Optical-Guided Surgery in Patients with Oral and Oropharyngeal Squamous Cell Carcinoma (OSCC & OPSCC) Using a Novel uPAR-targeting Near-Infrared Imaging Agent FG001: An Explorative Phase II Clinical Trial – a Case Example

Amanda Oester Andersen[+,1], Anders Christensen[+,1,2], Karina Juhl[2,4], Mads Lawaetz[1], Christoffer Holst Hahn[1], Nicklas Rubaek[1], Irene Wessel[1], Giedrius Lelkaitis[3], Katalin Kiss[3], Natasja Paaske[1], Anne Poulsen[4], Christian von Buchwald[1], Andreas Kjaer[2]

[1]Department of Otorhinolaryngology, Head and Neck Surgery and Audiology, Rigshospitalet, University of Copenhagen, Denmark
[2]Department of Clinical Physiology and Nuclear Medicine & Cluster for Molecular Imaging, Copenhagen University Hospital – Rigshospitalet & Department of Biomedical Sciences, University of Copenhagen, Denmark
[3]Departement of Pathology, Rigshospitalet, University of Copenhagen, Denmark
[4]FluoGuide A/S, Copenhagen, Denmark

INTRODUCTION

Identification and delineation of cancerous tissue is a key element in the surgical treatment of cancer to achieve adequate resection margins. Realtime intraoperative optical-guided surgery (OGS) combined with a tumor-specific fluorescent imaging agent has been proposed as a compelling strategy to enhance surgical vision and thereby assist the surgeon in more precise resection of the tumor [1,2]. FG001 (AE105-Glu-Glu-ICG) is designed to bind to the urokinase-type plasminogen activator receptor (uPAR), which is a receptor in the plasminogen activator (PA) system that is overexpressed in a variety of carcinoma types [3,4]. Extensive preclinically testing of FG001 has been preformed in mice models [5]. Furthermore, a first-in-human study (EudraCT no.: 2020-003089-38) with 40 GBM patients has been completed successfully – indicating high safety and tolerability.

OBJECTIVE

To explore the potential and specificity of the novel uPAR-targeting near-infrared (NIR) optical imaging agent FG001 in a case example for detection of oral squamous cell carcinoma (OSCC).

METHODS

In an ongoing phase II clinical trial, a 74-year-old male subject with a lateral tongue tumor (T3N2cM0) was enrolled. The patient was systemically administered 16 mg of FG001 and surgically treated for OSCC. The tumor and metastasis was visualized peri- and intraoperatively with a dedicated NIR camera-system. Following surgery, the microscopic tumor-specificity of FG001 was confirmed with histological evaluation of the tumor cross-section. The patient was safety-monitored with AE-registrations (CTCAE v5.0), blood samples, and ECGs.

