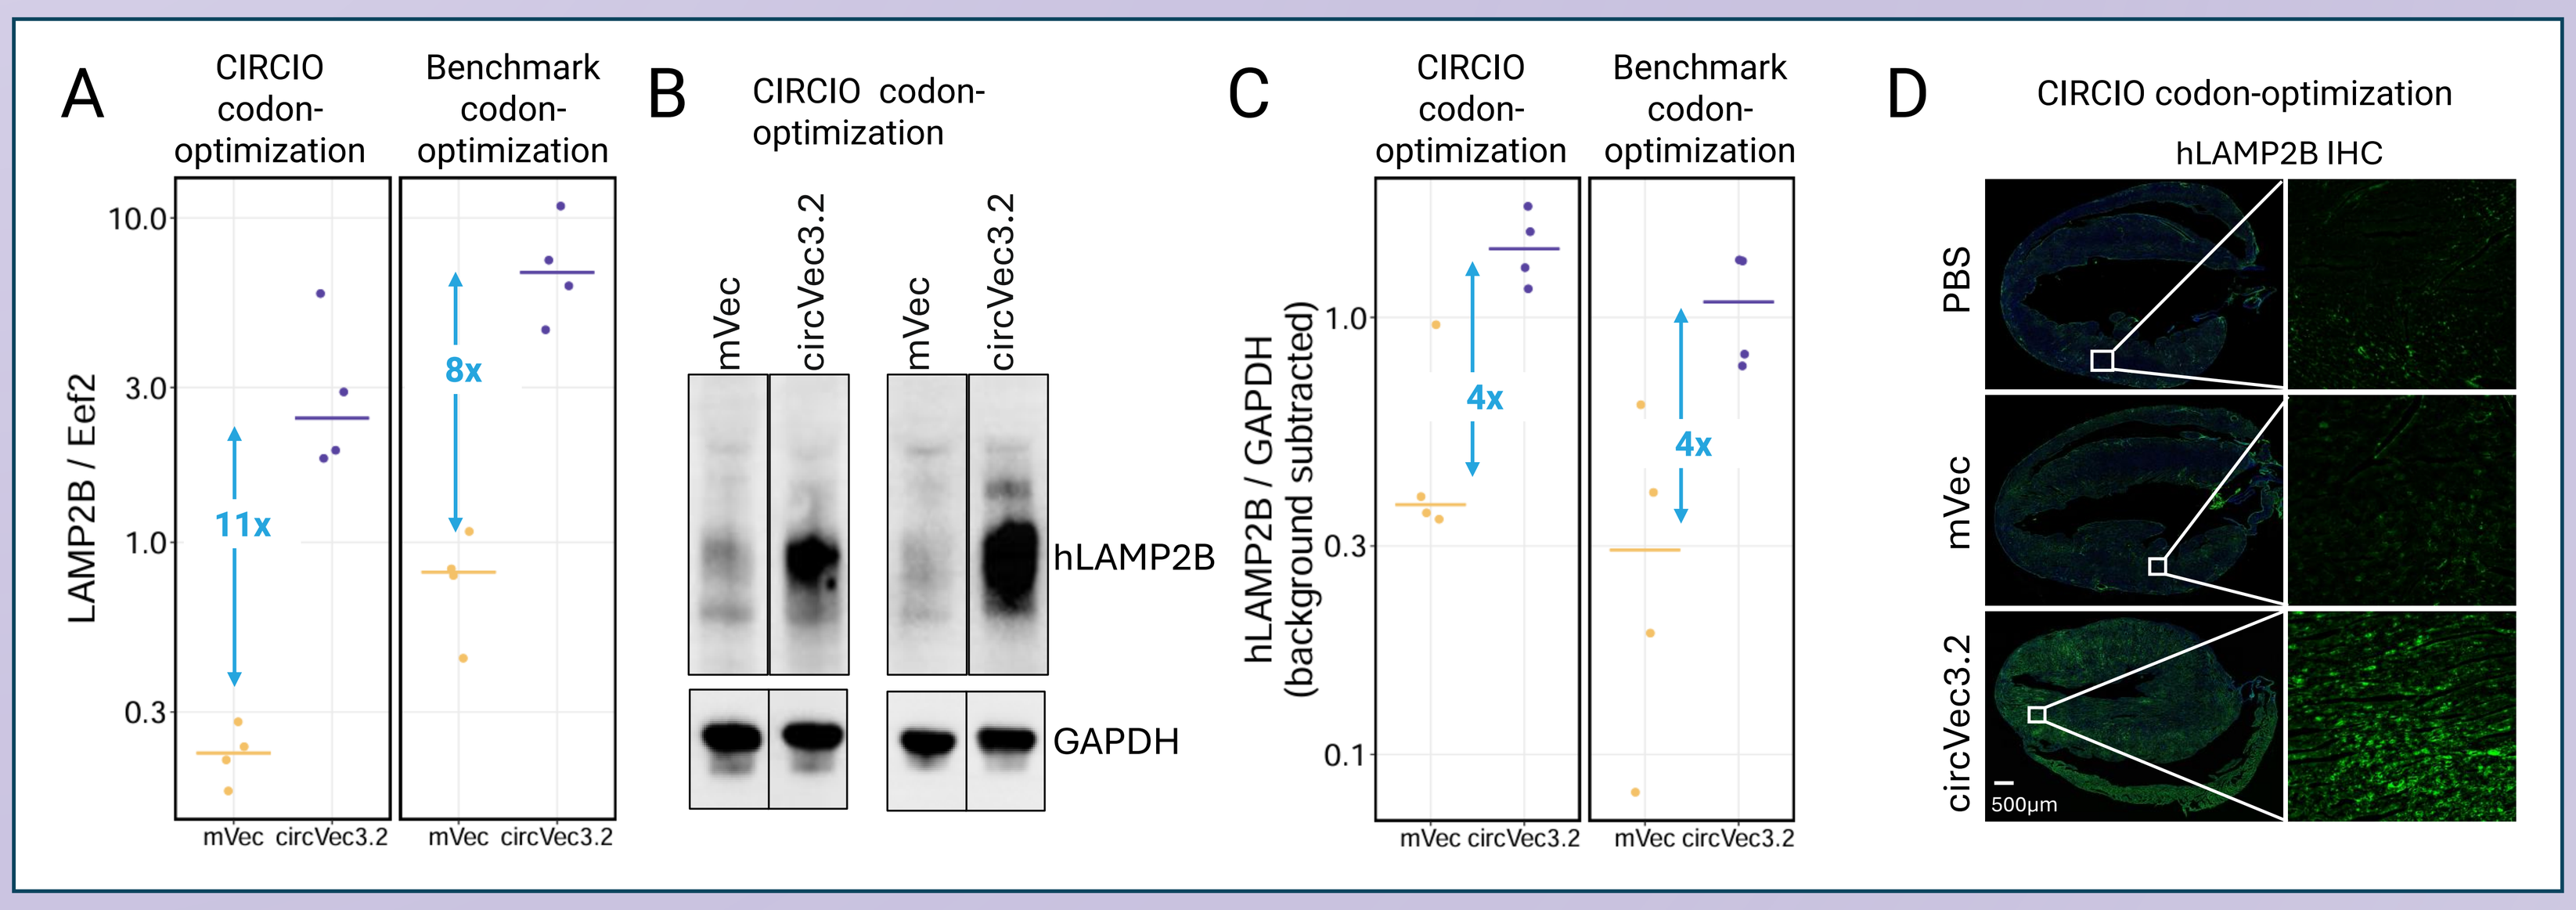
A
CIRCIO codon-optimization
Benchmark codon-optimization
LAMP2B / Eef2
11x
8x
mVec circVec3.2
B
CIRCIO codon-optimization
mVec
circVec3.2
hLAMP2B
GAPDH
C
hLAMP2B / GAPDH (background subtracted)
4x
D
CIRCIO codon-optimization
hLAMP2B IHC
PBS
mVec
circVec3.2
500µm

