A pilot study of Engineered Adenovirus ONCOS-102 in combination with pembrolizumab (pembro) in checkpoint inhibitor refractory advanced or unresectable melanoma

Alexander Shoushtari [1], Anthony J. Olszanski [2], Marta Nyakas [3], Thomas J. Hornyak [4], Jedd D. Wolchok [1], Victor Levitsky [5], Anne-Sophie Møller [5], Lukasz Kuryk [5], Karianne Risberg Handeland [5], Magnus Jäderberg [5]

[1] Memorial Sloan Kettering Cancer Center, New York, USA, [2] Fox Chase Cancer Center, Philadelphia, USA, [3] Department of Clinical Cancer Research, Oslo University Hospital, Oslo, Norway
[4] Department of Dermatology and University of Maryland Greenebaum Comprehensive Cancer Center, University of Maryland School of Medicine, Baltimore, USA
[5] Research & Development, Targovax ASA, Oslo, Norway

Poster number 1083P

INTRODUCTION

Although PD-1 and CTLA-4 blockade have led to broad improvements in prognosis for unresectable stage 3 and stage 4 melanomas most patients continue to suffer from disease progression and over half of patients will still die of their disease. PD-1 resistant melanomas remain challenging to treat and rational treatment strategies are still needed for patients with melanomas that progress despite PD-1 blockade.

Oncolytic viruses are immunomodulating agents with complementary mechanisms of antitumor efficacy to PD-1 blockade in their ability to promote tumor immune surveillance. Pre-clinically, ONCOS-102 (Ad5/3-D24-GM-CSF) and pembrolizumab have been shown to act ~~be~~ synergistically in a melanoma model. Clinically, both induction of innate and adaptive immune responses have been seen in various solid tumors.

We show that combining ONCOS-102 with PD-1 blockade is safe and provide preliminary evidence of efficacy in patients whose melanomas progressed despite prior anti-PD-1 therapy.

OBJECTIVE

The primary objective of the study is to assess the safety of ONCOS-102 in combination with pembrolizumab either given sequentially (Part 1) or combined (Part 2) for patients with aPD-1 resistant melanoma. Secondary objectives include overall response rate by RECIST 1.1 and the immune activation in tumor mass and peripheral blood, as well as correlation of immune markers and clinical outcome.

MECHANISM OF ACTION

ONCOS-102 is a serotype 5 adenovirus armed with a granulocyte-macrophage colony stimulating factor (GM-CSF) for enhanced immune stimulation with a unique ability to both prime and boost immune responses.
ONCOS-102 represents a promising immunotherapy strategy for advanced cancer as it directly recruits antigen presenting cells (APC) to the tumor site leading to induction of adaptive tumor-specific CD8+ T cell response (**Fig. 1**).

Fig 1: Mechanism of action of ONCOS-102.