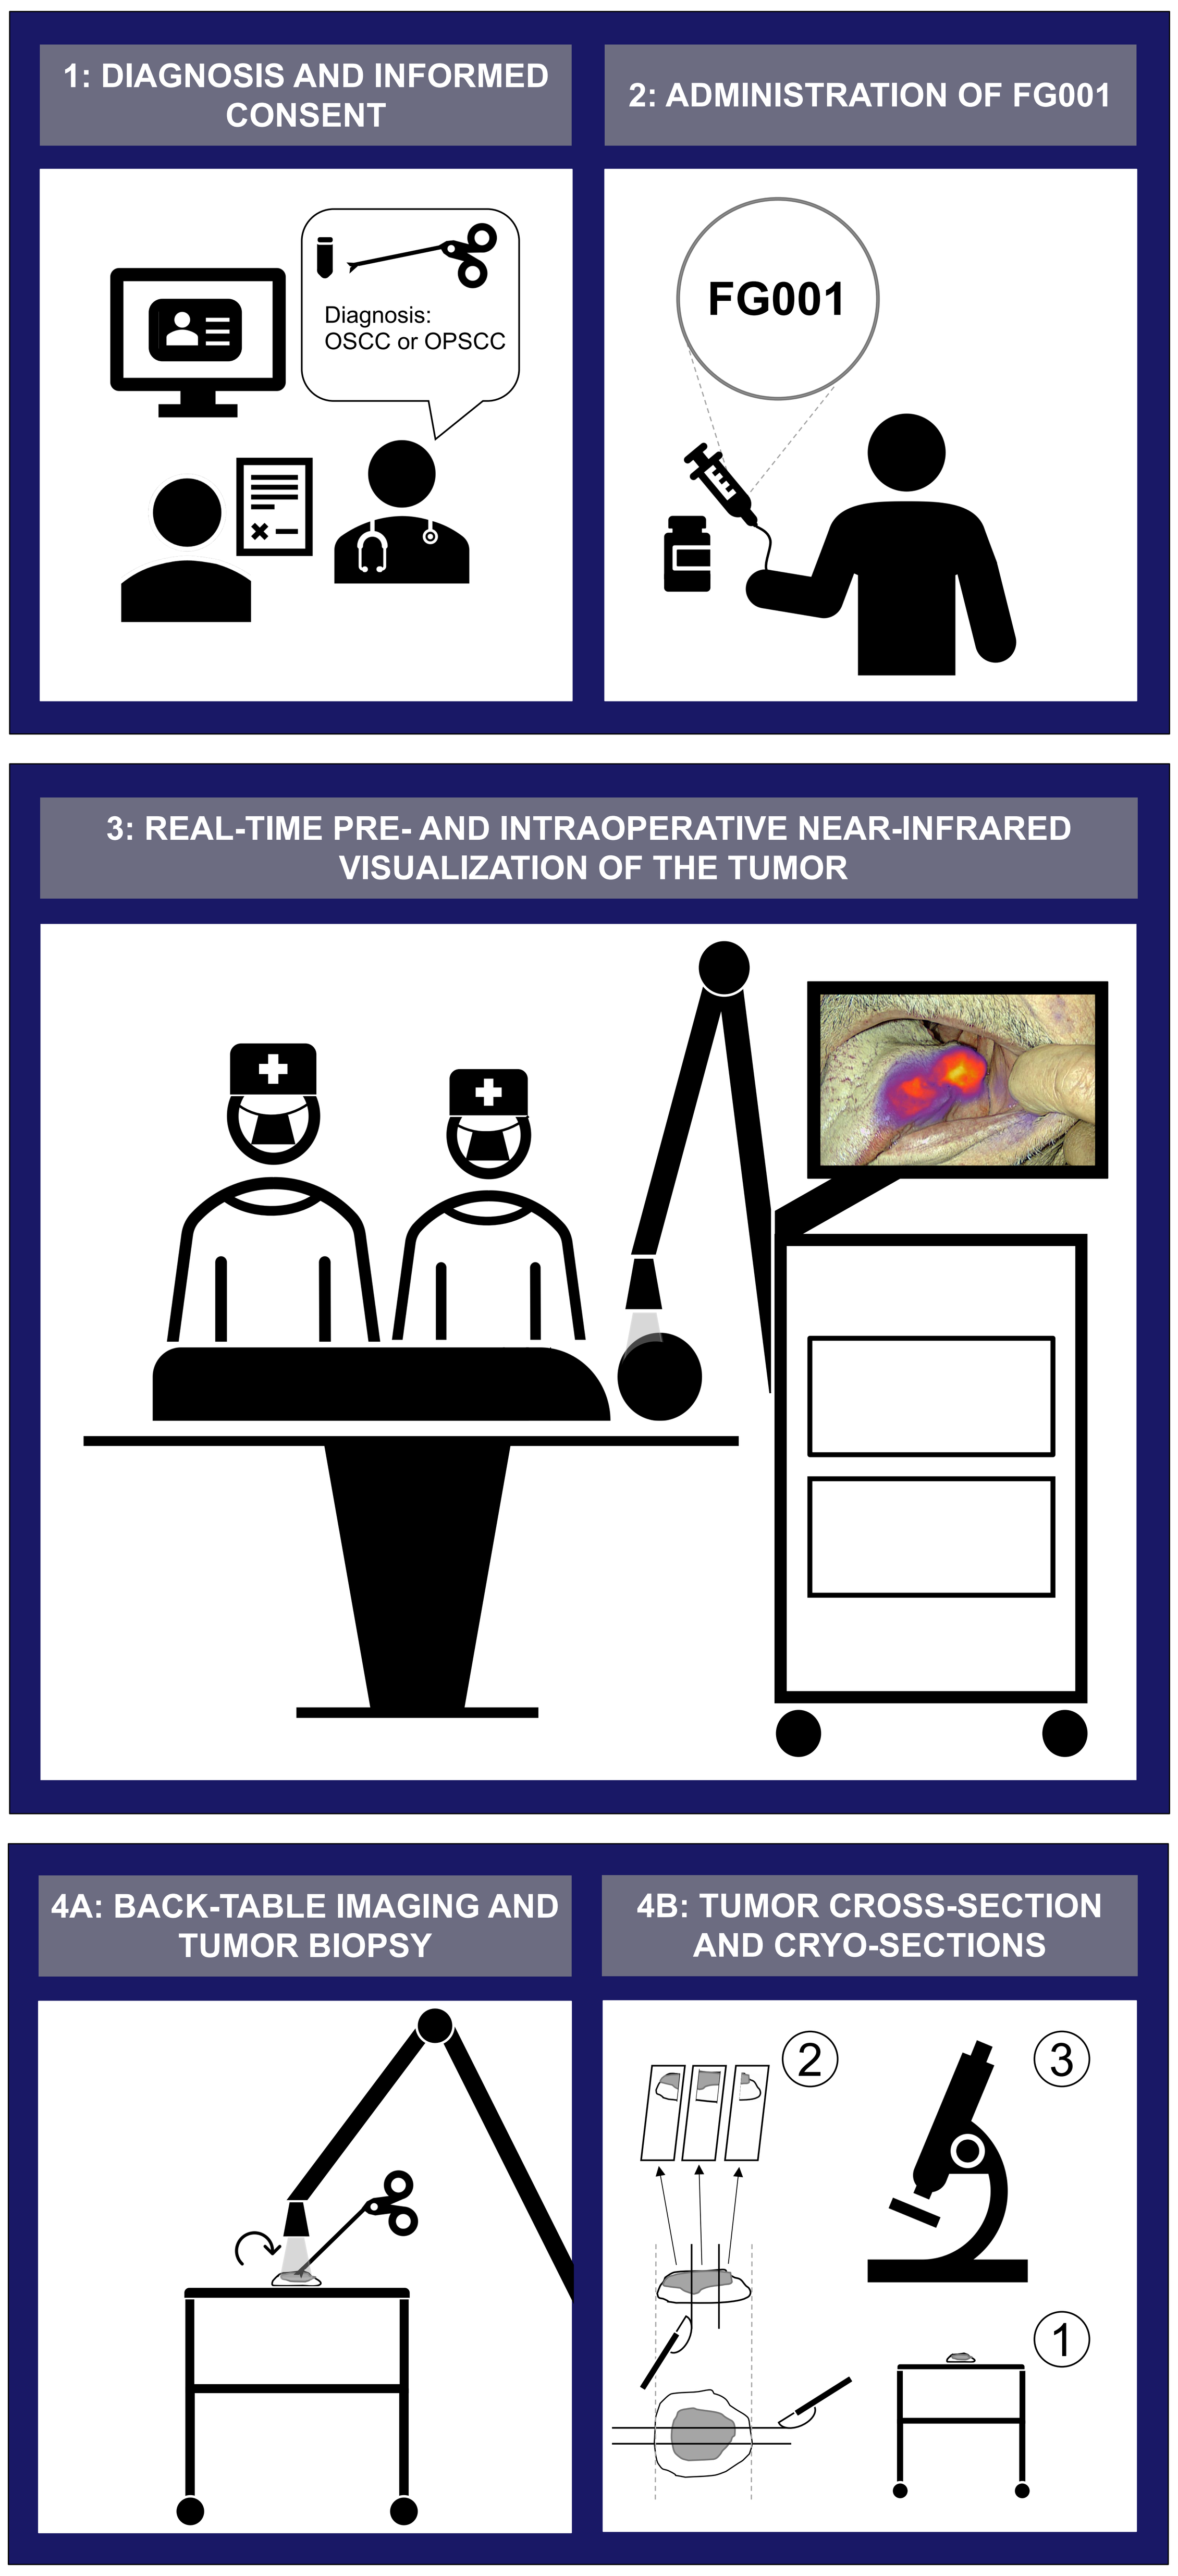

1: DIAGNOSIS AND INFORMED CONSENT
Diagnosis:
OSCC or OPSCC
2: ADMINISTRATION OF FG001
FG001
3: REAL-TIME PRE- AND INTRAOPERATIVE NEAR-INFRARED VISUALIZATION OF THE TUMOR
4A: BACK-TABLE IMAGING AND TUMOR BIOPSY
4B: TUMOR CROSS-SECTION AND CRYO-SECTIONS
2
3
1

Figure 1: Clinical workflow from patient inclusion to macro- and microscopic tumor-specificity evaluation of FG001.