Figure 6: Comparison of hLAMP2B expression in heart from AAV9-circVec3.2 and AAV9-mVec using 5e13vg/kg. A) RNA quantification by RT-qPCR on heart samples 42 days after injection of CIRCIO and industry benchmark codon-optimized hLAMP2B expression vectors (AAV9-circVec3.2 and AAV9-mVec) with a heart specific promoter. **B-C)** Representative western blots (each mVec/circVec pair cropped from same blot, B) and quantification of relative hLAMP2B protein expression in heart (C). **D)** IHC analysis of hLAMP2B protein expression in mouse heart 42 days post AAV injection.

7 Circio pipeline

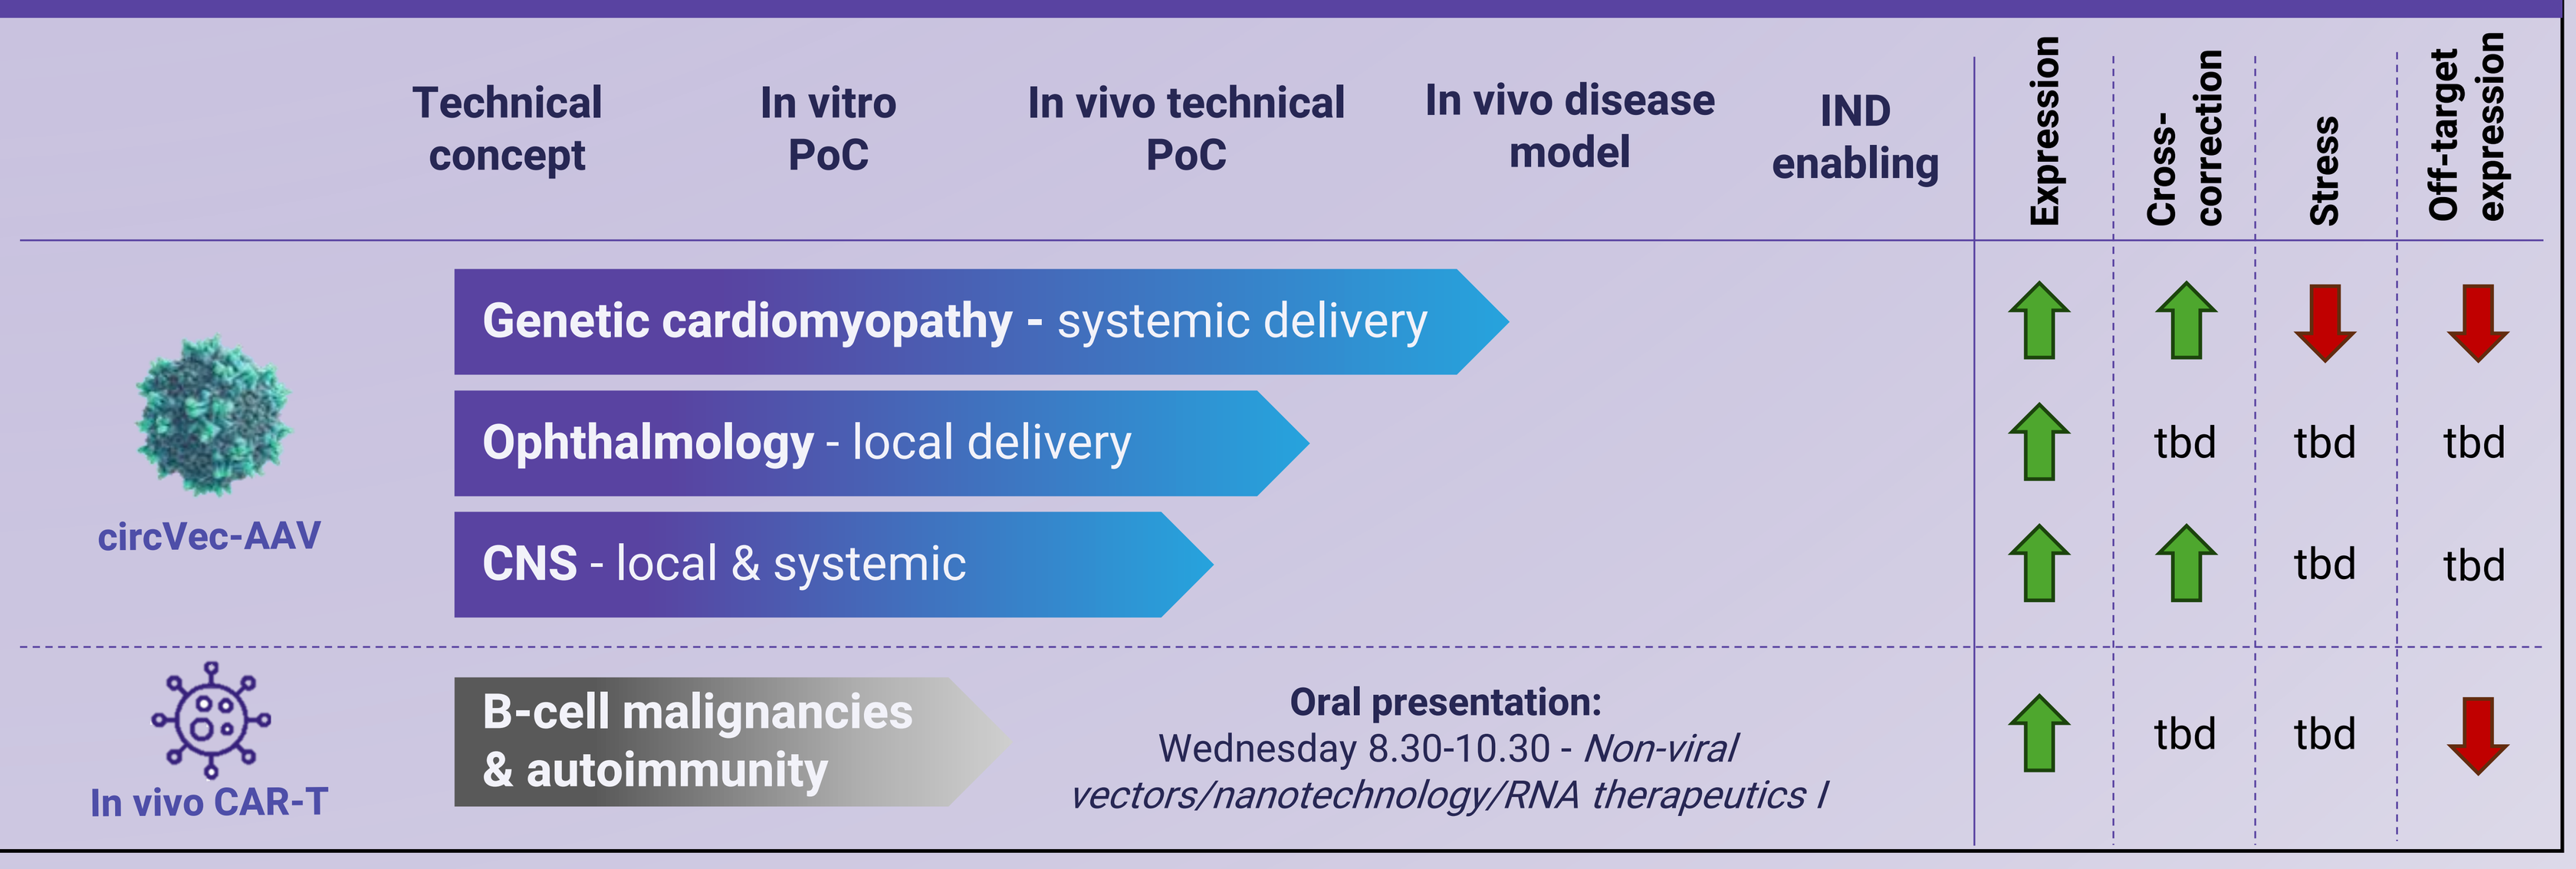
Technical concept
In vitro PoC
In vivo technical PoC
In vivo disease model
IND enabling
Expression
Cross-correction
Stress
Off-target expression
circVec-AAV
Genetic cardiomyopathy - systemic delivery
Ophthalmology - local delivery
tbd
tbd
tbd
CNS - local & systemic
tbd
tbd
In vivo CAR-T
B-cell malignancies & autoimmunity
Oral presentation:
Wednesday 8.30-10.30 - Non-viral vectors/nanotechnology/RNA therapeutics I
tbd
tbd

Conclusions

thomas.hansen@circio.com

- **40x enhanced protein expression in heart from AAV9-circVec:** Prolonged circRNA stability leads to accumulation of RNA and enhanced protein expression compared to conventional mRNA-expressing AAV
- **Reduced stress response and off-target liver expression from circVec**: Significantly lower activation of UPR pathway by as well as lower reduced off-target expression adds to the safety and specificity of circVec-based gene therapy in heart
- **80% circRNA penetrance in heart using low dose (5e12vg/kg) i.v. administration:** RNAscope analysis shows higher and broader expression of circRNA compared to mRNA from equivalent vector copy numbers in heart
- **Superior LAMP2B expression from circVec:** Using two different codon-optimized hLAMP2B ORFs, circVec significantly outperforms mVec on RNA and protein levels in wild-type mice

Get PDF using the QR code (do not distribute without permission)