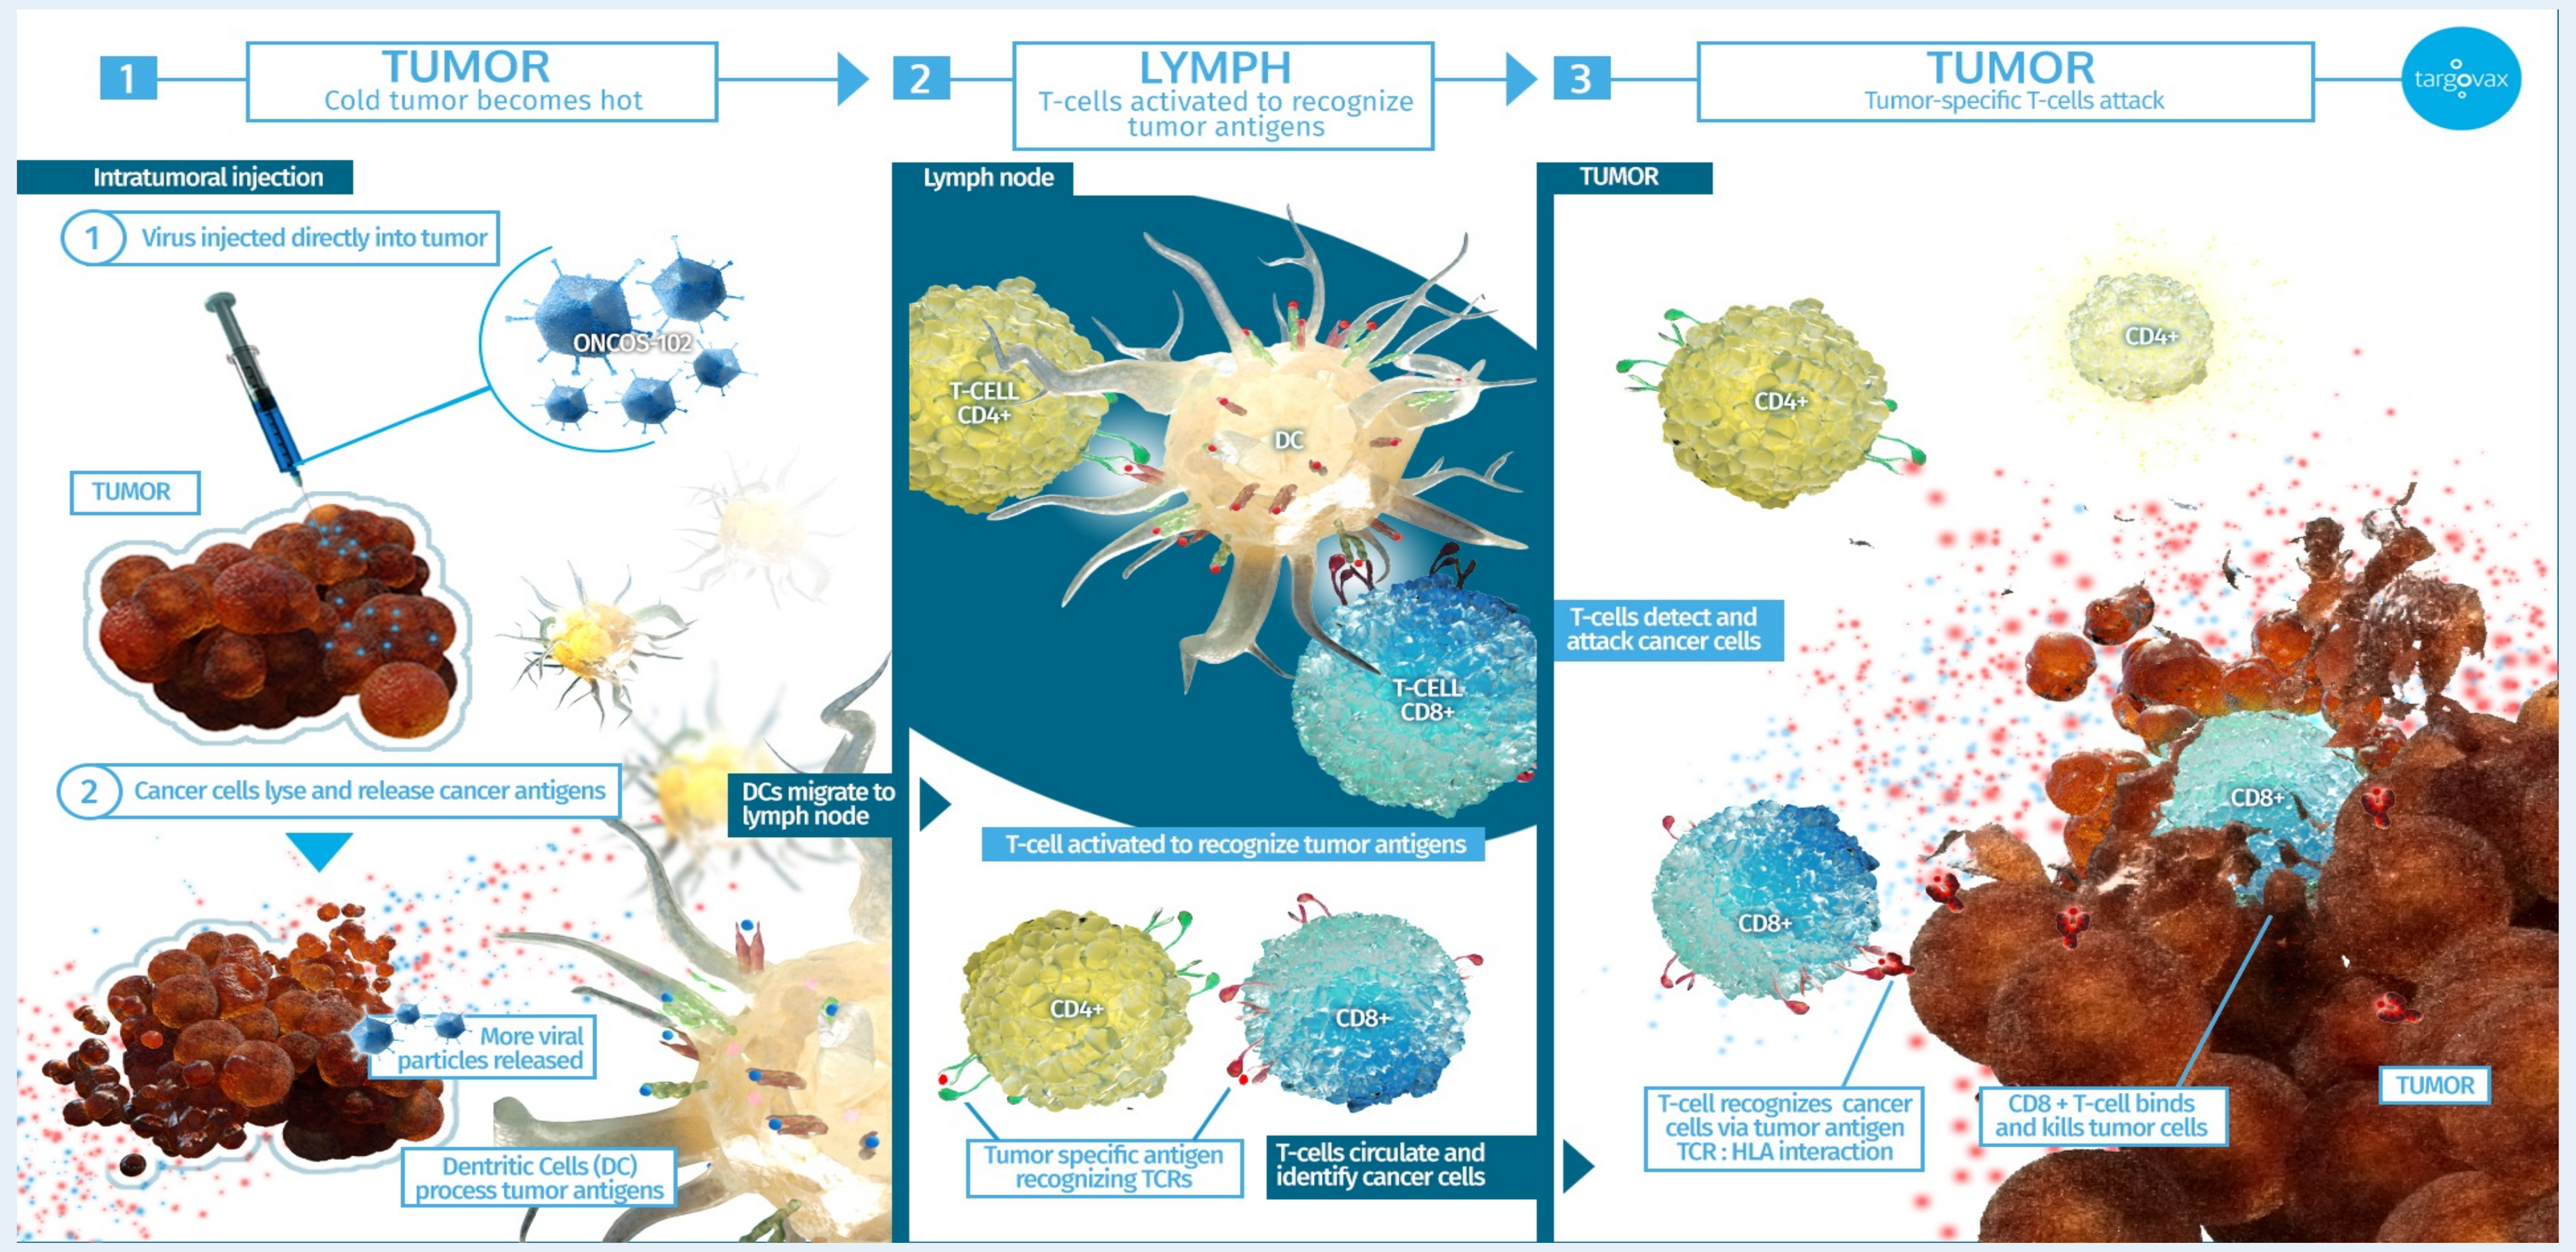
1 TUMOR Cold tumor becomes hot
2 LYMPH T-cells activated to recognize tumor antigens
3 TUMOR Tumor-specific T-cells attack
Intratumoral injection
1 Virus injected directly into tumor
ONCOS-102
TUMOR
2 Cancer cells lyse and release cancer antigens
More viral particles released
Dentritic Cells (DC) process tumor antigens
DCs migrate to lymph node
Lymph node
T-CELL CD4+
DC
T-CELL CD8+
T-cell activated to recognize tumor antigens
CD4+
CD8+
Tumor specific antigen recognizing TCRs
T-cells circulate and identify cancer cells
TUMOR
CD4+
T-cells detect and attack cancer cells
CD8+
T-cell recognizes cancer cells via tumor antigen TCR : HLA interaction
CD8 + T-cell binds and kills tumor cells
TUMOR

METHOD

This is a phase 1 prospective, open-label, multi-center pilot safety study. Patients with advanced or unreachable melanoma who had experienced radiographic progression of disease despite prior PD-1 blockade were eligible as long as they have at least 1 lesion measurable by RECIST 1.1. The study consists of two parts: Part I with sequential ONCOS-102 injection (day 1, 4, 8) with cyclophosphamide (CPO) priming followed by Pembrolizumab (up to 8 doses every 3 weeks, n=9) and Part 2, with initial ONCOS-102 injections (day 1, 4, 8, 15) primed with CPO followed by a treatment phase where ONCOS-102 and Pembrolizumab is given in combination (up to 8 times every 3 weeks, n=12, **Fig 2**). Samples for immune analysis (Biopsy, PBMC, Serum, whole blood and shedding samples) were collected throughout the study. The median time from failed aPD1 and study start was 1.9 months. Part 2 patients had significantly more advanced disease with more lesions and relatively more stage IV to stage III patients.