RESULTS

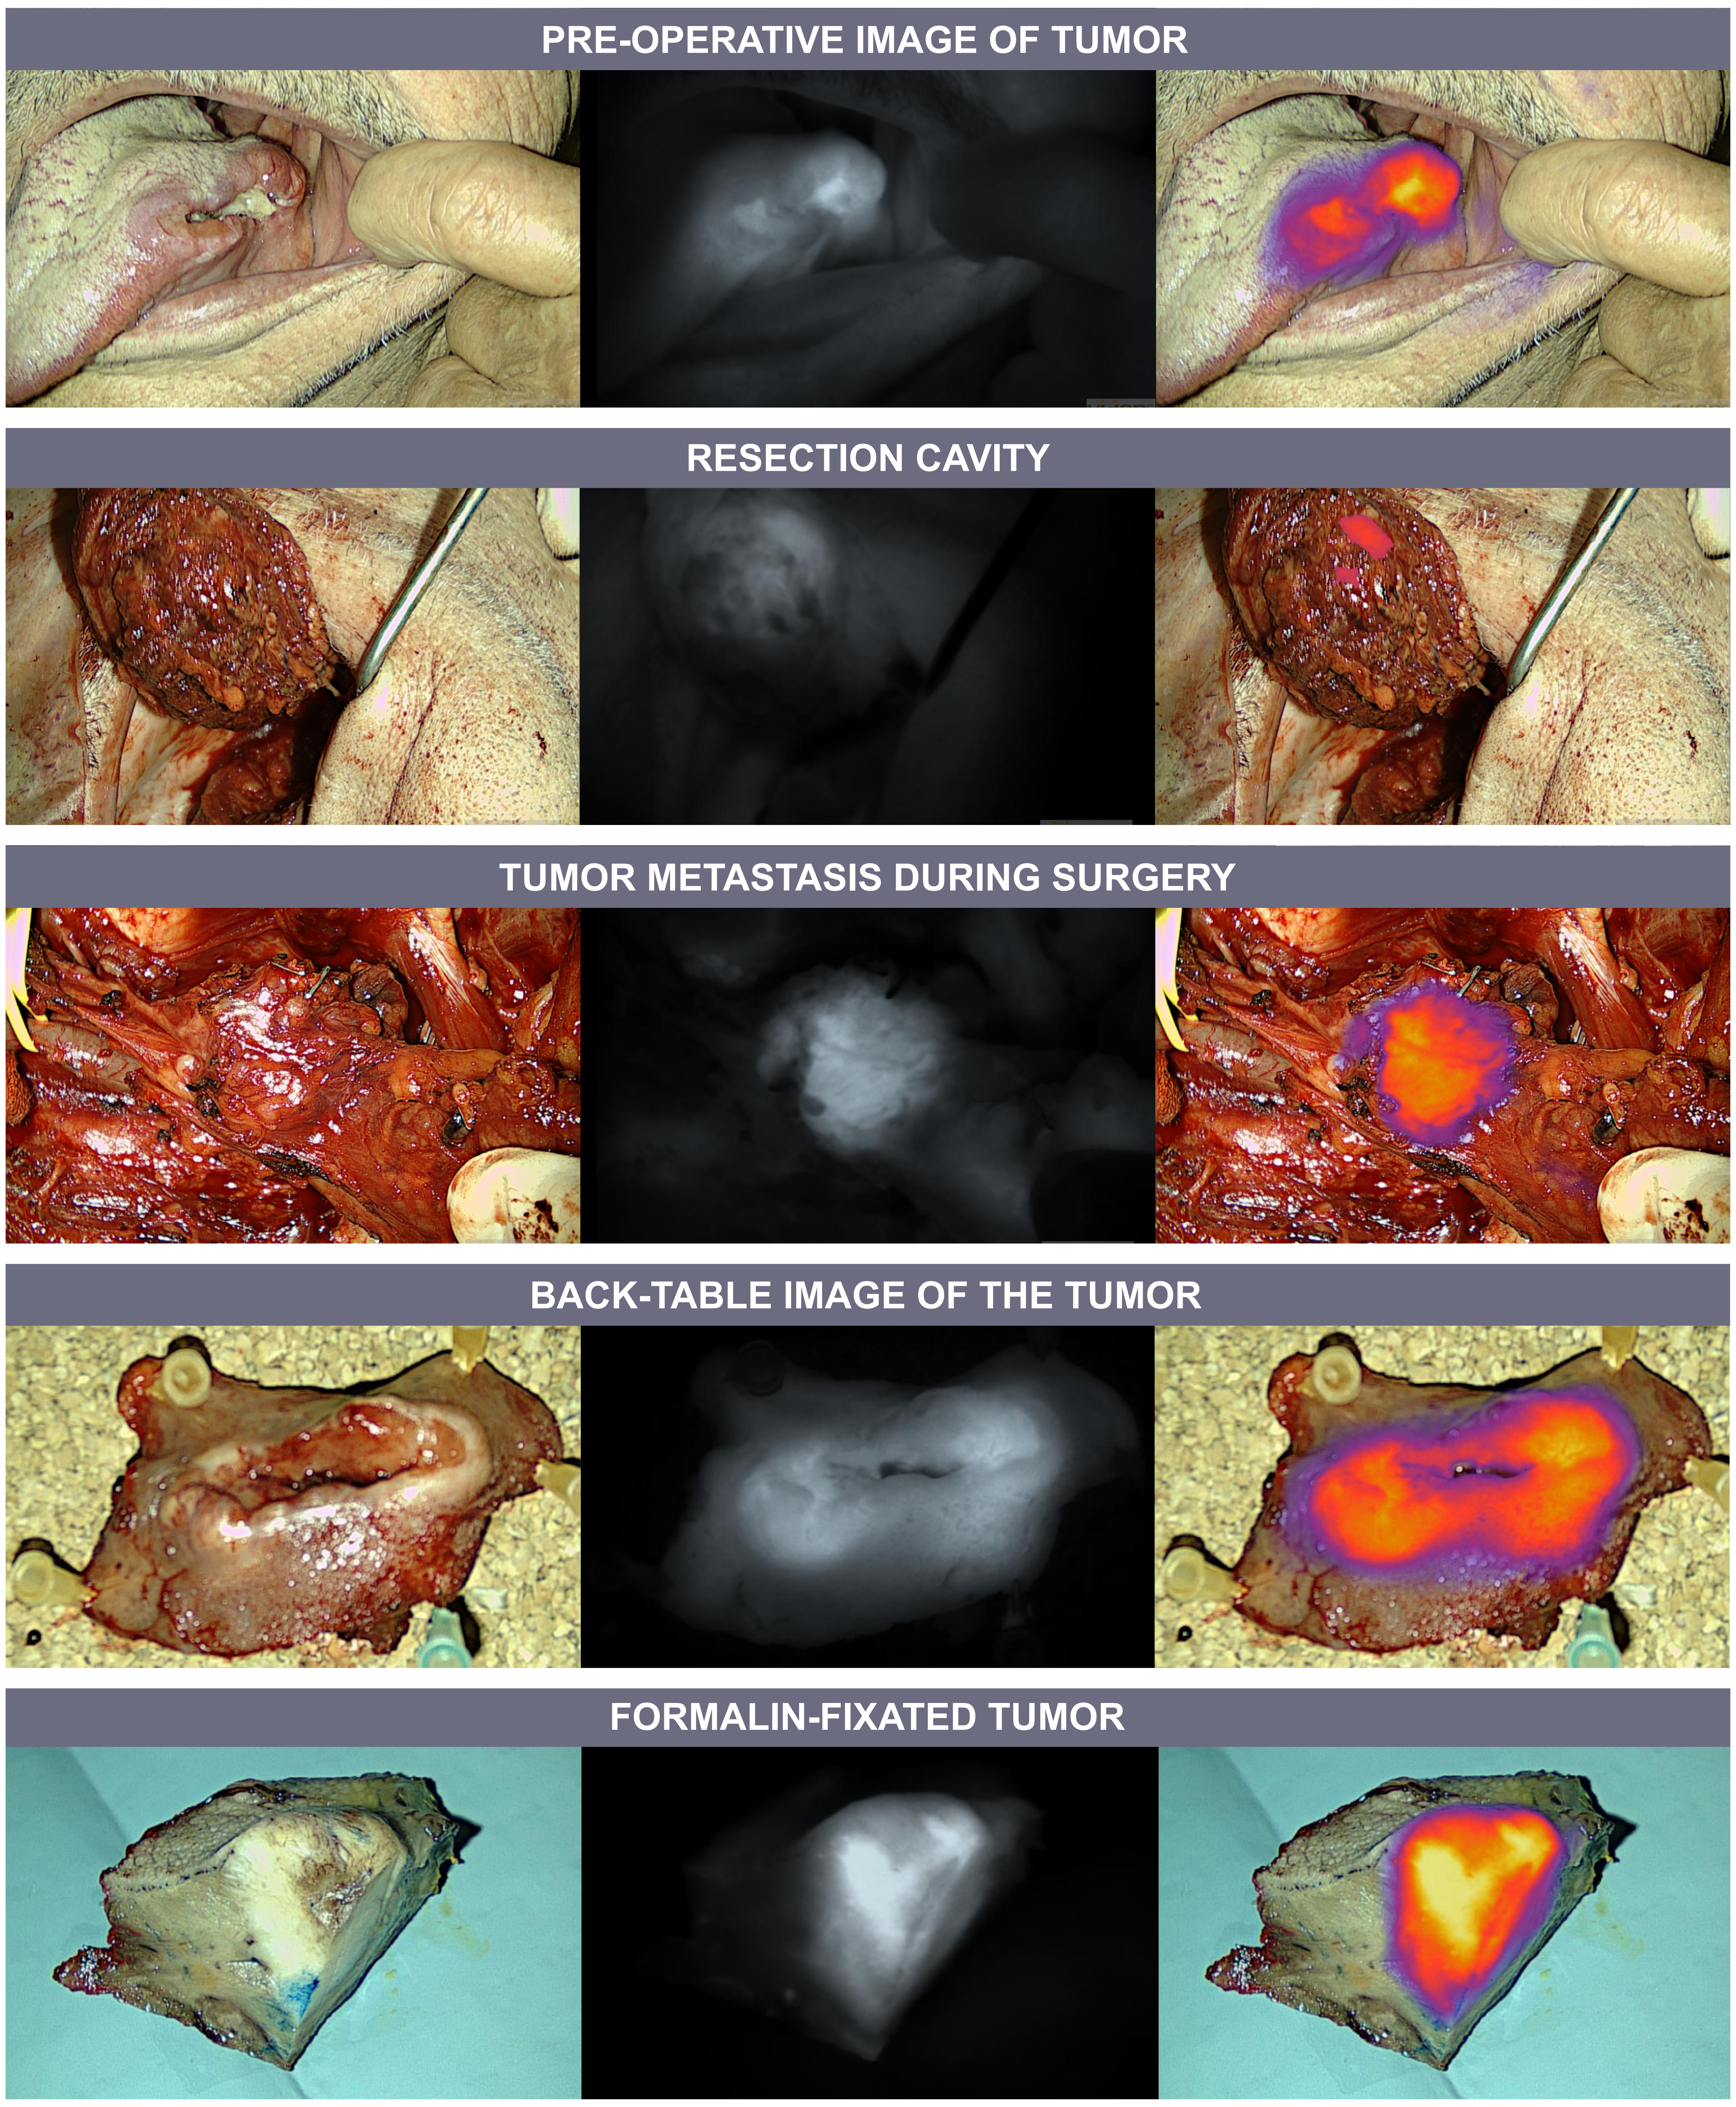

PRE-OPERATIVE IMAGE OF TUMOR
RESECTION CAVITY
TUMOR METASTASIS DURING SURGERY
BACK-TABLE IMAGE OF THE TUMOR
FORMALIN-FIXATED TUMOR