Fig 2: Treatment schedule.

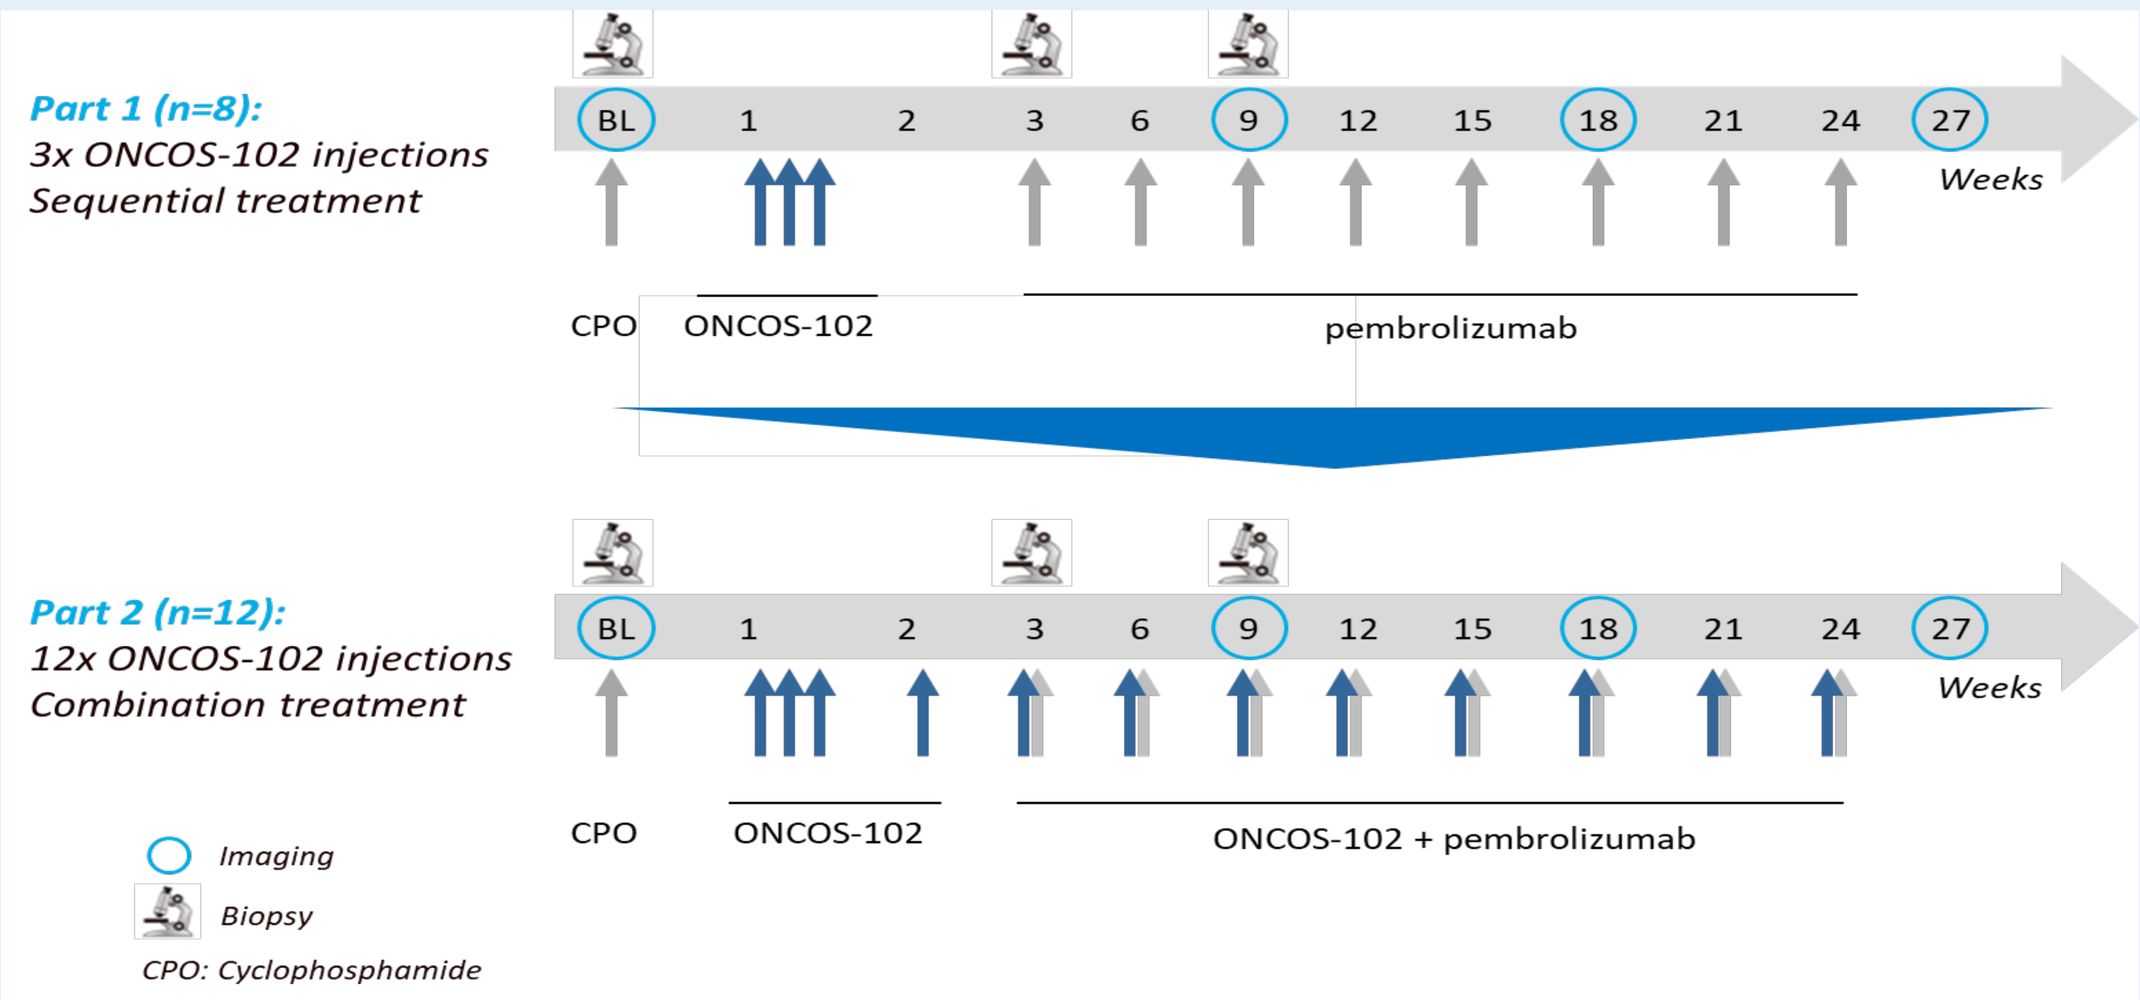
Part 1 (n=8):
3x ONCOS-102 injections
Sequential treatment
BL 1 2 3 6 9 12 15 18 21 24 27
Weeks
CPO ONCOS-102 pembrolizumab
Part 2 (n=12):
12x ONCOS-102 injections
Combination treatment
CPO ONCOS-102 ONCOS-102 + pembrolizumab
Imaging
Biopsy
CPO: Cyclophosphamide