Figure 2: Real-time NIR imaging of the OSCC patient tumor and metastasis taken at the indicated time-point. To the left showing: white light, middle: NIR, and right: overlay of NIR signal on white light image.

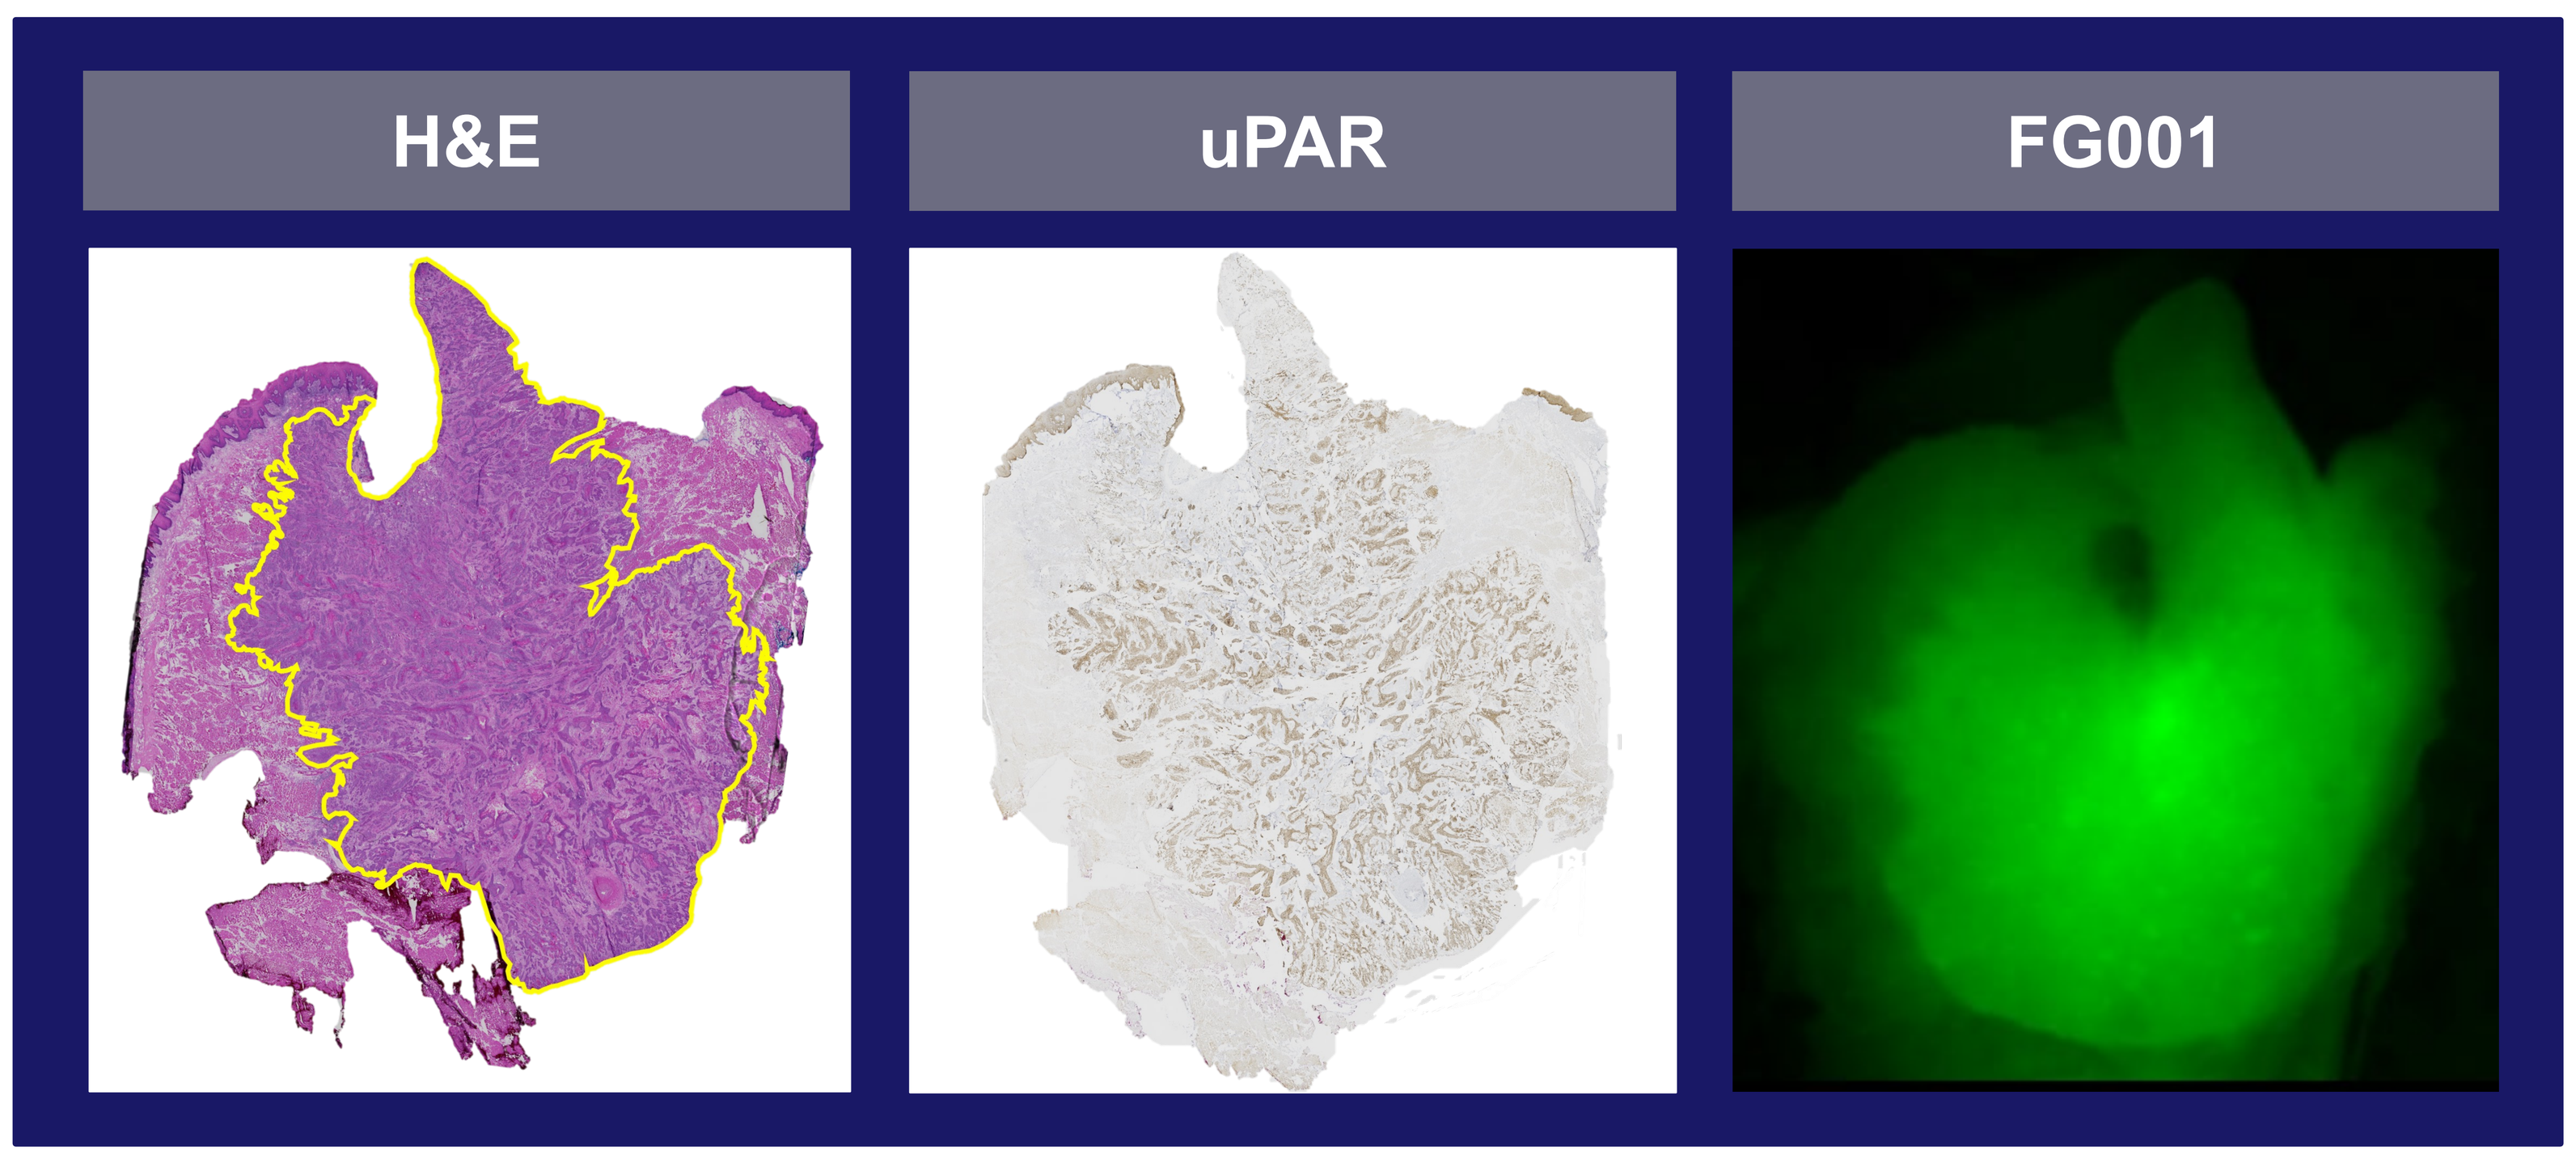

H&E
uPAR
FG001

Figure 3: Microscopic tumor-specificity evaluation of FG001 using cryo-sections from a tumor cross-section (below). The cryo-sections were stained with hematoxylin and eosin (H&E) and for uPAR-expression (in brown). The FG001 signal was captured with the NIR camera-system on the back-table.

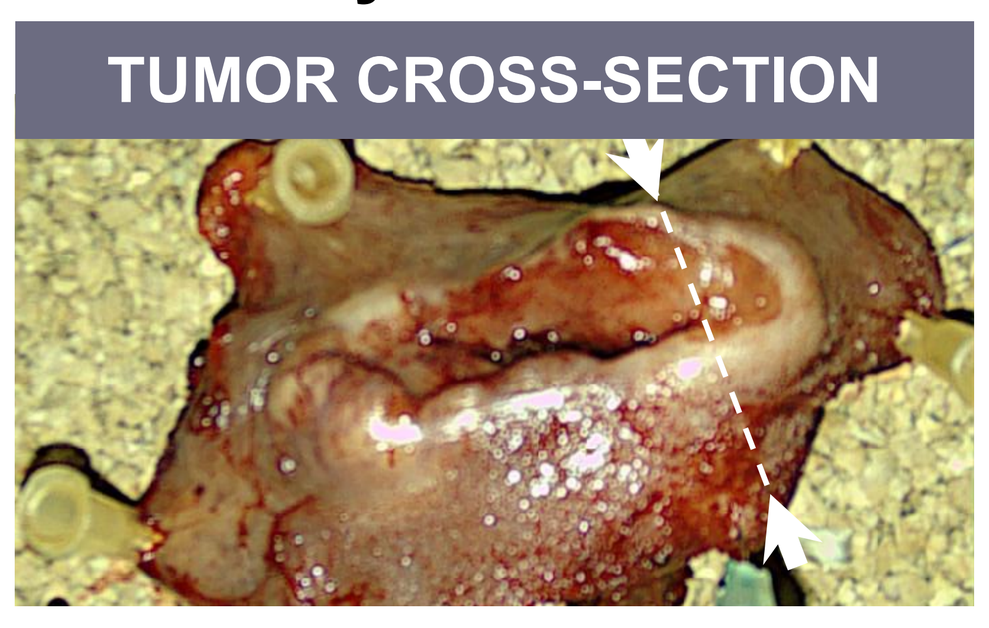

TUMOR CROSS-SECTION

CONCLUSION

In this case example, FG001 was successfully used to visualize, with sufficient contrast, tumor and the metastasis of a patient with OSCC. The signal