RESULTS – Clinical efficacy

Twenty patients were evaluable for efficacy. Across the study, 7 of 20 patients (35%) achieved a best objective response (BORR) during the treatment period using both RECIST 1.1 and irRECIST, including 1 patient with a complete response in recurrent in-transit lesion. In Part 1, 3 patients had CR or PR during the study according to RECIST 1.1 and irRECIST resulting in a BORR of 38%. In Part 2, 4 of 12 patients had CR or PR during the study according to RECIST 1.1 and irRECIST resulting in a BORR of 33% (**Fig 3**). A case example (image) is shown in **Fig 4.** Reduction in size of non-injected lesions was seen in 12/36 (33%) of targeted measured non-injected lesions with examples of complete disappearance of lesions in two patients. The combination treatment was well tolerated, mostly grade 1/2 with five grade 3/4 events in three patents attributed to pembrolizumab, ONCOS-102 or both.

Fig 3: Relative change (percent) in tumor burden measured by PET/CT from baseline to best response.

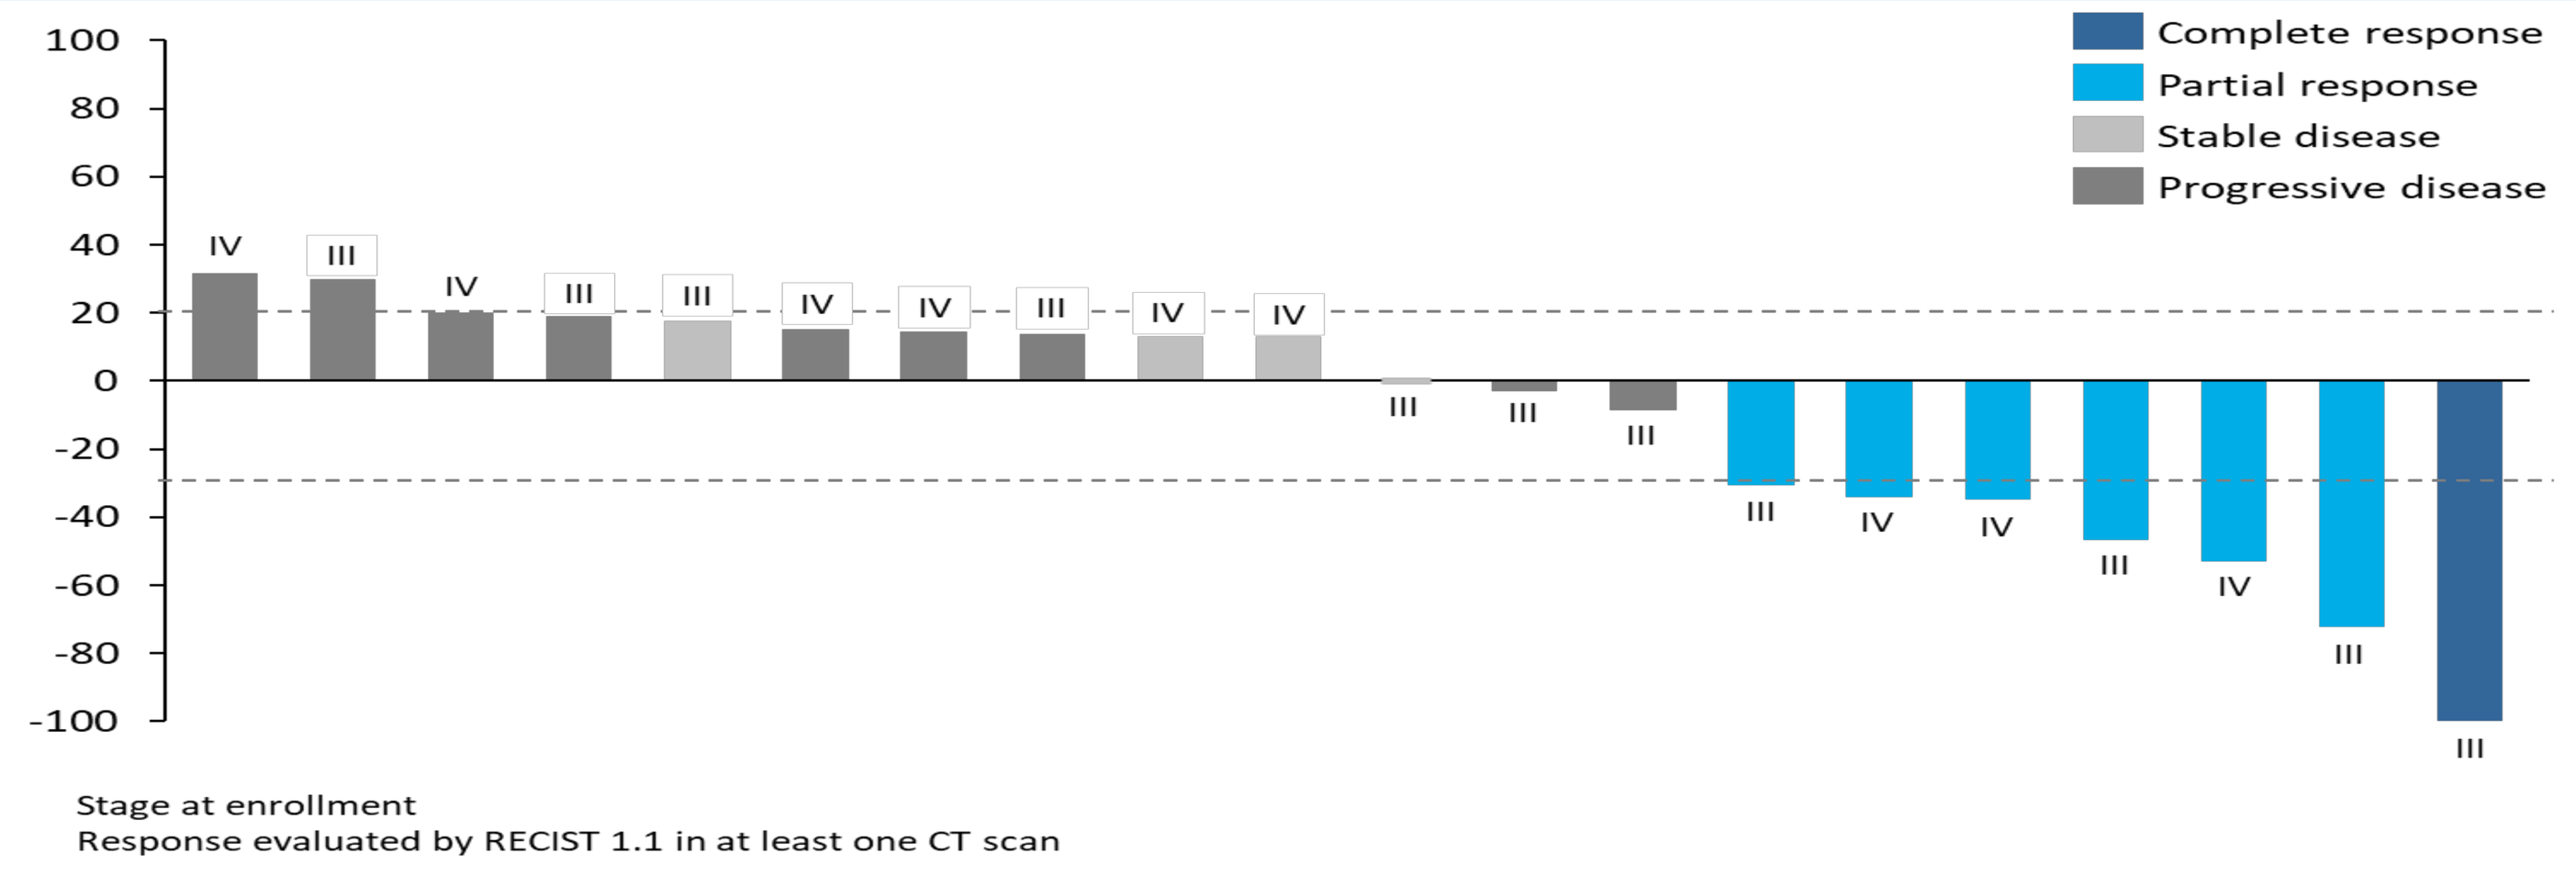
Complete response
Partial response
Stable disease
Progressive disease
100 80 60 40 20 0 -20 -40 -60 -80 -100
IV III IV III III IV IV III IV IV III III III III IV IV III IV III III
Stage at enrollment
Response evaluated by RECIST 1.1 in at least one CT scan

Fig 4: Case example of patient with partial response (week 9-27) refractory to both T-VEC and anti-PD-1.

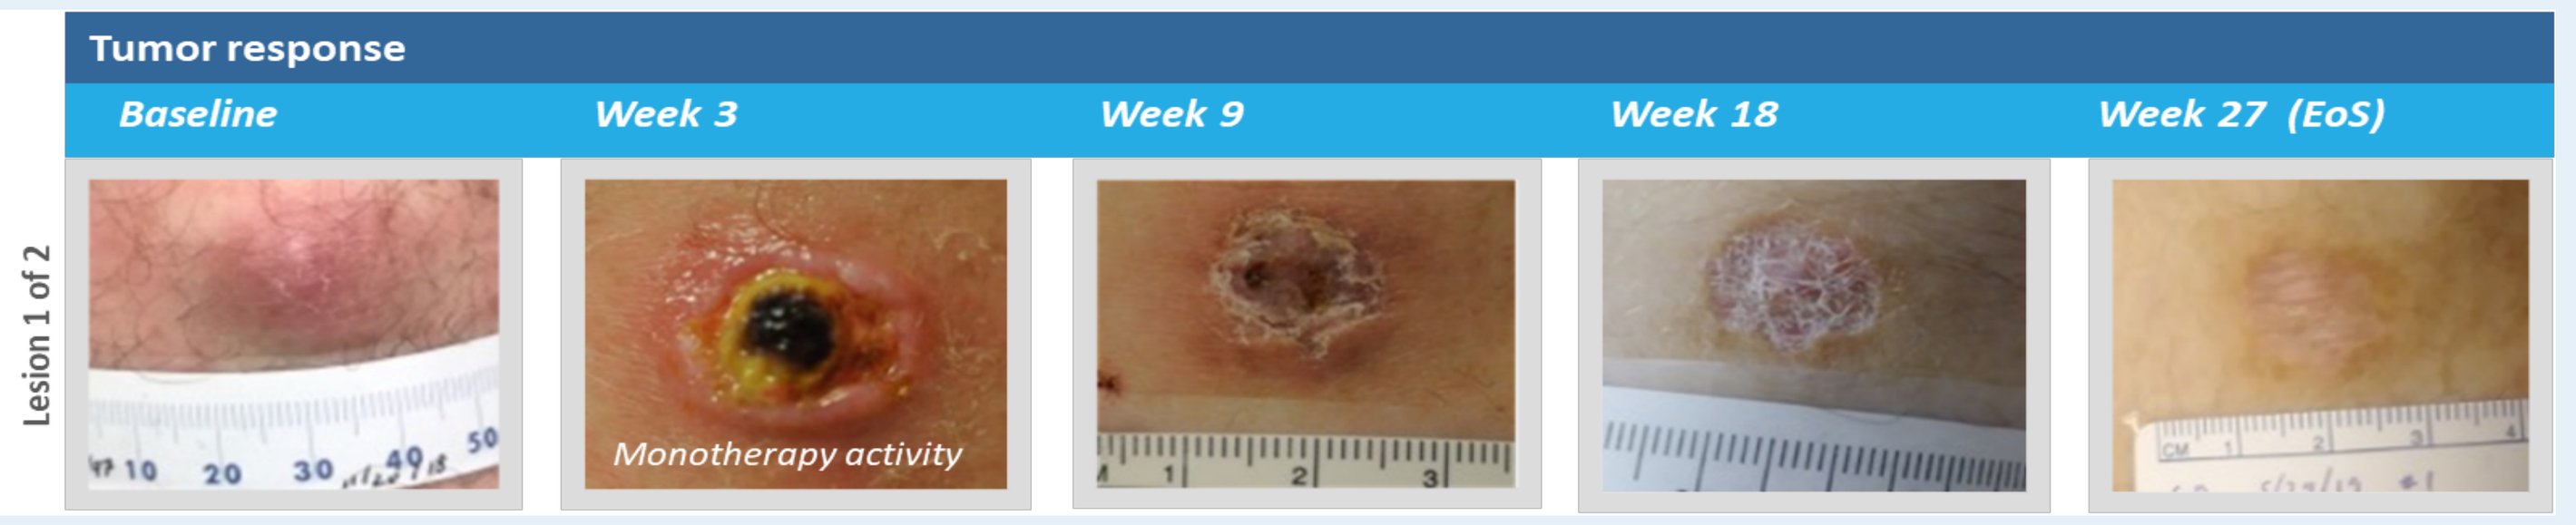
Tumor response
Baseline
Week 3
Week 9
Week 18
Week 27 (EoS)
Lesion 1 of 2
Monotherapy activity

RESULTS – Immune activation

Analysis of immune cell subsets in biopsies (multiplex immunohistochemistry ((mIHC) / multicolour immunofluorescence (mIF) taken at baseline and on Day 22 and/or Day 64 of the same tumours indicated immuno-stimulatory effects of ONCOS-102 in combination with anti-PD1. Several biologically meaningful changes such as increased frequencies of CD8+ or CD4+ T-cells appeared to be more prominent in tumour samples from CR, PR and even SD patients as compared to patients with PD, in spite of the fact the observed differences between the groups of patients did not reach statistical significance (**Fig 5**). A case example showing mIHC from a patient with complete response (**Fig 6**).