was preserved after formalin-fixation at the pathological evaluation. Histological co-localization of the tumor tissue, uPAR-expression, and FG001 signal was confirmed microscopically. The safety data and further conclusions awaits trial finalization and database closure.

REFERENCES

[1]: Mieog JSD, Achterberg FB, Zlitni A, Hutteman M, Burggraaf J, Swijnenburg RJ, et al. Fundamentals and developments in fluorescence-guided cancer surgery. Nat Rev Clin Oncol 2022;19:9–22. https://doi.org/10.1038/s41571-021-00548-3.

[2]: Hernot S, Van Manen L, Debie P, Sven J, Mieog D, Lucas Vahrmeijer A. Review Latest developments in molecular tracers for fluorescence image-guided cancer surgery. vol. 20. 2019.

[3]: Persson M, Kjaer A. Urokinase-type plasminogen activator receptor (uPAR) as a promising new imaging target: Potential clinical applications. Clin Physiol Funct Imaging 2013;33:329–37. https://doi.org/10.1111/cpf.12037.

[4]: Christensen A, Kiss K, Lelkaitis G, Juhl K, Persson M, Charabi BW, et al. Urokinase-type plasminogen activator receptor (uPAR), tissue factor (TF) and epidermal growth factor receptor (EGFR): Tumor expression patterns and prognostic value in oral cancer. BMC Cancer 2017;17. https://doi.org/10.1186/s12885-017-3563-3.

[5]: Christensen A, Juhl K, Persson M, Charabi BW, Mortensen J, Kiss K, et al. Oncotarget 15407 www.impactjournals.com/oncotarget uPAR-targeted optical near-infrared (NIR) fluorescence imaging and PET for image-guided surgery in head and neck cancer: proof-of-concept in orthotopic xenograft model. vol. 8. 2017.