Fig 5: Immune cell infiltrates: CD8+ and CD4+ T cells (average level by tumor).

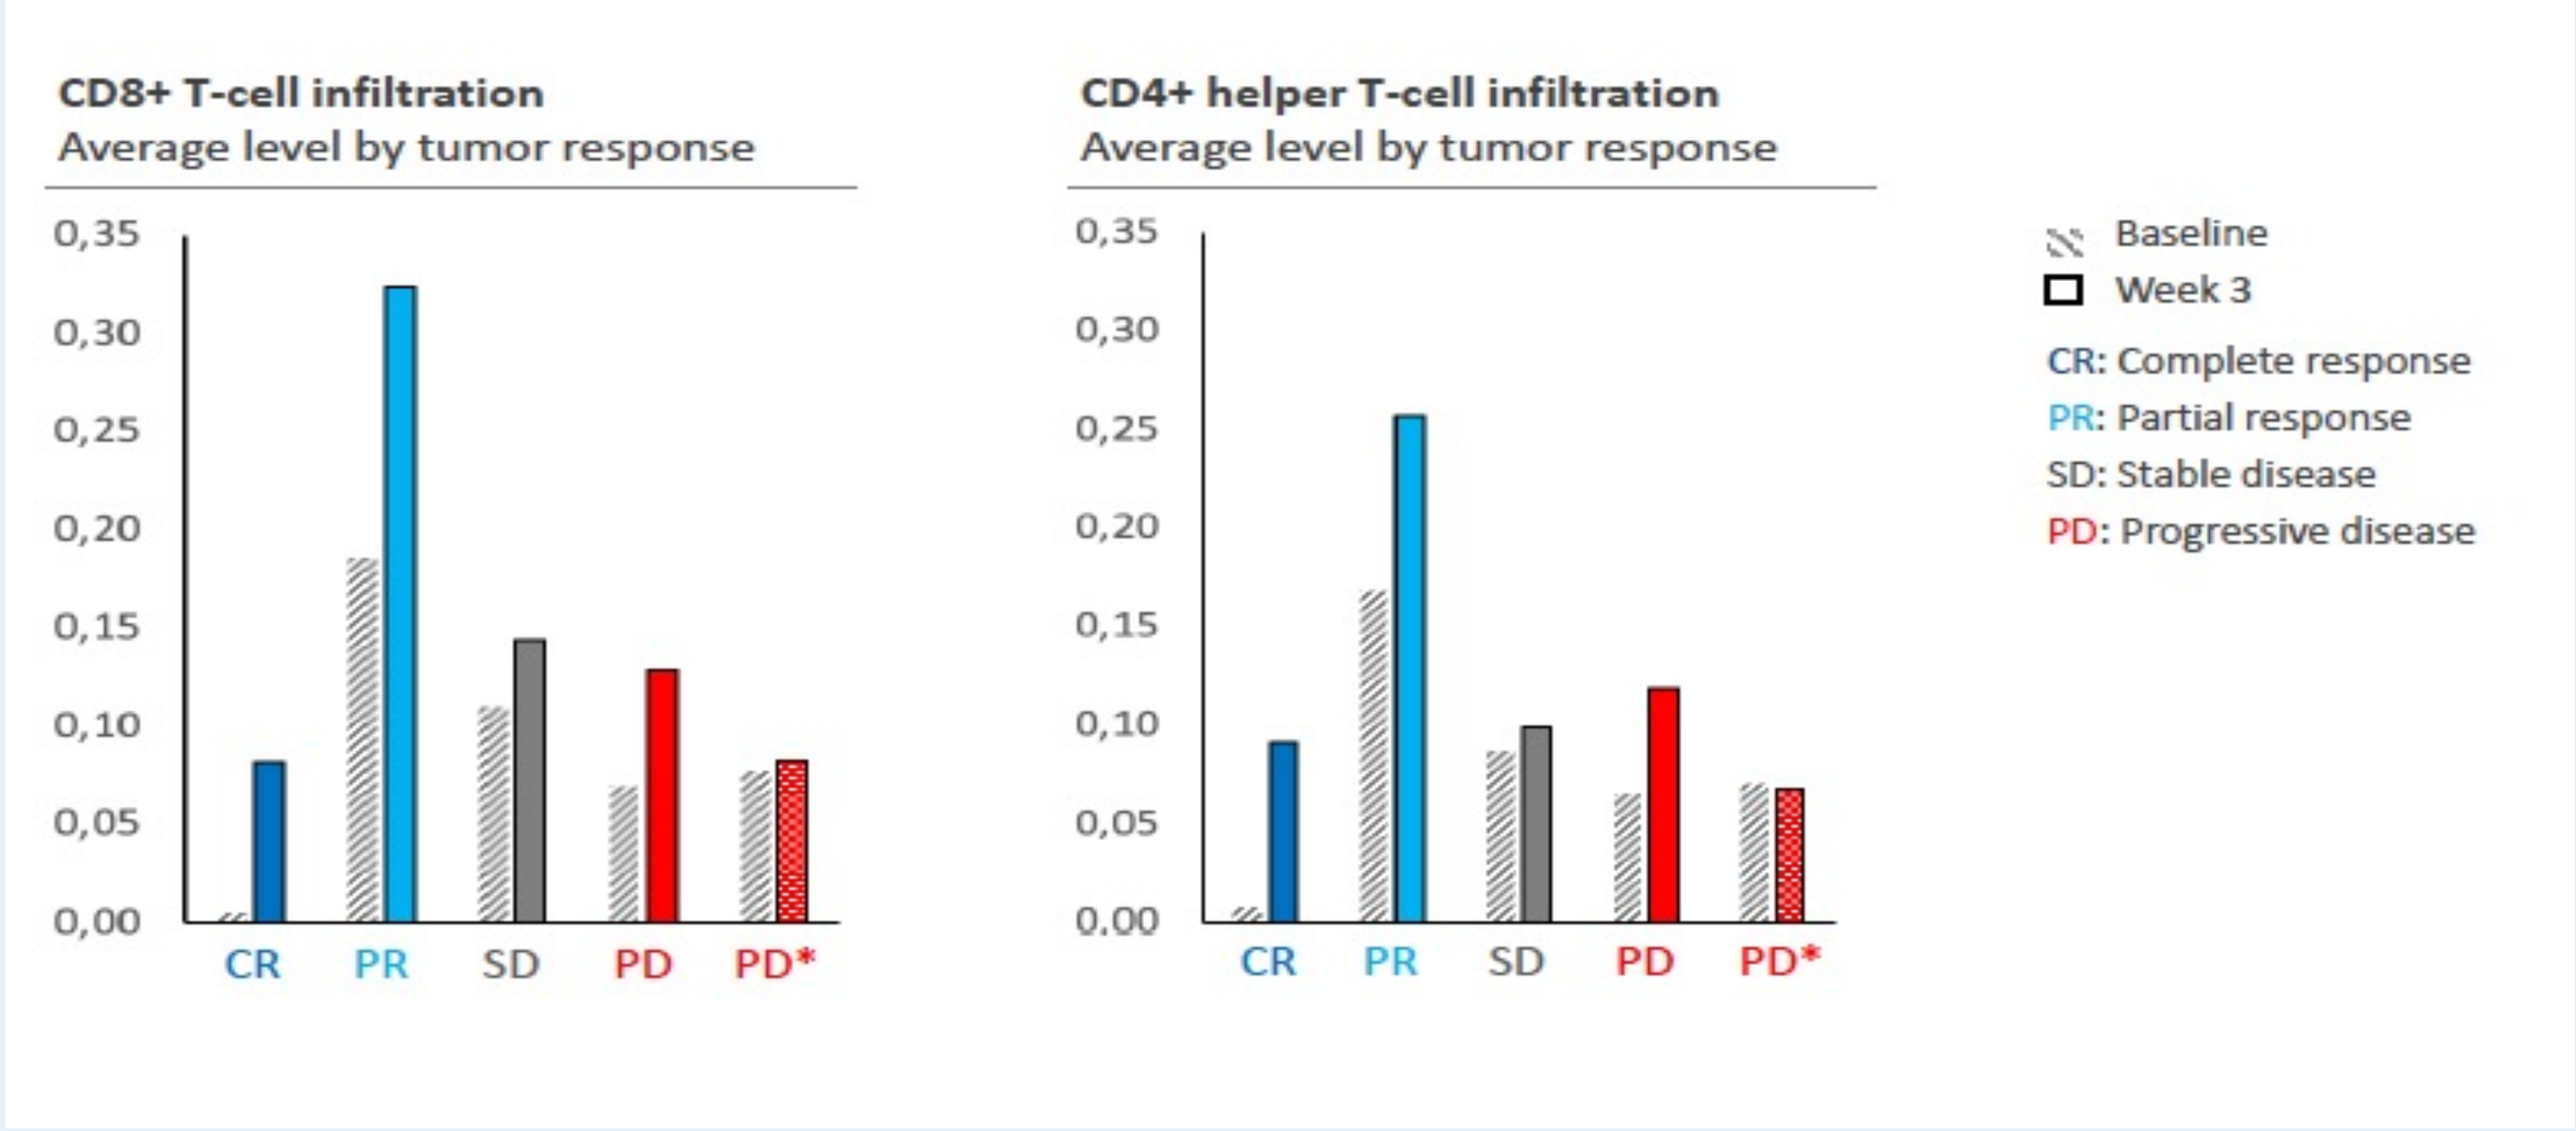
CD8+ T-cell infiltration
Average level by tumor response
CD4+ helper T-cell infiltration
Average level by tumor response
0,35 0,30 0,25 0,20 0,15 0,10 0,05 0,00
CR PR SD PD PD*
Baseline
Week 3
CR: Complete response
PR: Partial response
SD: Stable disease
PD: Progressive disease

Fig 6: Case example of tumor infiltrating lymphocytes in patient with Complete response

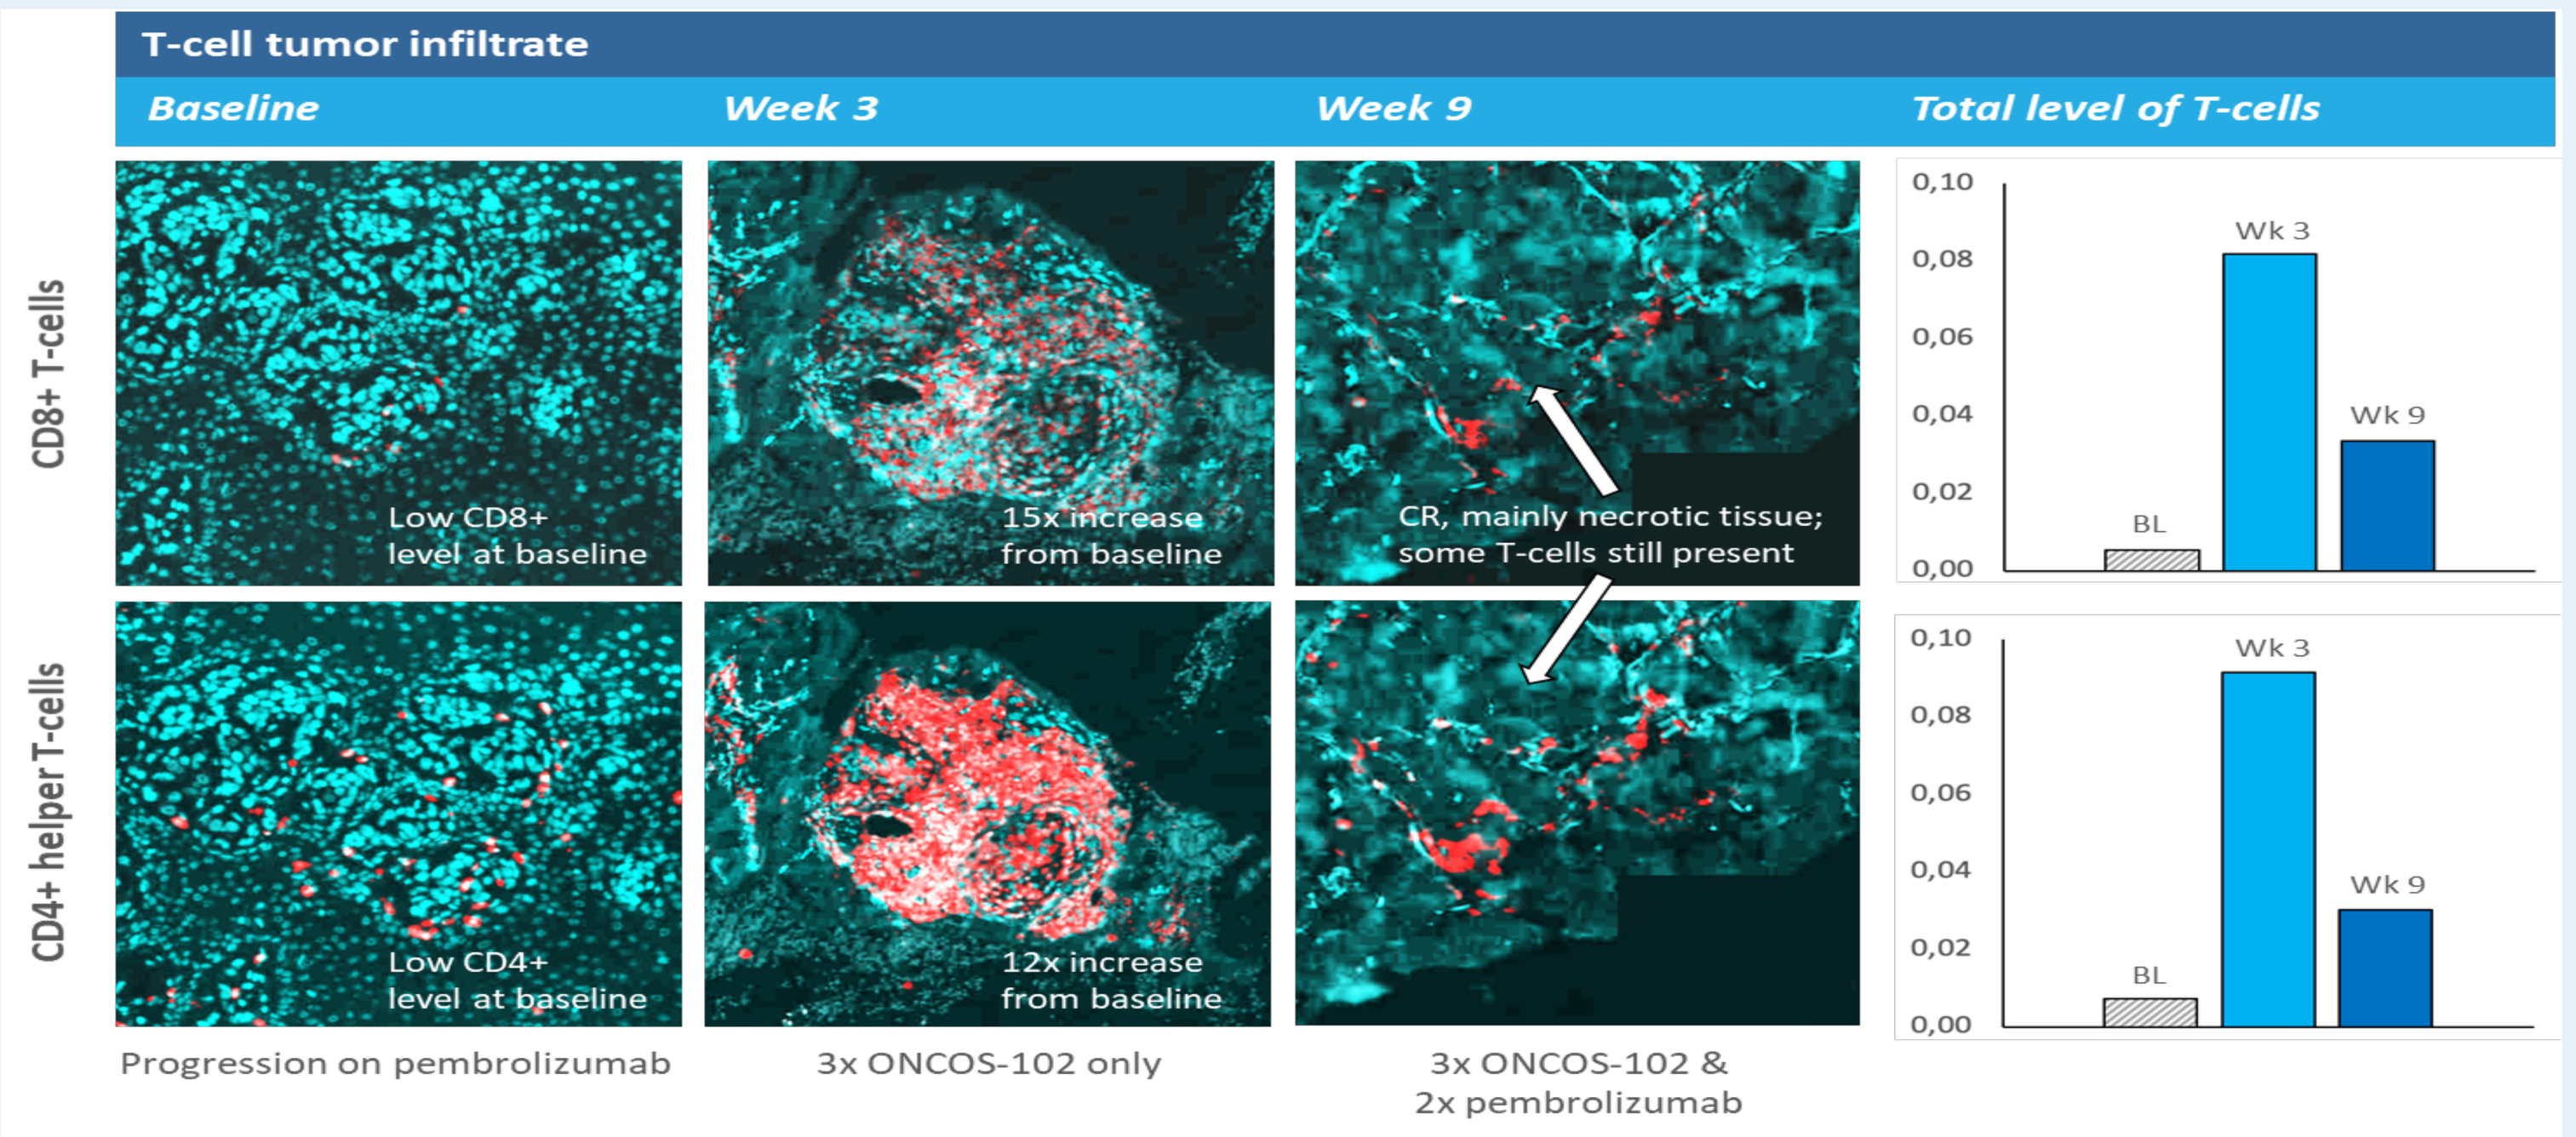
T-cell tumor infiltrate
Baseline
Week 3
Week 9
Total level of T-cells
CD8+ T-cells
CD4+ helper T-cells
Low CD8+ level at baseline
15x increase from baseline
CR, mainly necrotic tissue; some T-cells still present
Low CD4+ level at baseline
12x increase from baseline
Progression on pembrolizumab
3x ONCOS-102 only
3x ONCOS-102 & 2x pembrolizumab
0,10 0,08 0,06 0,04 0,02 0,00
BL
Wk 3
Wk 9

CONCLUSION

We conclude that co-administration of ONCOS-102 and pembrolizumab is safe and feasible for patients with melanomas progressing on PD-1 blockade. Rapid clinical objective responses were seen in patients treated both sequentially and in combination, and immune markers demonstrating induction of beneficial TME changes support the role of ONCOS-102 as a complementary treatment with aPD1 and other IO modalities.

Disclosure

Financial Interests: Bristol-Myers Squibb, Immunocore. Royalties: UpToDate. Research Funding: Targovax ASA, Bristol-Myers Squibb, Immunocore, Polaris, Xcovery, Pfizer, Novartis, Checkmate Pharmaceuticals

www.Targovax.com
E-mail : shoushta@mskcc.org
E-mail: magnus.jaderberg@targovax.com